ProGem1: A Phase I/II study of a first-in-class nucleotide analogue Acelarin (NUC-1031) in patients with advanced solid tumours

Imperial College London

Essam Ghazaly[1], Ivana Rizzuto[2], Hani Gabra[3], Nagy Habib[3], Robert Leonard[3], Harpreet Wasan[3], Christopher McGuigan[4], Sarah Blagden[2,3]

1) Centre for Haemato-Oncology, Barts Cancer Institute, Queen Mary University of London, UK; 2) NIHR/Wellcome Trust Imperial Clinical Research Facility, Imperial College, Du Cane Road, London, UK; 3) Department of Oncology, Imperial College, Du Cane Road, London, UK; 4) School of Pharmacy and Pharmaceutical Sciences, Cardiff University, King Edward VII Avenue, Cardiff, UK

Barts Cancer Institute

BACKGROUND

ProTides: Nucleotide Analogues

- New generation of anti-cancer agents
- Innovative phosphoramidate technology
- Overcome key cancer resistance mechanisms
- Broader utility to benefit a wide range of cancer patients
- Promising efficacy and safety profile

NUC-1031: The First Anti-Cancer ProTide

- Designed to overcome all the key resistant mechanisms associated with gemcitabine:
 - o Cellular uptake independent of nucleoside transporters
 - o Activation independent of deoxycytidine kinase
 - o Avoids cytidine deaminase inactivation

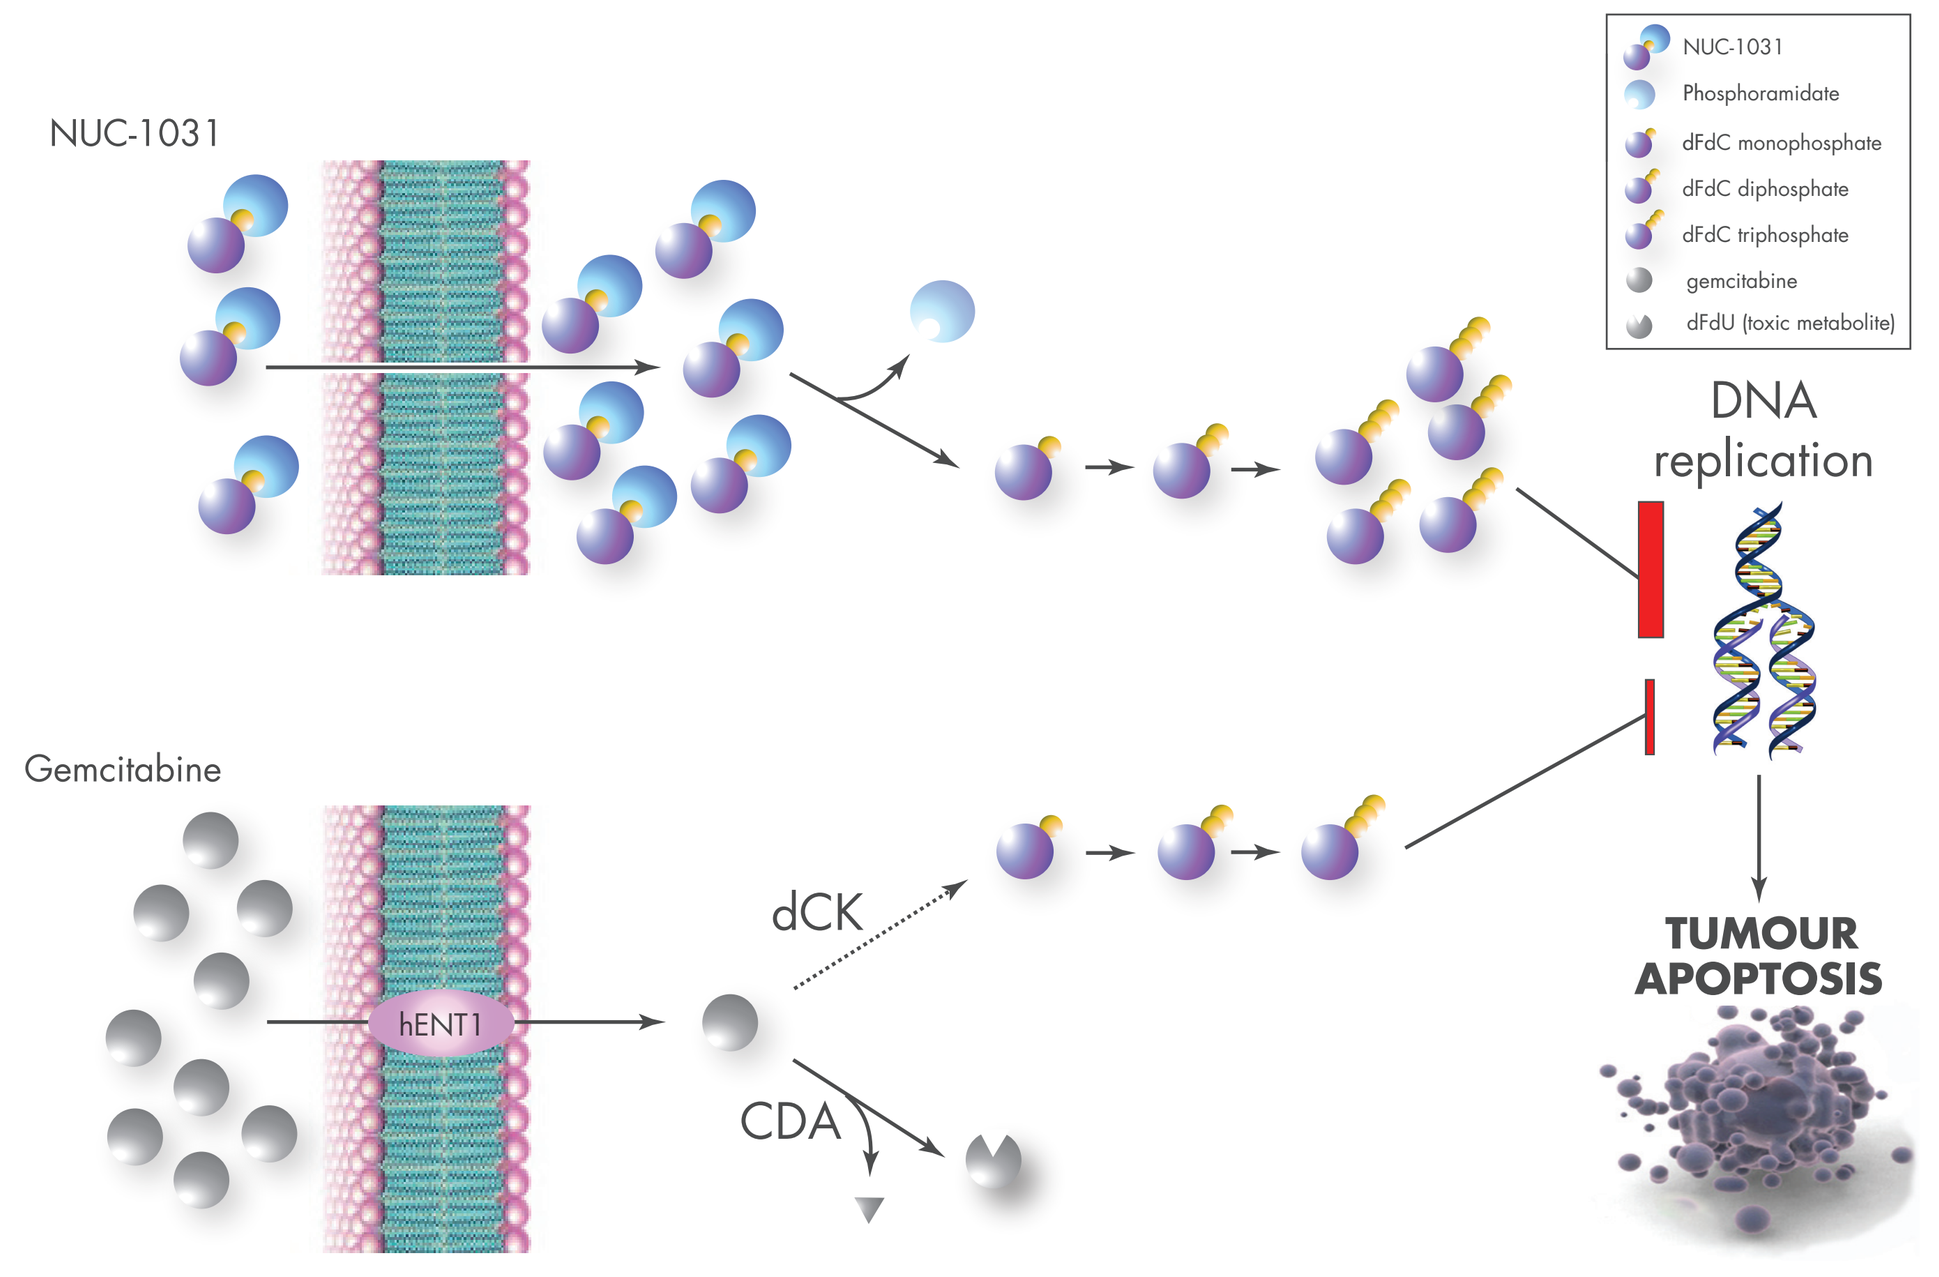
NUC-1031
NUC-1031
Phosphoramidate
dFdC monophosphate
dFdC diphosphate
dFdC triphosphate
gemcitabine
dFdU (toxic metabolite)
DNA replication
Gemcitabine
hENT1
dCK
CDA
TUMOUR APOPTOSIS

Figure 1. NUC-1031 bypasses key gemcitabine resistance pathways

STUDY DESIGN

Objectives

- Primary
 - o Determine recommended Phase II dose
 - o Assess safety profile
- Secondary
 - o Define PK and PD profiles
 - o Evaluate preliminary anti-tumour activity

Methods

- Sequential dose-escalating cohorts (3+3 design), with NUC-1031 administered as a 10 minute IV injection
- Schedule A: NUC-1031 administered on days 1, 8 & 15 of a 4 week cycle
- Schedule B: NUC-1031 administered on days 1, 5, 8, 12, 15, 19 of a 4 week cycle

Patient Population

- Patients aged ≥18 years with advanced, solid tumours relapsed/refractory to all standard treatments

RESULTS

Patients Characteristics

- 36 patients at submission date
- 26 female, 10 male
- Mean age 56 years (range 20-76)
- Average number of previous chemotherapy regimens 2.4 (range 1-6)
- Primary tumour sites: Ovary 10; Pancreas 6; Biliary 4; Breast 3; Colon 3; CUP 3; Mesothelioma 3; NSCL 2; Thymus 1; Renal 1

Pharmacokinetics

Plasma

- NUC-1031 plasma half life is more favourable than gemcitabine (7.3 hours versus 1.5 hours respectively)

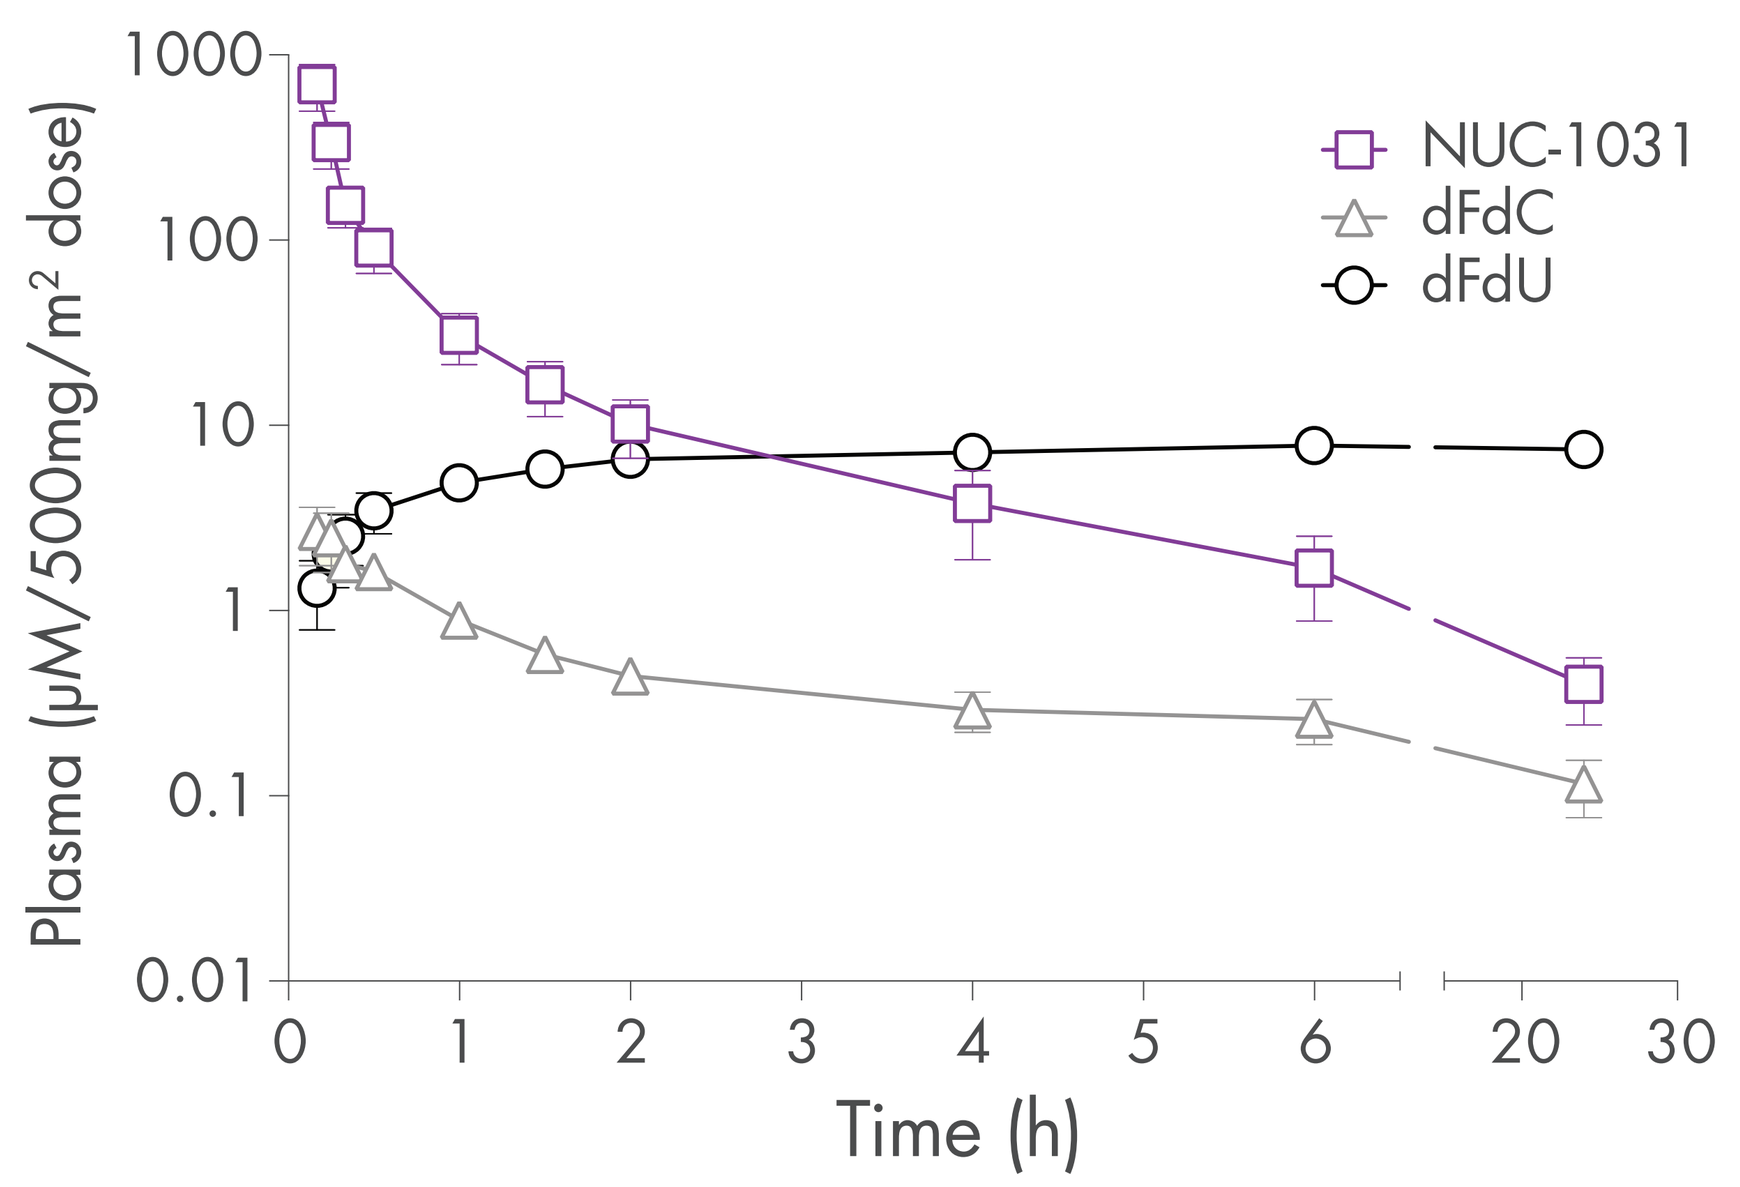
Plasma (μM/500mg/m² dose)
1000
100
10
1
0.1
0.01
NUC-1031
dFdC
dFdU
0
1
2
3
4
5
6
20
30
Time (h)

Figure 2. Plasma concentrations of NUC-1031, dFdC and dFdU

Intracellular dFdCTP

- C_{max} reached at 30 minutes end of injection
- Long half life: 12.1 hours
- Even at 24 hours NUC-1031 achieves levels of dFdCTP higher than reported for gemcitabine at its C_{max} at 2 hours

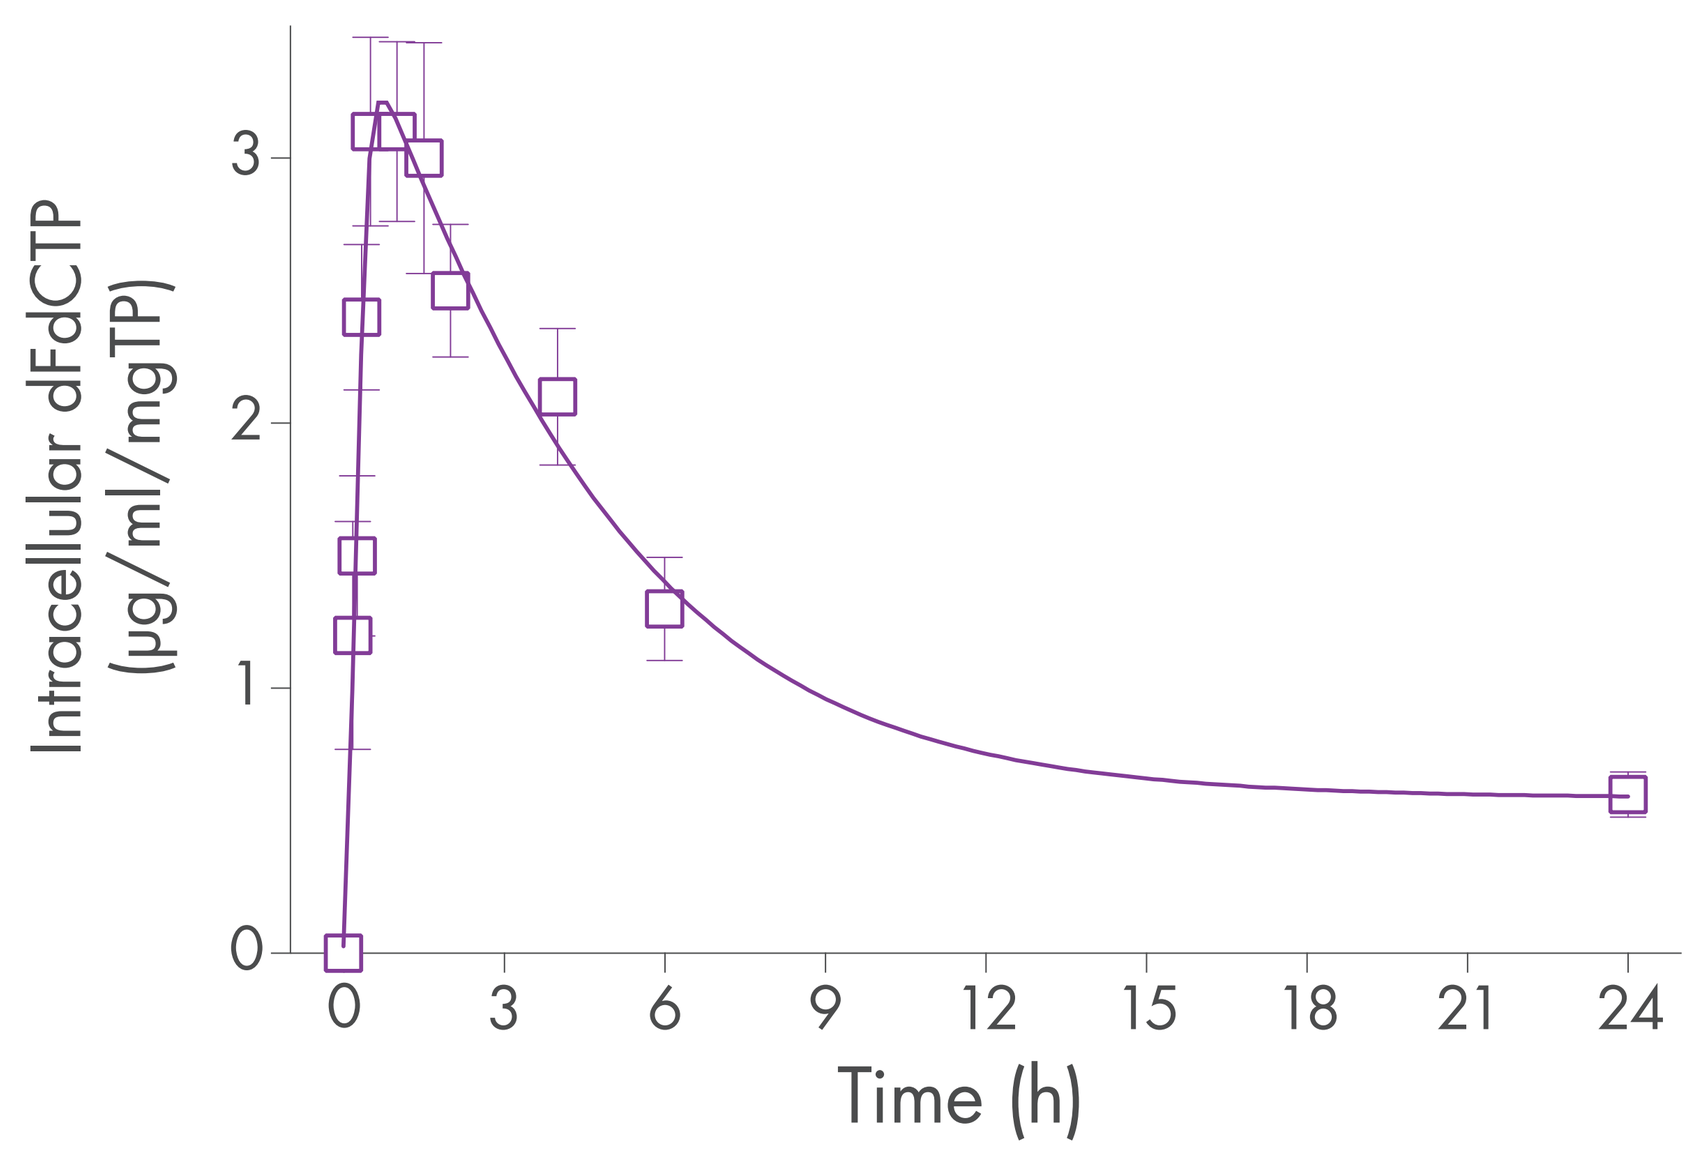
Intracellular dFdCTP (μg/ml/mgTP)
3
2
1
0
0
3
6
9
12
15
18
21
24
Time (h)

Figure 3. Intracellular concentrations (mean ±SD) of dFdCTP achieved by NUC-1031

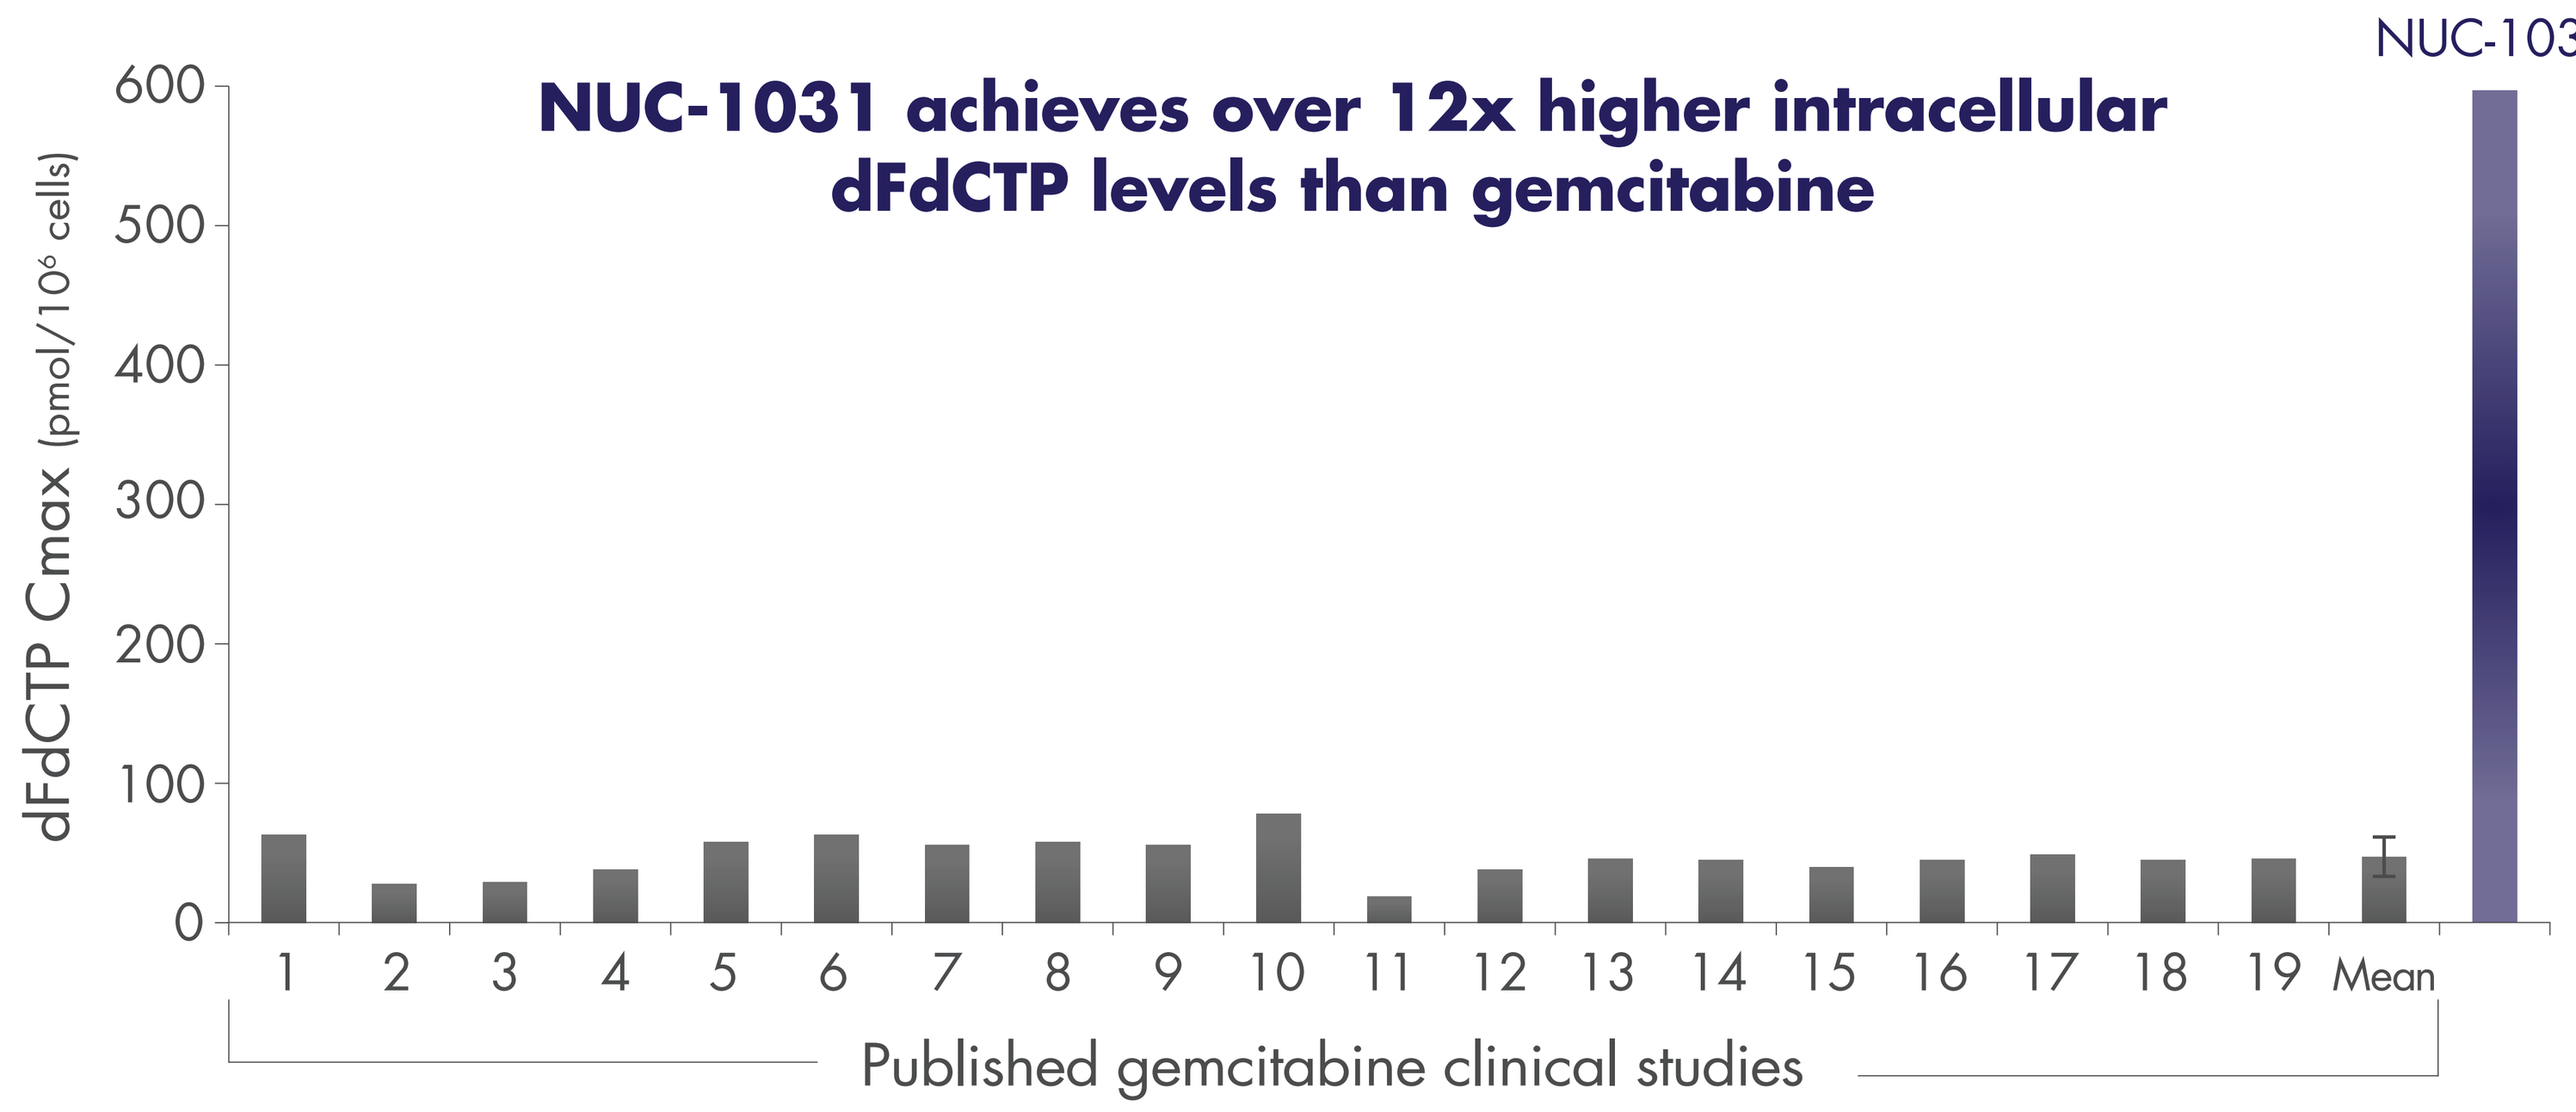
NUC-1031 achieves over 12x higher intracellular dFdCTP levels than gemcitabine
NUC-1031
dFdCTP Cmax (pmol/10⁶ cells)
600
500
400
300
200
100
0
1
2
3
4
5
6
7
8
9
10
11
12
13
14
15
16
17
18
19
Mean
Published gemcitabine clinical studies

Patient Safety

- No unexpected Adverse Events (AEs)
- Most common AEs* Grade 1 or 2 were: anaemia (67%); fatigue (67%); transaminitis (64%); thrombocytopaenia (53%)
- 14 Serious Adverse Events*
- Only 3 patients had Grade 4 AEs: thrombocytopaenia; hypomagnesaemia; sepsis + dyspnoea*
- 2 DLTs were observed:
 - o Grade 3 elevated ALT (725 mg/m²)
 - o Grade 4 thrombocytopaenia (750 mg/m²)

AEs Grade 3 or 4 occurring in ≥ 5% patients*

	Schedule A							Sch.B
	500 mg/m²	625 mg/m²	675 mg/m²	725 mg/m²	750 mg/m²	825 mg/m²	1000 mg/m²	375 mg/m²
Neutropaenia				1	4		1	1
Thrombocytopaenia	1				2			1
Leucopaenia				1				2
Anaemia				1	1			
Decreased WBC				1	1			1
Transaminitis				1				4
Hypomagnesaemia		1			1			
Lung infection					1			1
Dyspnoea	1				1			

*Considered definitely, probably or possibly related to NUC-1031

Efficacy

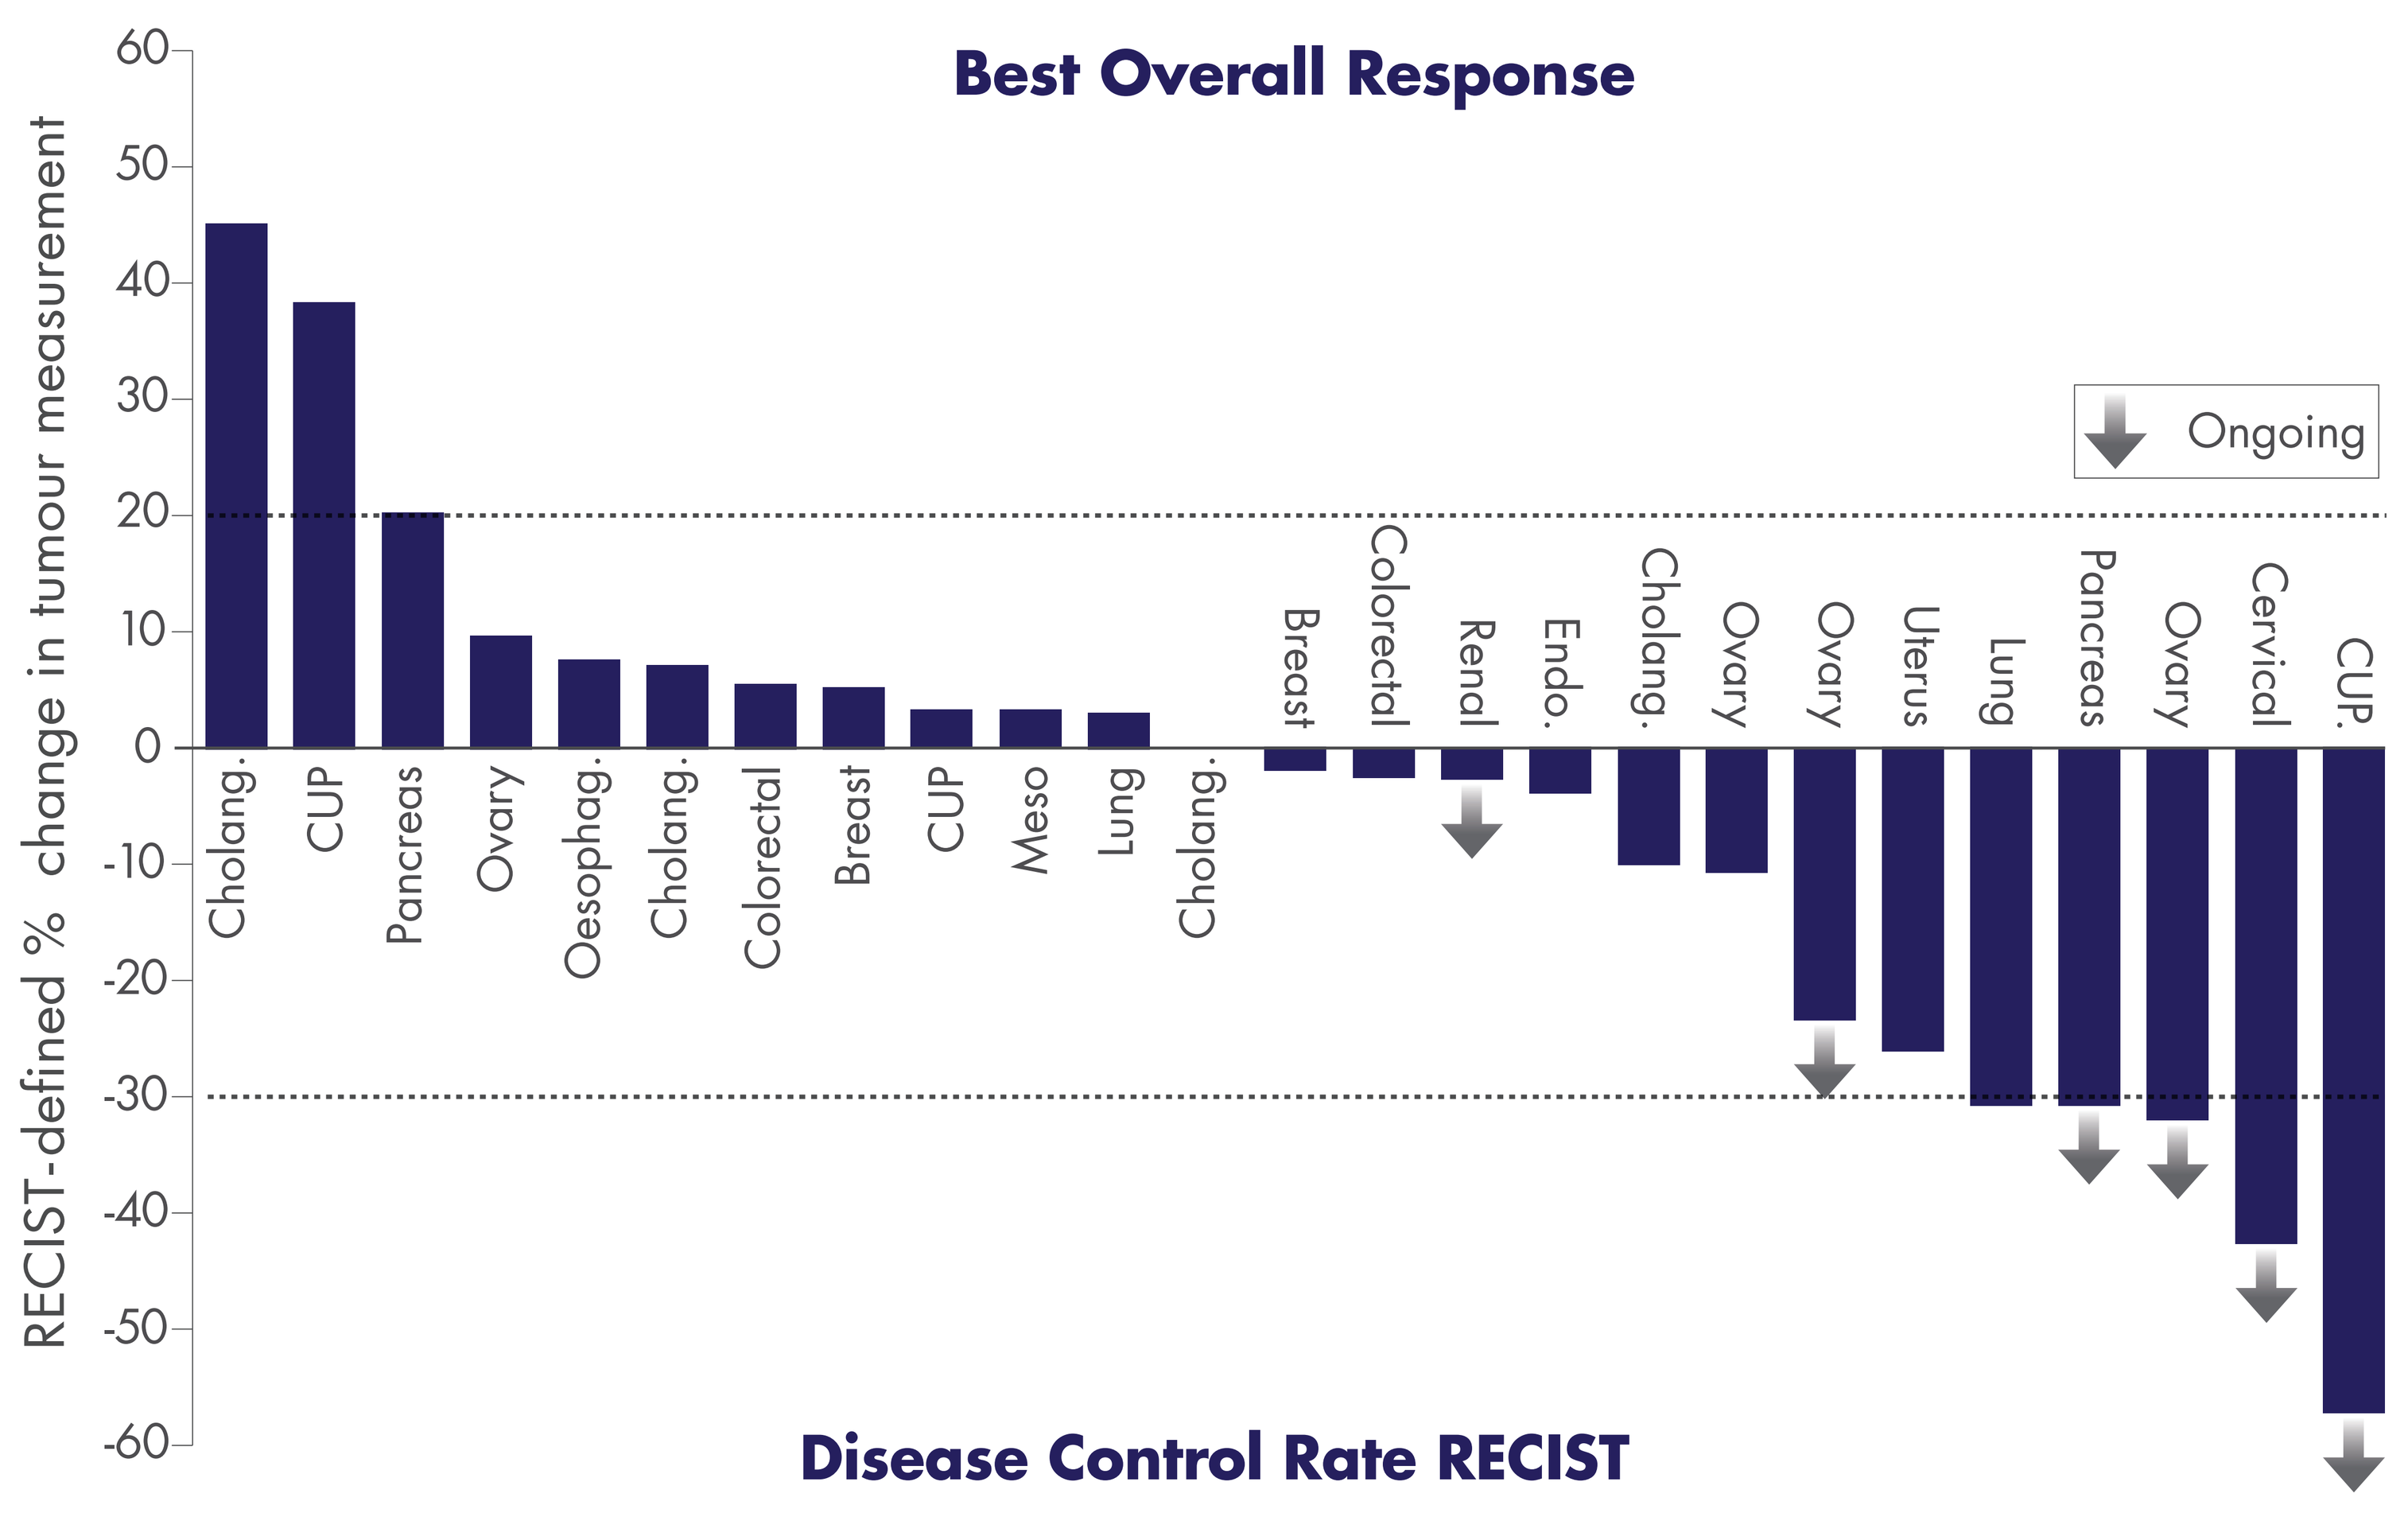
Best Overall Response
RECIST-defined % change in tumour measurement
60
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
Ongoing
Cholang.
CUP
Pancreas
Ovary
Oesophag.
Cholang.
Colorectal
Breast
CUP
Meso
Lung
Cholang.
Breast
Colorectal
Renal
Endo.
Cholang.
Ovary
Ovary
Uterus
Lung
Pancreas
Ovary
Cervical
CUP.

Disease Control Rate RECIST

	All Patients		Evaluable Patients	
	n	%	n	%
Total	36	100	25	100
Partial Response	5	14	5	20
Stable Disease	17	47	17	68
Disease Control	22	**61**	22	**88**

Evaluable patients ≥ 2 Cycles of NUC-1031

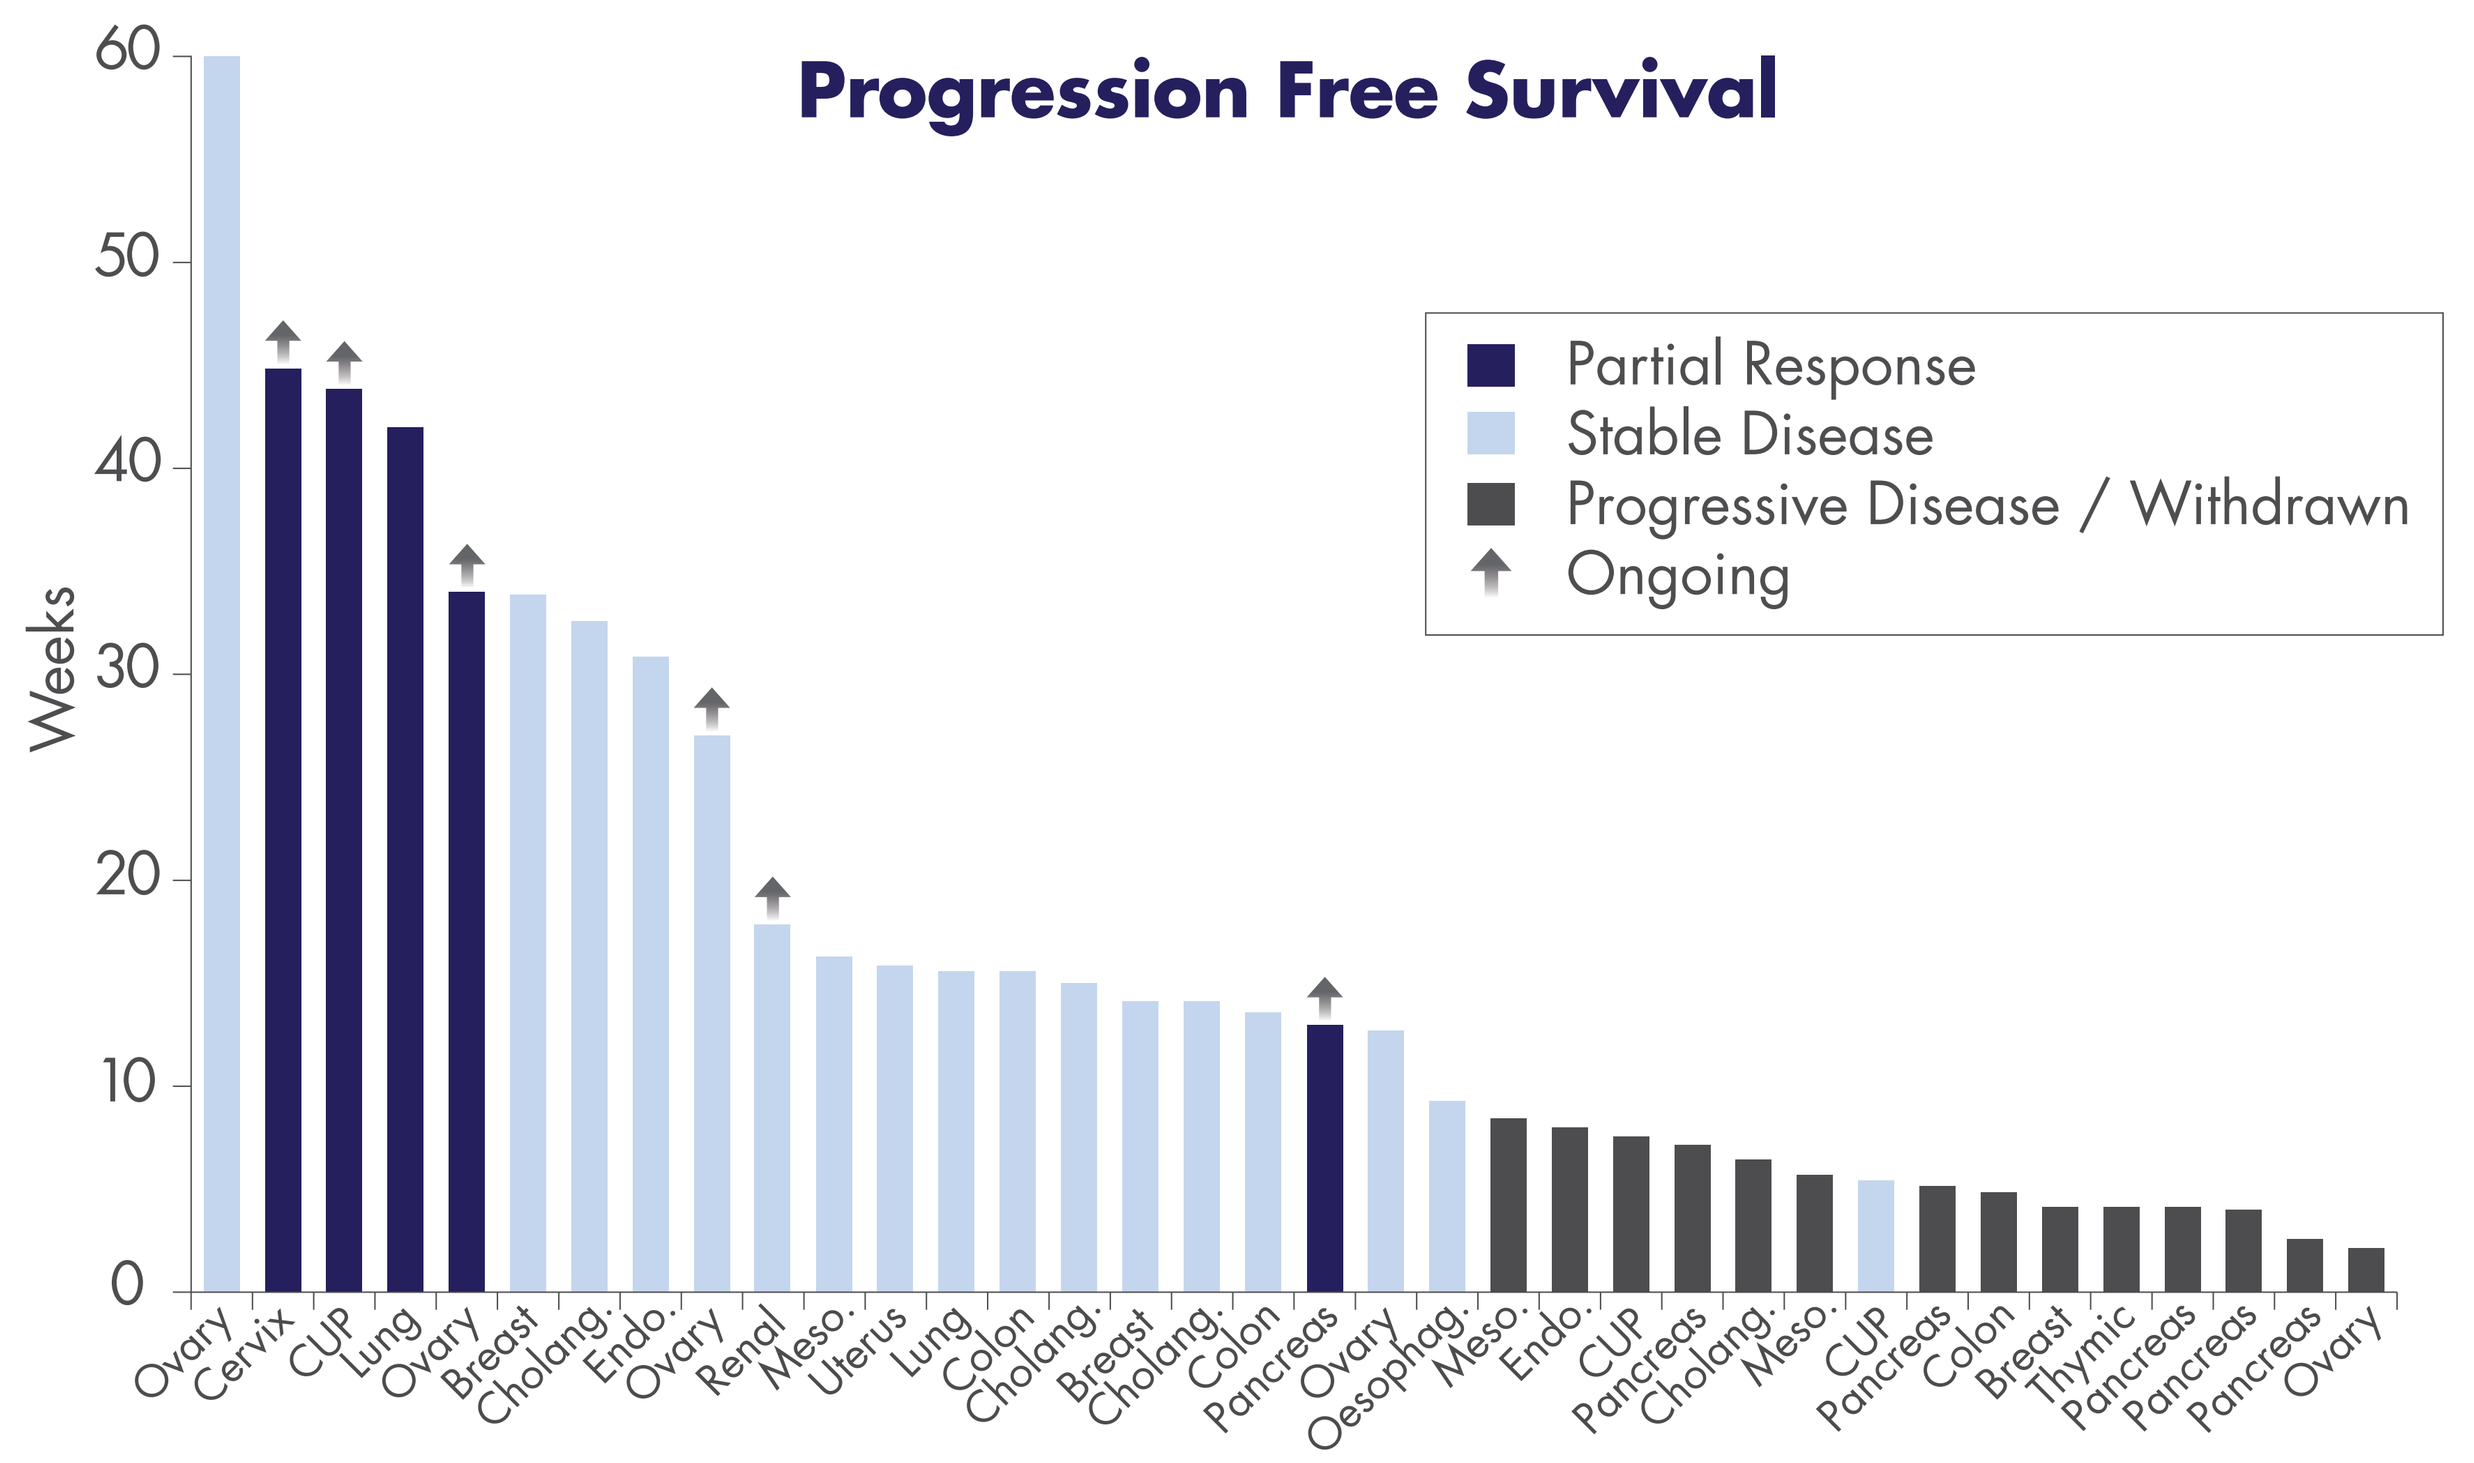
Progression Free Survival
Weeks
60
50
40
30
20
10
0
Partial Response
Stable Disease
Progressive Disease / Withdrawn
Ongoing
Ovary
Cervix
CUP
Lung
Ovary
Breast
Cholang.
Endo.
Ovary
Renal
Meso.
Uterus
Lung
Colon
Cholang.
Breast
Cholang.
Colon
Pancreas
Ovary
Oesophag.
Meso.
Endo.
CUP
Pancreas
Cholang.
Meso.
CUP
Pancreas
Colon
Breast
Thymic
Pancreas
Pancreas
Pancreas
Ovary

Patient Case Studies

Biliary

Female, 48 years, cholangiocarcinoma with multi-site metastases.
Refractory to gemcitabine + cisplatin.
Stable Disease on NUC-1031 with 10% reduction in tumour volume.
PFS = 8 months.

CUP

Male, 54 years, unknown primary with lung and liver metastases.
Progressed on epirubicin + cisplatin + capecitabine.
Partial Response on NUC-1031, with 58% reduction in tumour volume.
PFS = 11 months ongoing.

Ovary

Female, 61 years, ovarian adenocarcinoma with multi-site metastases.
Several prior lines of chemotherapy and relapsed within 1 year of carboplatin regimen.
Partial Response on NUC-1031 with >30% reduction in tumour volume and 91% reduction in CA125 level.
PFS = 8 months ongoing.

Pancreas

Female, 69 years, pancreatic cancer with liver metastases.
Progressed on gemcitabine.
Partial Response on NUC-1031, with 30% reduction in tumour volume, 92% reduction in CEA level and 73% reduction in CA19.9 level.
PFS = 3 months ongoing.

CONCLUSIONS

NUC-1031

- Impressive and durable disease control in a high proportion of evaluable patients
- Active in a broad range of cancers
- Disease control in patients refractory to/relapsed on prior chemotherapy, including gemcitabine
- Well tolerated with no unexpected AEs
- Generates high intracellular levels of the active agent dFdCTP
- Overcomes key cancer resistance mechanisms
- Molecular profiling studies ongoing to characterise target population
- Phase III global studies planned in pancreatic, biliary, ovarian and NSCL cancers

ASCO 2014

Imperial College acknowledges support from NIHR/Cancer Research UK Experimental Cancer Medicine Centre and NHS funding to their NIHR Biomedical Research Centre.