NUC-3373 is a potent TS inhibitor and induces DNA damage in NSCLC cancer cells regardless of histological subtype

Boback Kaghazchi[1], Alison L. Dickson[1,2], Greice Zickuhr[1], Oliver J. Read[1,2], David J. Harrison[1,2], Jennifer Bré[1,2]

1) University of St Andrews, UK
2) NuCana plc, UK

Abstract Number: 185 **Email:** bk68@st-andrews.ac.uk

BACKGROUND

- Lung 2nd most common cancer
- Incidence: 2.2 million
- Leading cause of cancer mortality[1]
- NSCLC accounts for ~85% cases of lung cancer, majority are adeno-or squamous carcinoma
 - Systemic treatments include immune checkpoint inhibitors, chemotherapy, and targeted therapies

Thymidylate Synthase (TS) inhibitors in NSCLC

- 5-FU has limited clinical utility due to high tumor expression of DPD[2]
- Pemetrexed is a multi-targeted anti-folate that inhibits TS
 - Used in adenocarcinoma but not recommended in squamous subtypes due to high basal TS expression[3]
 - Upregulates PD-L1 and potentiates T-cell-mediated cytotoxicity when combined with anti-PD-L1 therapy[4]

NUC-3373: A potent TS inhibitor with additional DNA-mediated mode of action

- ProTide transformation of FUDR-MP[5,6,] active anti-cancer metabolite of 5-FU
 - Resistant to breakdown by DPD
 - Low levels of toxic metabolites (FBAL, FUTP)[7,8]
 - Generates high intracellular levels of FUDR-MP resulting in potent TS inhibition and DNA damage[8]

Aim

- Investigate NUC-3373-mediated TS inhibition and effect in combination with a PD-L1 inhibitor

Hypotheses

- NUC-3373 inhibits TS and causes DNA damage in NSCLC cells, regardless of histological subtype
- NUC-3373 upregulates PD-L1 promoting immunogenic cell death

NUC-3373: Overcomes limitations associated with 5-FU

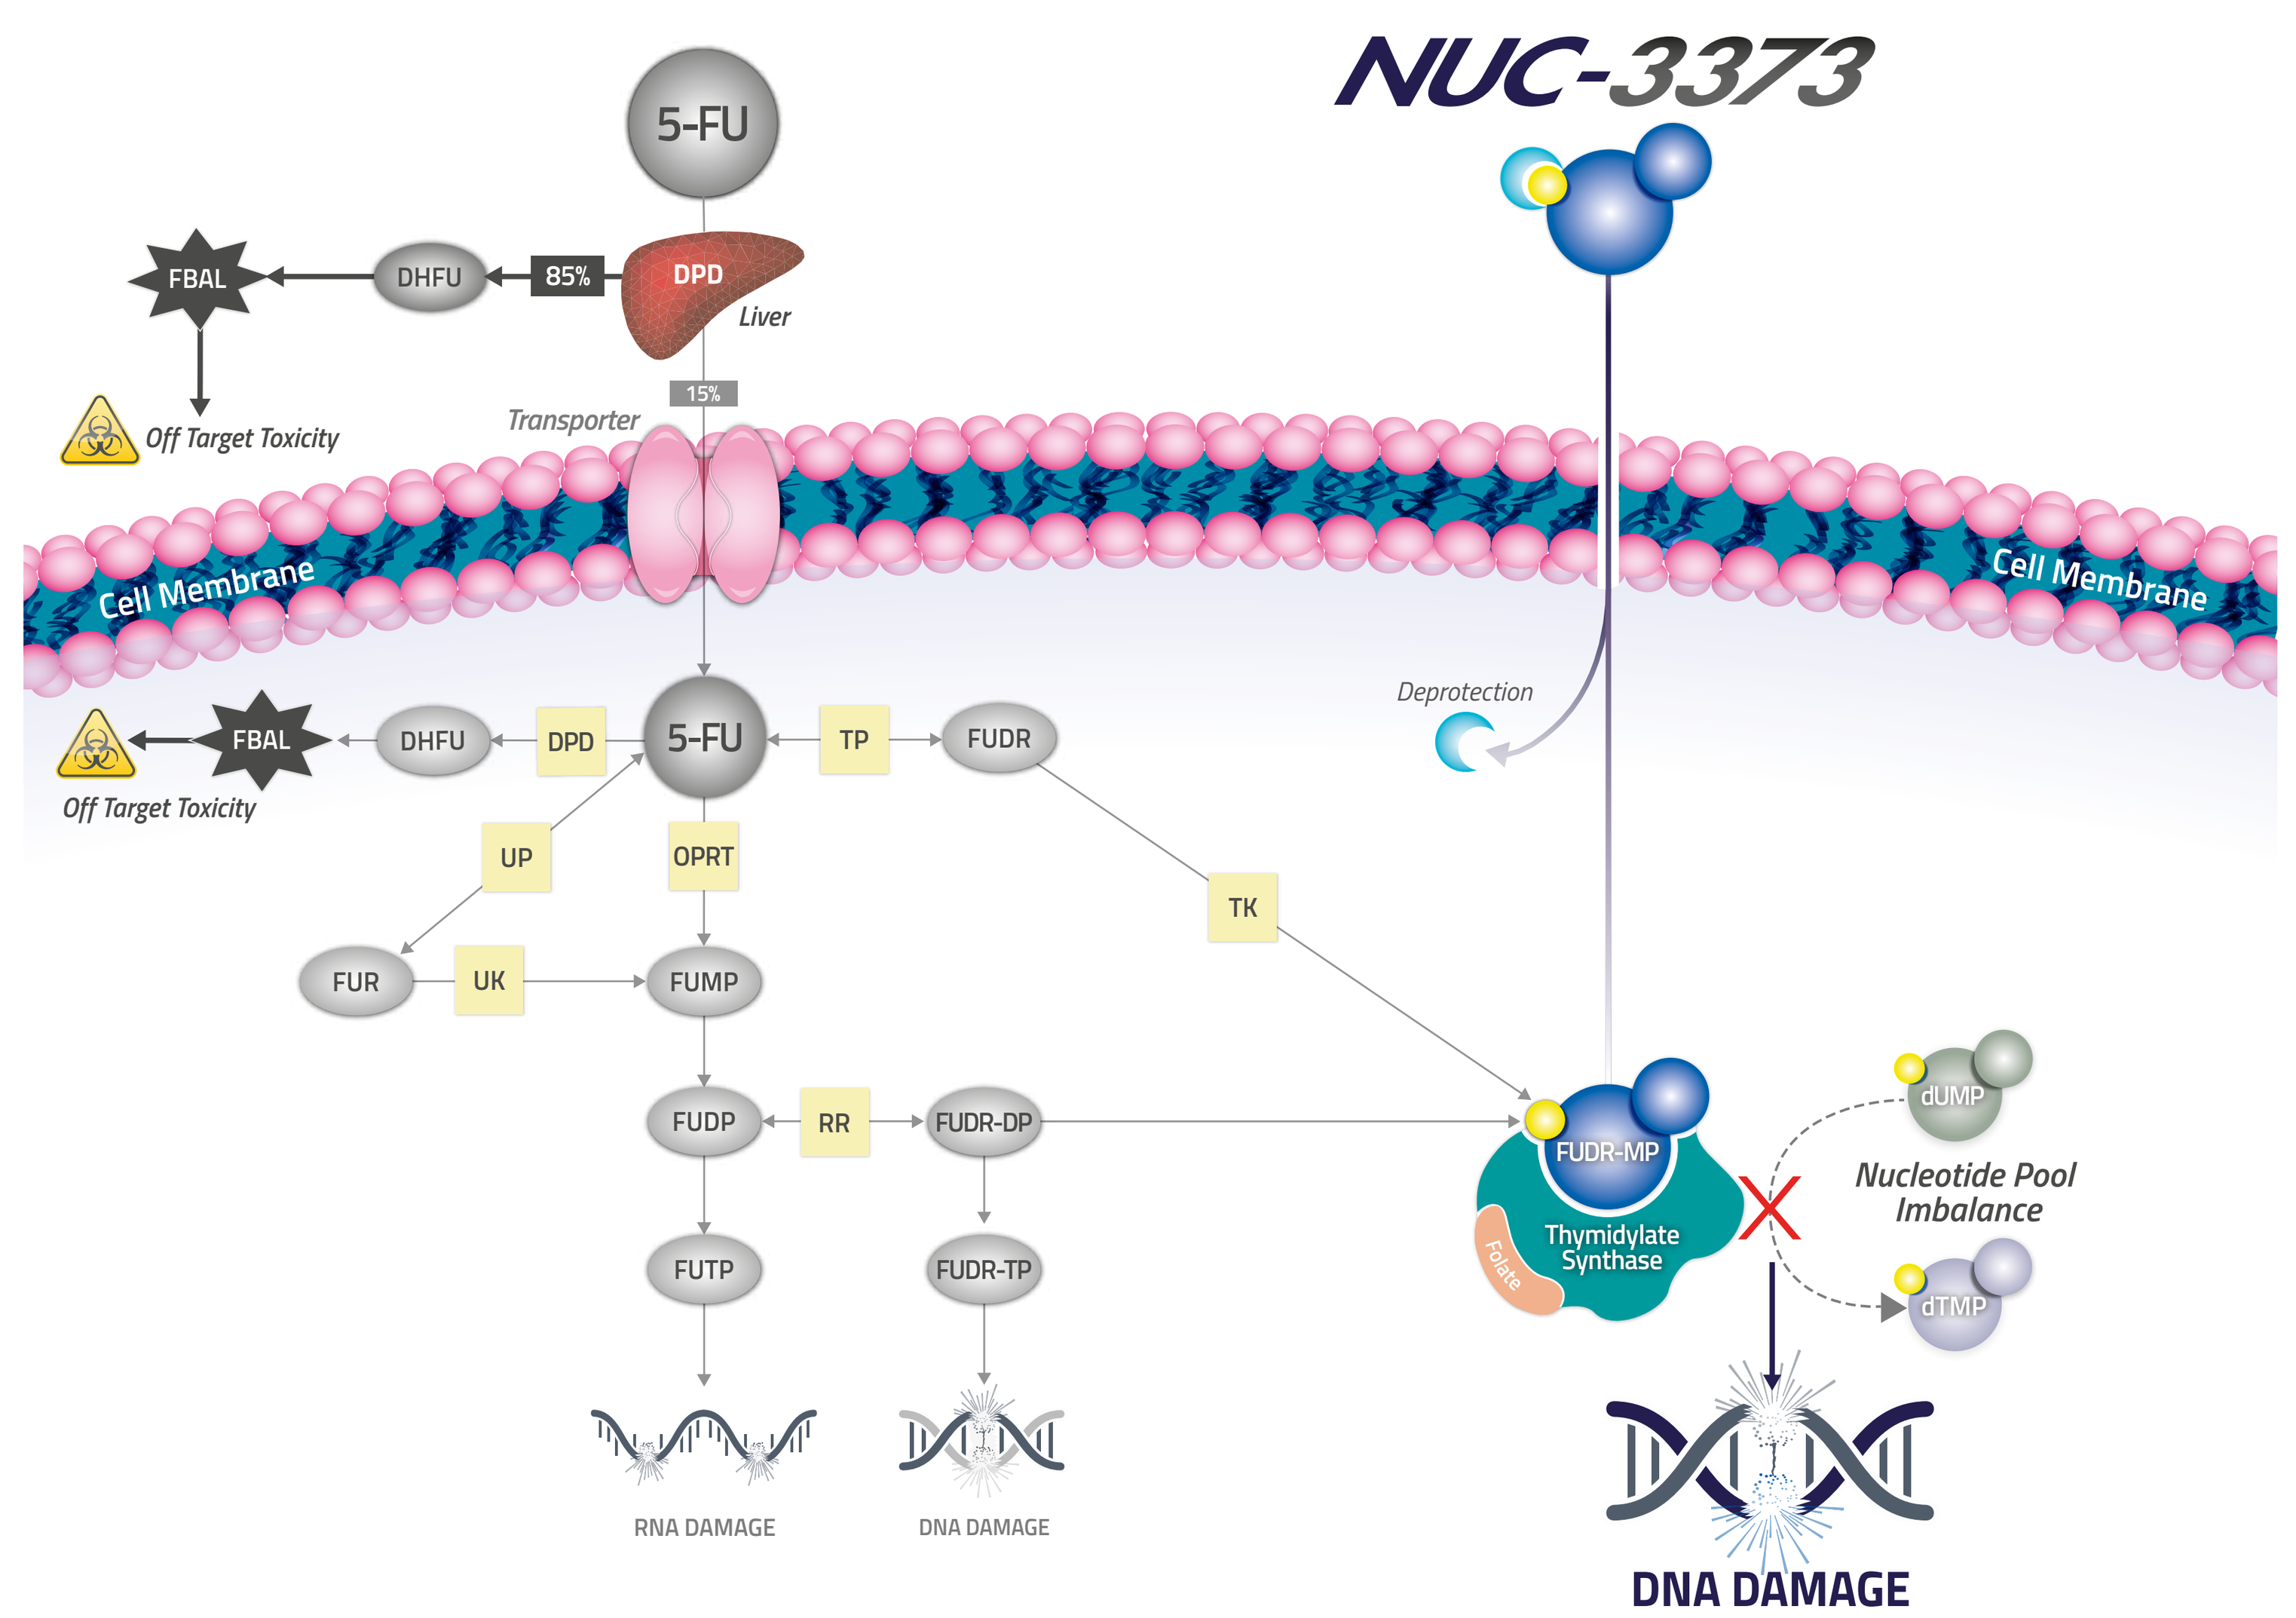
NUC-3373
5-FU
FBAL
DHFU
85%
DPD
Liver
15%
Off Target Toxicity
Transporter
Cell Membrane
Cell Membrane
Deprotection
FBAL
DHFU
DPD
5-FU
TP
FUDR
Off Target Toxicity
UP
OPRT
TK
FUR
UK
FUMP
FUDP
RR
FUDR-DP
FUDR-MP
Thymidylate Synthase
Folate
dUMP
Nucleotide Pool Imbalance
dTMP
FUTP
FUDR-TP
RNA DAMAGE
DNA DAMAGE
DNA DAMAGE

METHODS

Cell culture: Adenocarcinoma (A549) and squamous (Nx002) NSCLC cell lines were treated with 1 or 10 μM NUC-3373 (sub-IC_{50}), pemetrexed or 5-FU for 6, 24 or 48 hours. Nx002 cells were treated with 10 μM NUC-3373 for 24 hours prior to co-culture with PBMCs to test T-cell mediated cell killing modulated by NUC-3373 ± 10 μg/ml pembrolizumab. Cell confluence assessed by automated cytometry analysis (Celigo).

TS ternary complexes: Free TS and TS ternary complex formation assessed by Western blot.

Metabolites: FUDR-MP, FdUr and dUMP levels determined by LC-MS/MS (LLOQs: FUDR-MP=5 nM, dUMP=0.2 nM, FdUr=0.1 nM).

DNA damage: DNA damage assessed by immunofluorescence using γ–H2AX and p-Chk1.

PD-L1: NSCLC cell surface expression measured by flow cytometry.

(for all experiments, n=3)

RESULTS

NUC-3373 generates high levels of FUDR-MP & forms TS ternary complexes

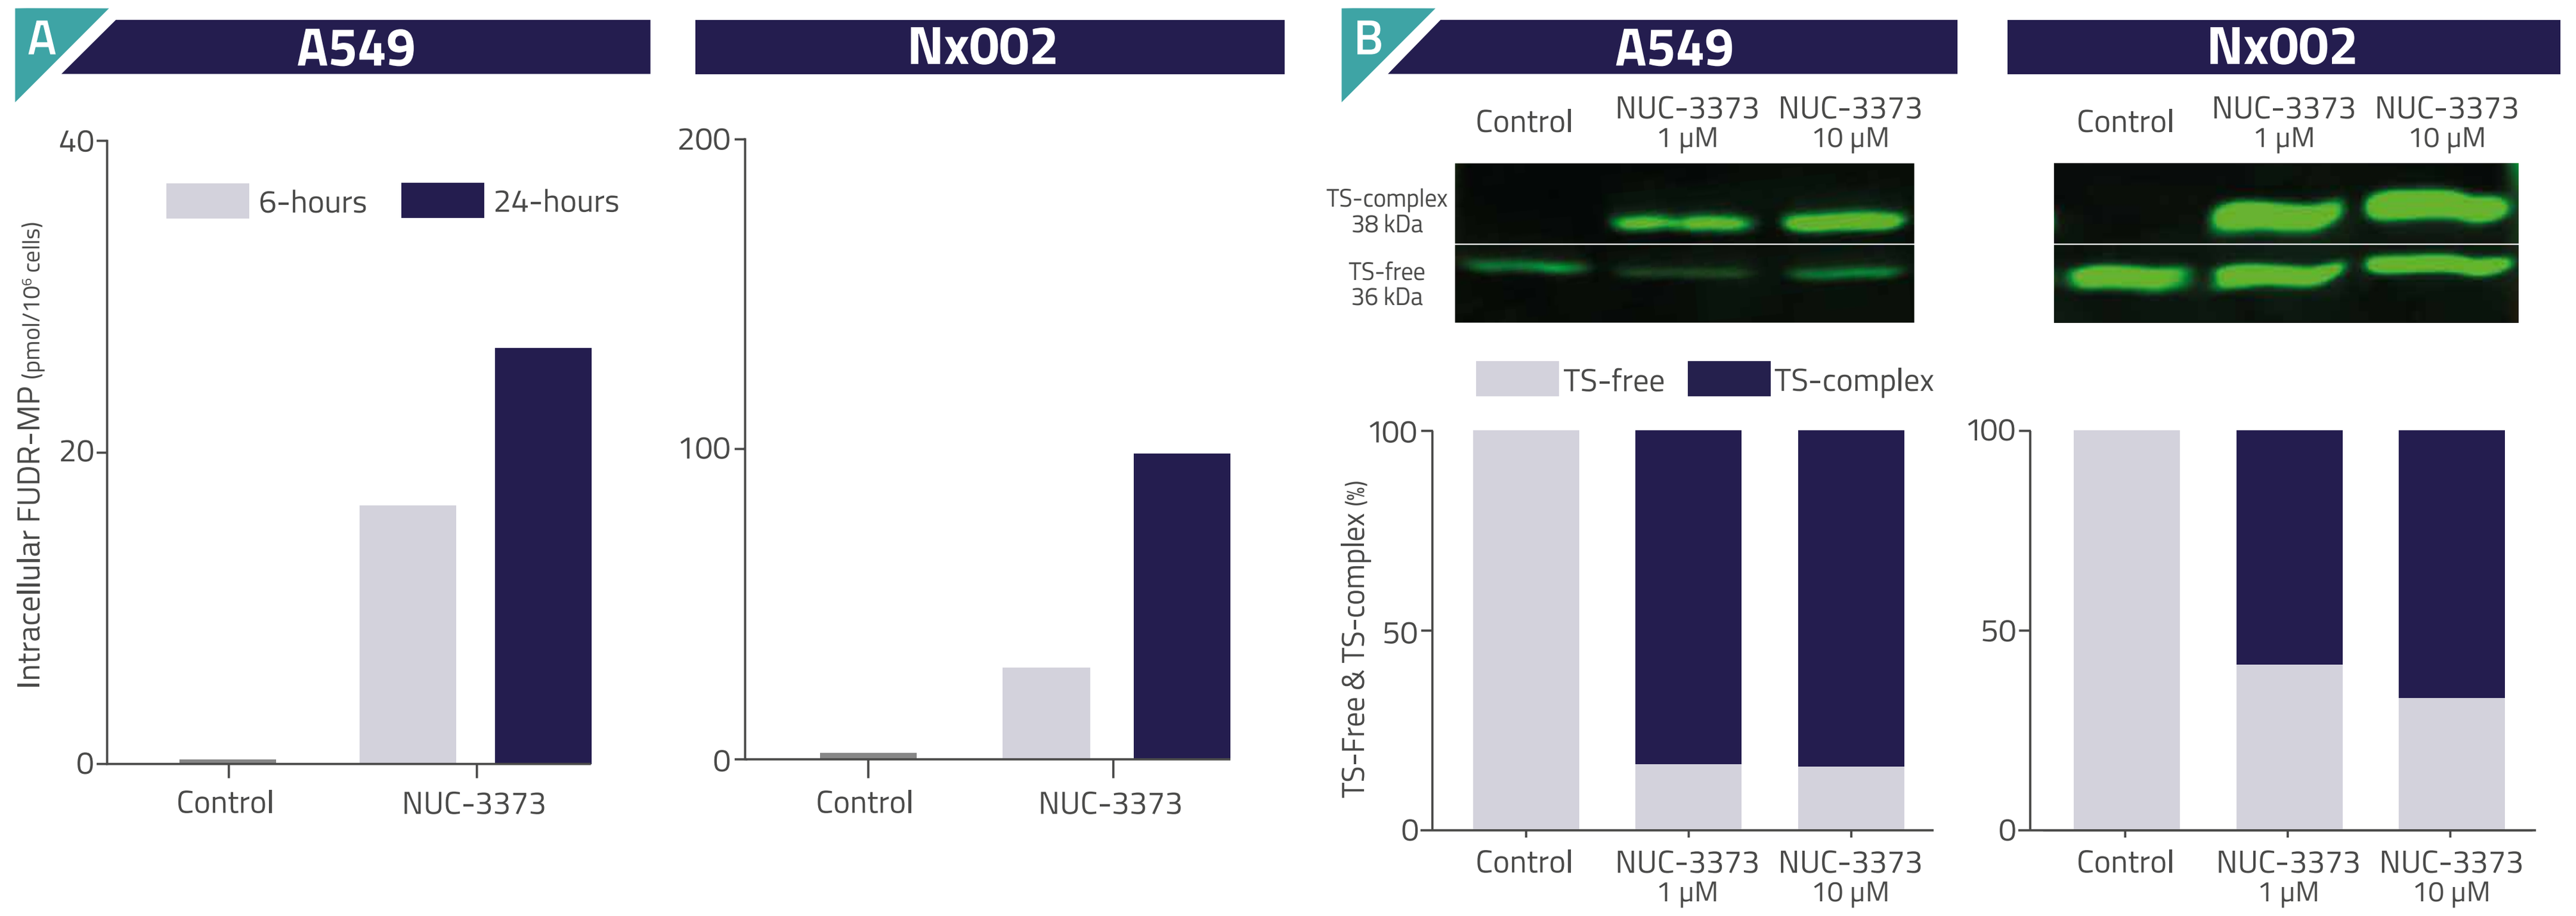
A
A549
Nx002
6-hours
24-hours
Intracellular FUDR-MP (pmol/10⁶ cells)
Control
NUC-3373
B
A549
Nx002
Control
NUC-3373 1 μM
NUC-3373 10 μM
TS-complex 38 kDa
TS-free 36 kDa
TS-free
TS-complex
TS-Free & TS-complex (%)

Figure 2. (A) FUDR-MP levels in NSCLC cells treated with 10 μM NUC-3373. (B) TS and TS ternary complex protein expression in NSCLC cells treated with 1 μM and10 μM NUC-3373 for 24 hours.

- NUC-3373 generates high intracellular levels of active anti-cancer metabolite FUDR-MP
- NUC-3373 promotes formation of TS ternary complexes (60-84% of total TS) in NSCLC cells
 - Ternary complex formation occurs in adenocarcinoma and squamous subtypes

NUC-3373 is a more potent TS inhibitor than pemetrexed & 5-FU

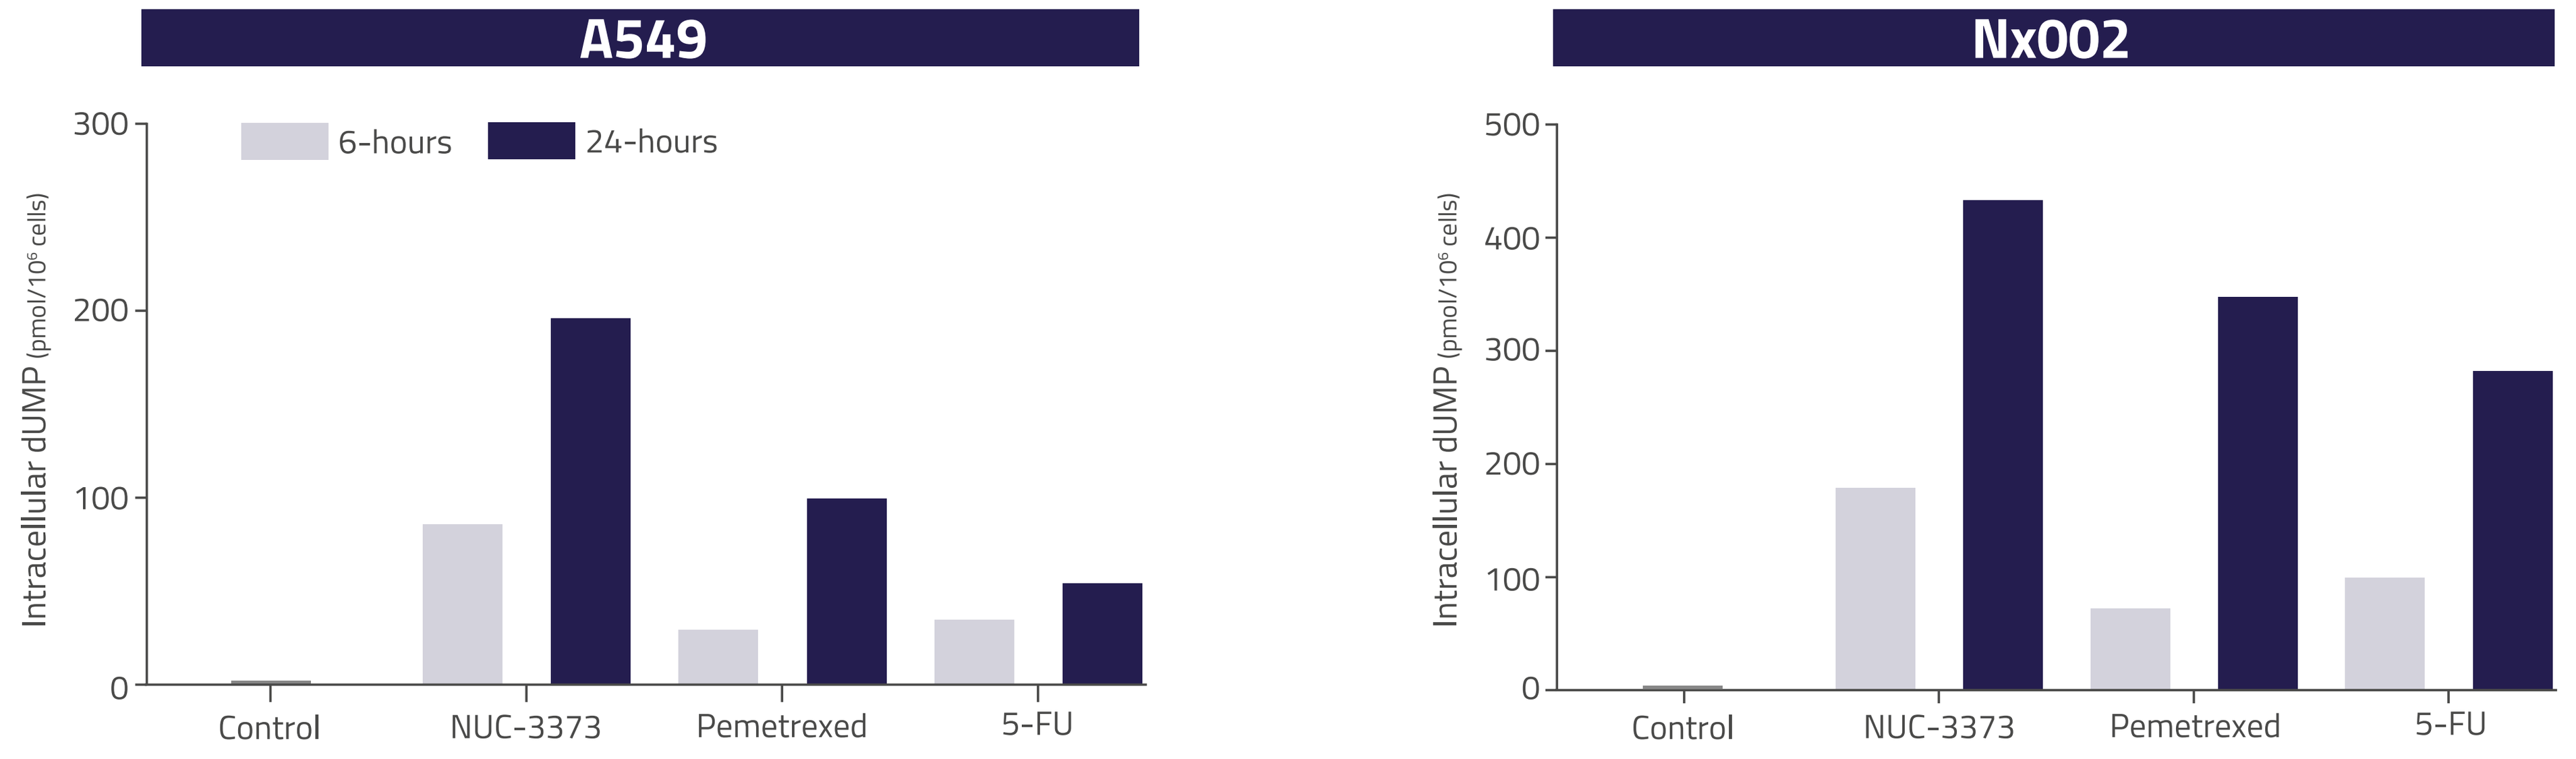
A549
Nx002
6-hours
24-hours
Intracellular dUMP (pmol/10⁶ cells)
Control
NUC-3373
Pemetrexed
5-FU

Figure 3: dUMP levels (surrogate of TS inhibition) in NSCLC cells treated with 10 μM NUC-3373, pemetrexed or 5-FU for 6 or 24 hours.

- NUC-3373 treatment results in greater levels of dUMP than pemetrexed or 5-FU treatment
 - Indicative of more effective TS inhibition
 - TS inhibition evident in adenocarcinoma and squamous subtypes

NUC-3373 is incorporated into DNA causing additional DNA damage

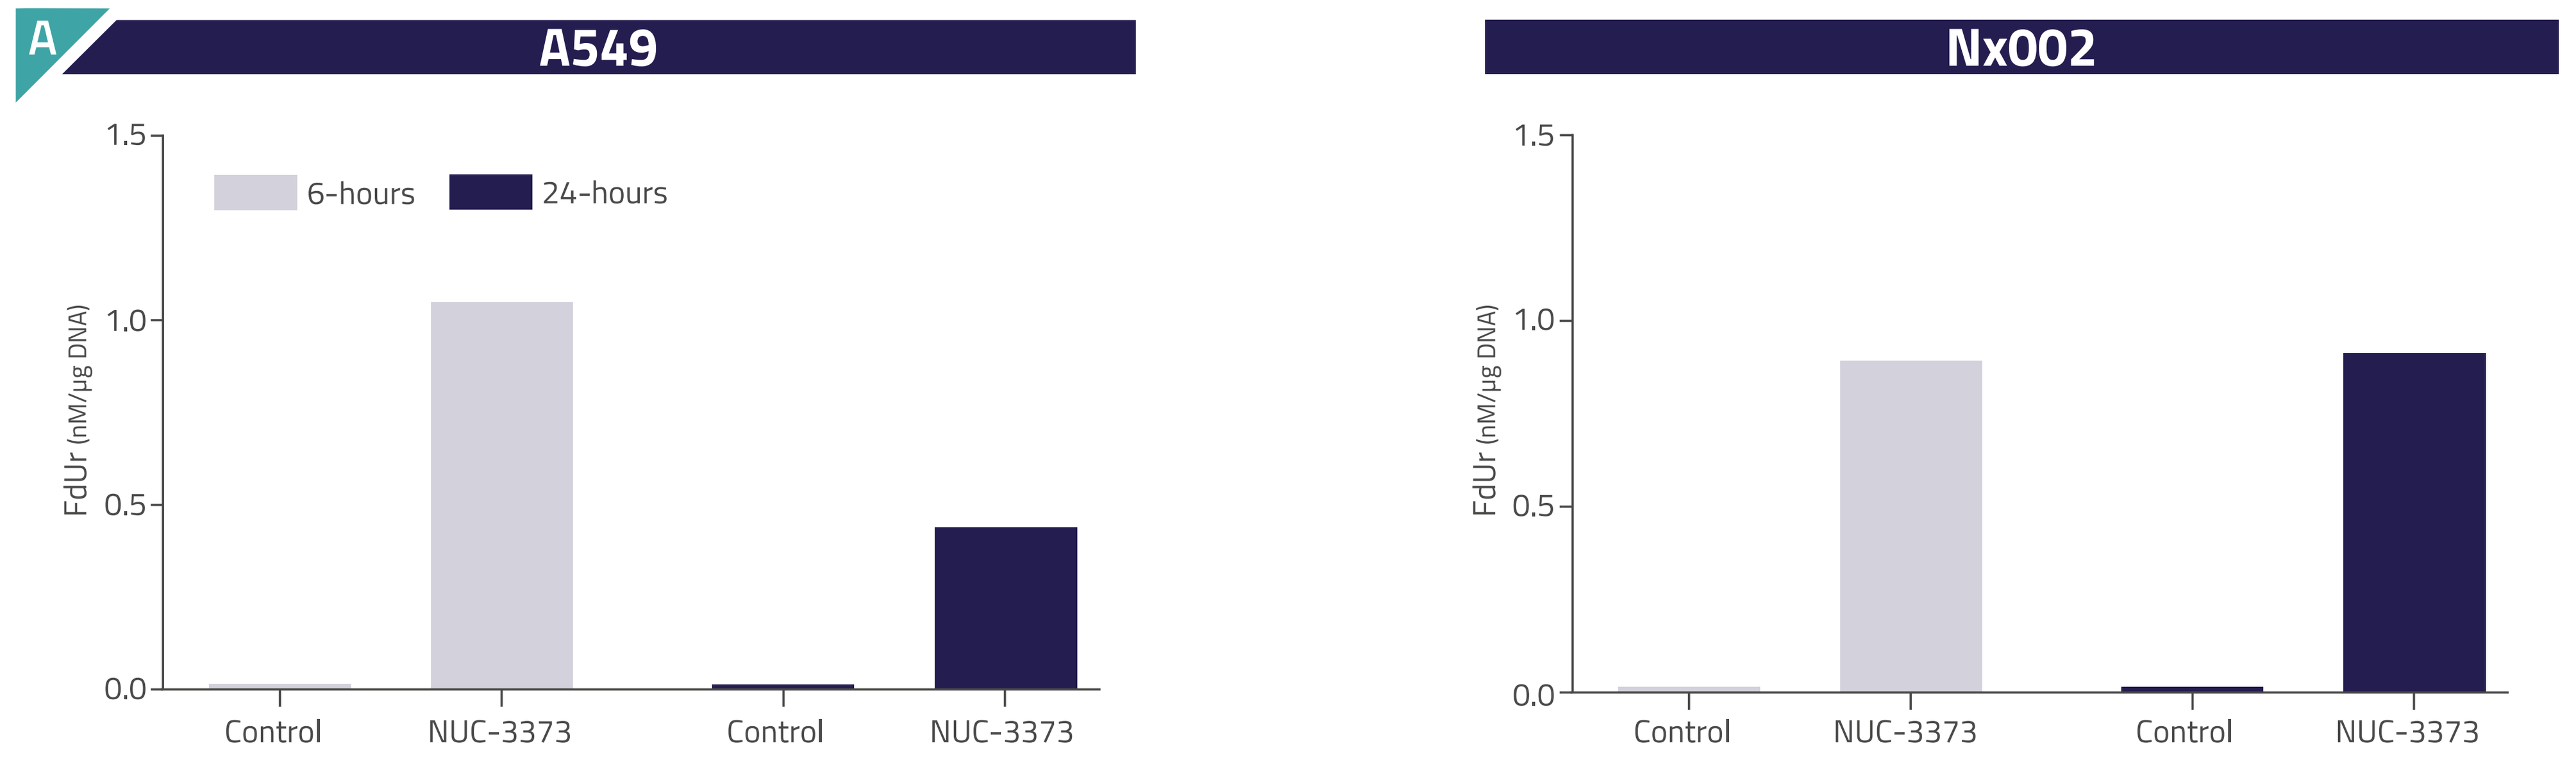
A
A549
Nx002
6-hours
24-hours
FdUr (nM/μg DNA)
Control
NUC-3373
Control
NUC-3373

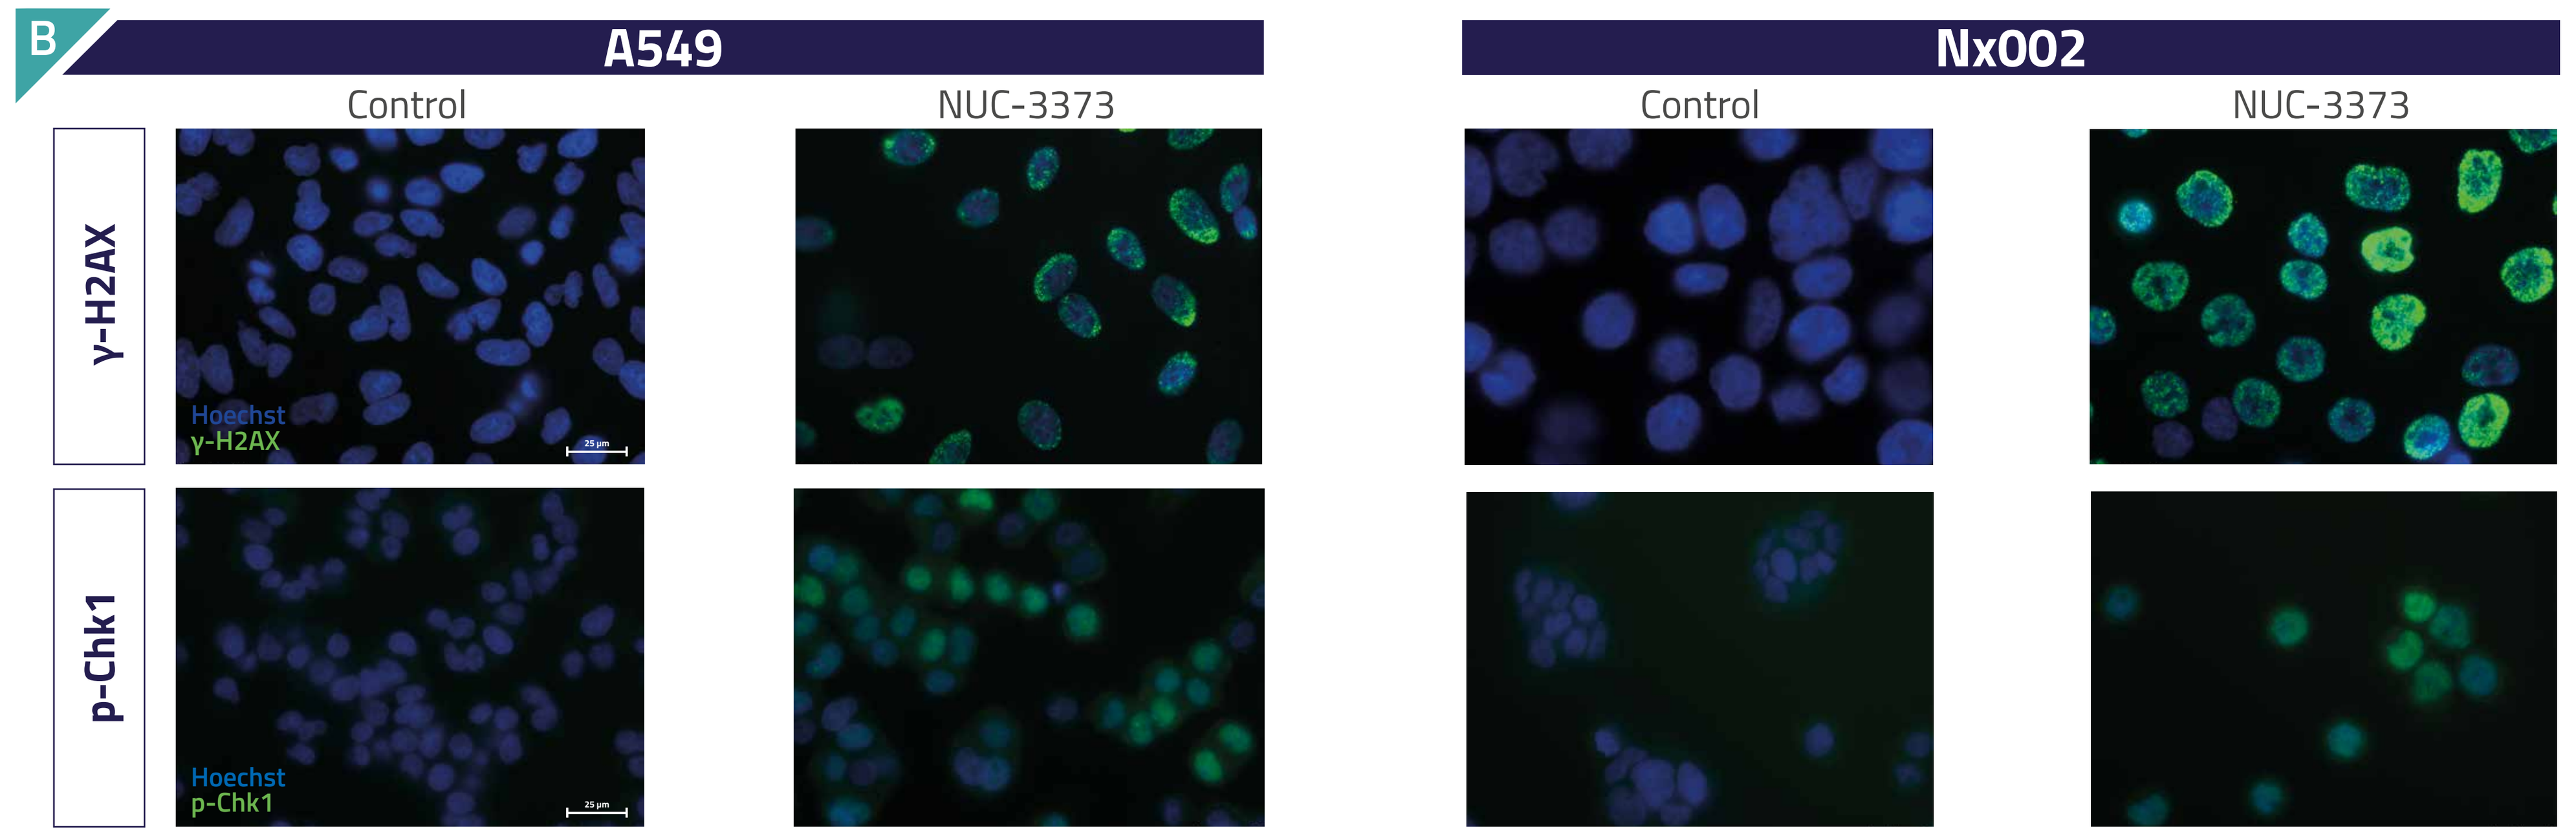
B
A549
Nx002
Control
NUC-3373
γ-H2AX
p-Chk1
Hoechst
γ-H2AX
Hoechst
p-Chk1

Figure 4. (A) FdUr levels (surrogate of FdUTP incorporation into DNA) in NSCLC cells treated with 10 μM NUC-3373 for 24 or 48 hours. (B) γ-H2AX & p-Chk1 expression in NSCLC cells treated with 10 μM NUC-3373 for 24 hours.

- NUC-3373 generates FUDR-TP (FdUTP) which is incorporated into DNA
 - DNA incorporation evident in both adenocarcinoma and squamous subtypes
- NUC-3373 causes DNA damage
 - Confirming a DNA-mediated mode of action in adenocarcinoma and squamous NSCLC cells

NUC-3373 increases PD-L1, potentiating T-cell-mediated cytotoxicity when combined with pembrolizumab

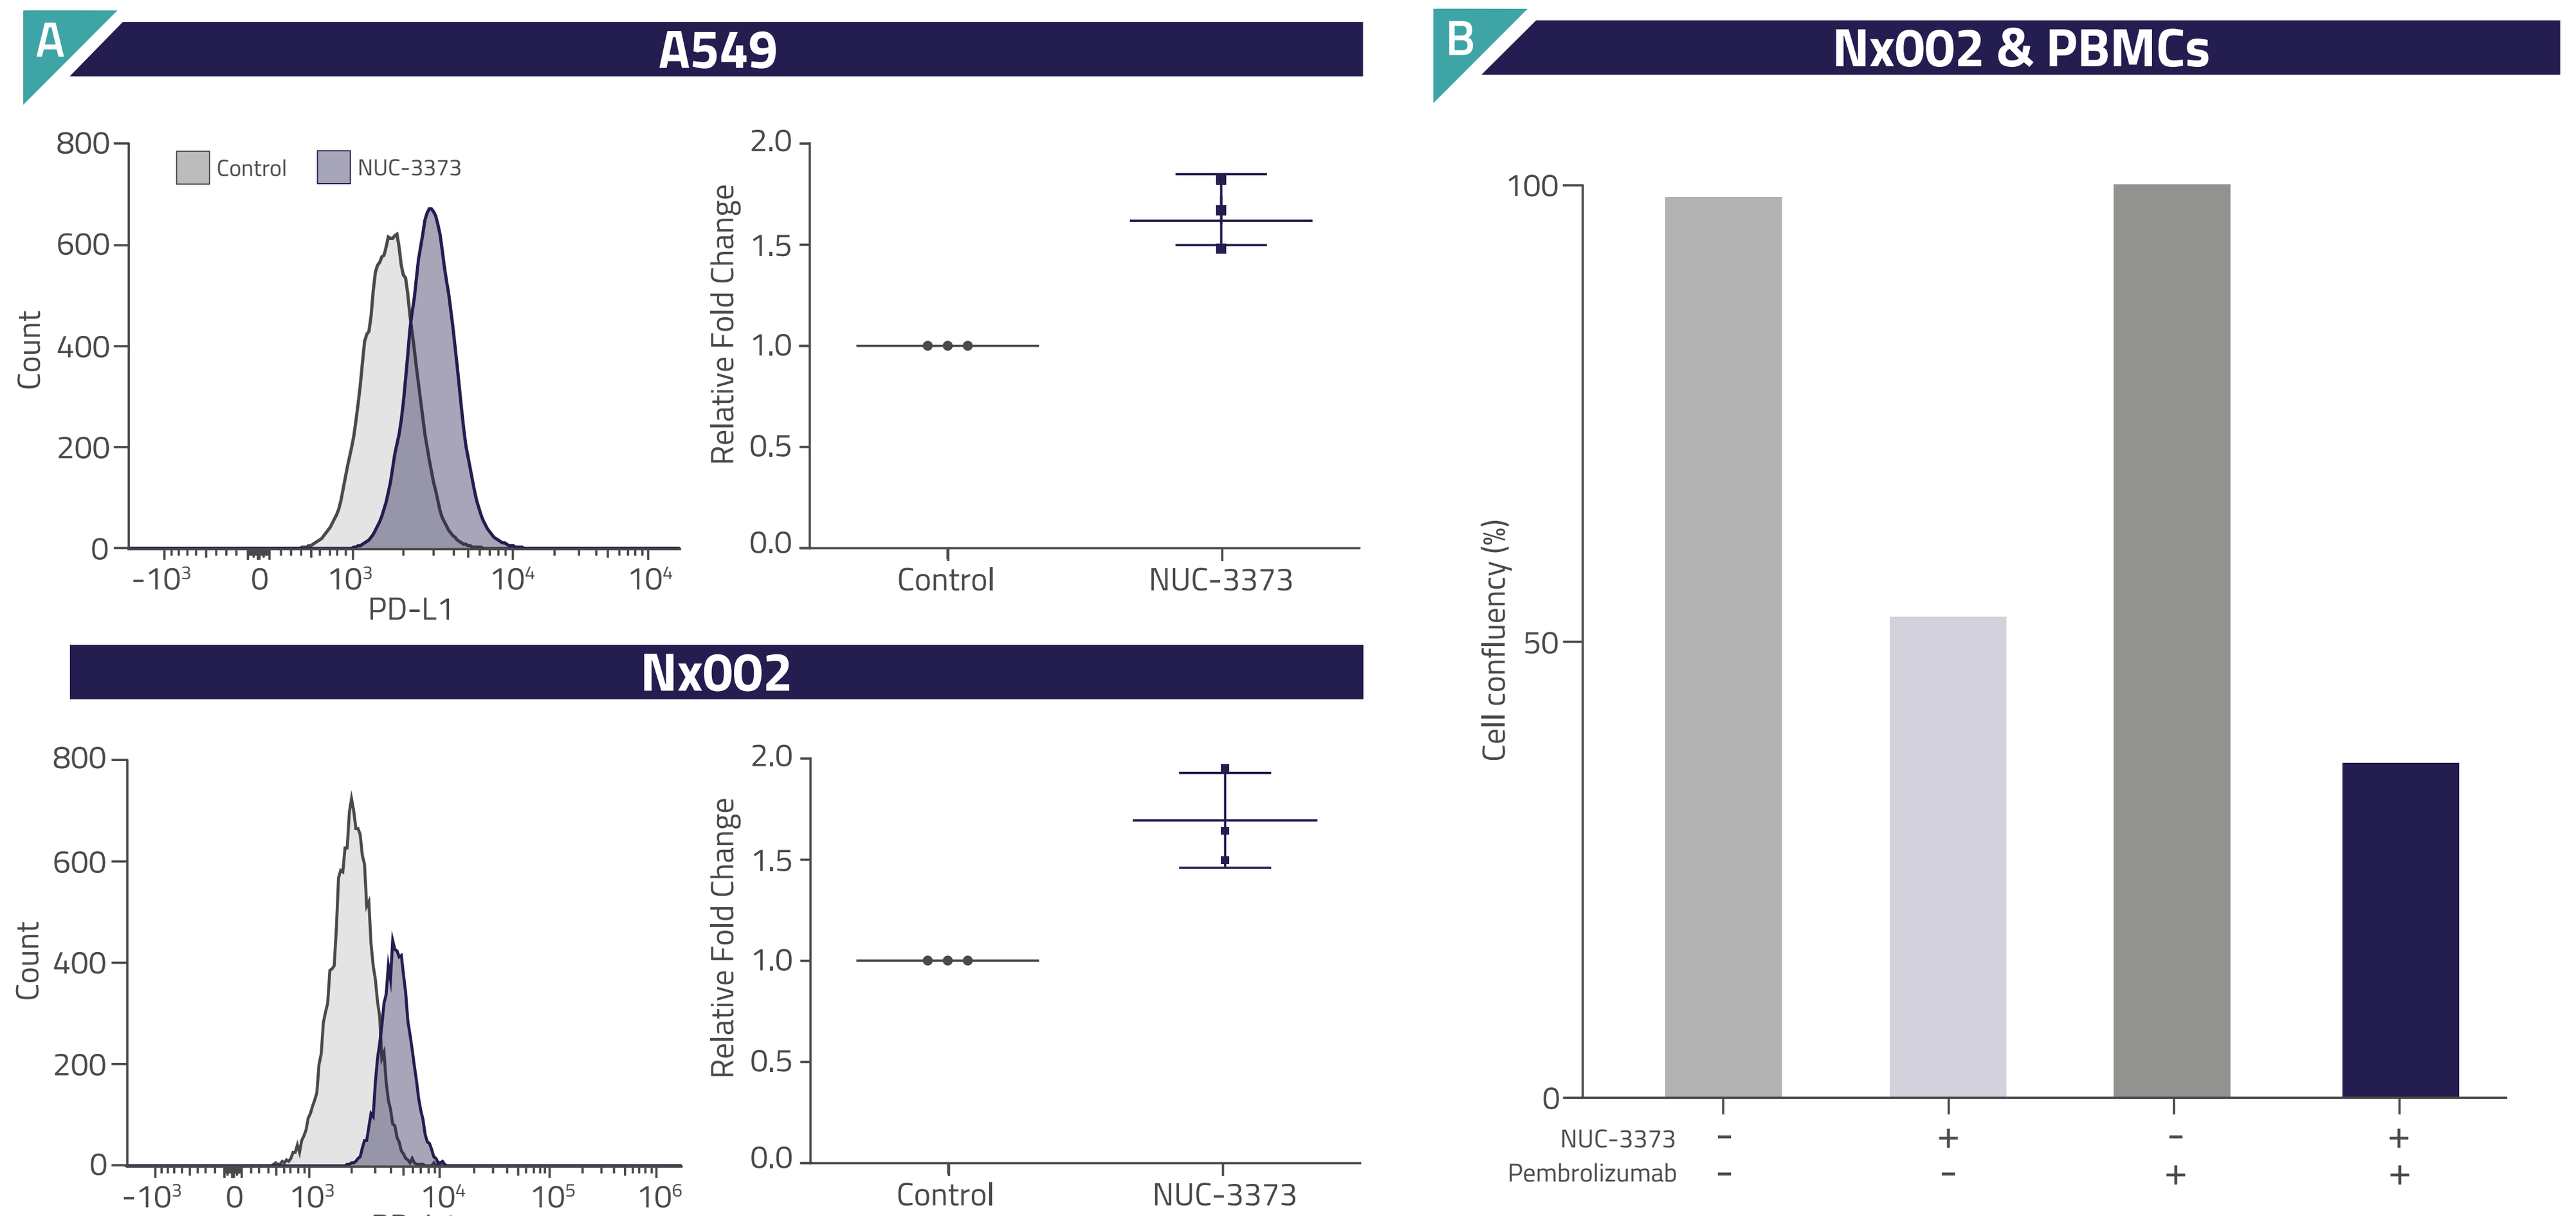
A
A549
Control
NUC-3373
Count
PD-L1
Relative Fold Change
Control
NUC-3373
Nx002
B
Nx002 & PBMCs
Cell confluency (%)
NUC-3373
Pembrolizumab

Figure 5: (A) PD-L1 surface expression in NSCLC cells treated with 10 μM NUC-3373 for 24 hours. (B) Confluence of Nx002 cells pre-treated with10 μM NUC-3373 and co-cultured with activated PBMCs for 72 hours (representative data shown).

- NUC-3373 increases cell surface PD-L1 expression in adenocarcinoma and squamous subtypes
- NUC-3373 promotes T-cell mediated cytotoxicity against NSCLC cells, which was increased by pembrolizumab

CONCLUSION

- NUC-3373 is a potent inhibitor of TS in NSCLC cells, regardless of histological subtype
- NUC-3373 is incorporated into DNA resulting in extensive DNA damage
- NUC-3373 increases cell surface expression of PD-L1 and potentiates T-cell-mediated cell death in combination with pembrolizumab
- NUC-3373 may be an effective treatment for NSCLC, regardless of histological subtype and basal TS expression

REFERENCES: 1. Ferlay J *et al.*, 2020. *Global Cancer Observatory*: Cancer Today. Lyon, France: International Agency for Research on Cancer. Available from https://gco.iarc.fr/today, accessed [12 Oct 2022] 2. Hirota T *et al.*, 2012. *Lung Cancer*; 77(1): 16-23 3. Scagliotti G *et al.*, 2009. *Oncologist*; 14(3): 253-263 4. Cavazzoni A *et al.*, 2020. *Cancers*; 12: 666 5. McGuigan C *et al.*, 2011. *Med Chem*; 27: 7247-7258 6. Vande Voorde J *et al.*, 2011. *Biochem Pharmacol*; 82: 441-452 7. Ciombor K *et al.*, 2020. *J Clin Oncol*; 38 (Suppl 4) 8. Bré J *et al.*, 2022. *Cancer Res*; 82 (Suppl 12): 1835

ABBREVIATIONS: 5-FU: 5-fluorouracil DHFU: dihydrofluorouracil DNA: deoxyribonucleic acid DPD: dihydropyrimidine dehydrogenase dTMP: deoxythymidine monophosphate dUMP: deoxyuridine monophosphate FBAL: α-fluoro-ß-alanine FdUr: fluorodeoxyuridine FUDP: fluorouridine diphosphate FUDR-DP: fluorodeoxyuridine diphosphate FUDR-MP: fluorodeoxyuridine monophosphate FUDR-TP: fluorodeoxyuridine triphosphate FUDR: fluorodeoxyuridine FUMP: fluorouridine monophosphate FUR: fluorouridine FUTP: fluorouridine triphosphate IC_{50}: half-maximal inhibitory concentration LC-MS/MS: liquid chromatography with tandem mass spectrometry NSCLC: non-small cell lung cancer OPRT: orotate phosphoribosyl transferase p-Chk1: phosphorylated checkpoint kinase 1 PBMC: peripheral blood mononuclear cell PD-L1: programmed cell death ligand 1 TK: thymidine kinase TP: thymidine phosphorylase TS: thymidylate synthase UK: uridine kinase UP: uridine phosphorylase γ–H2AX: phosphorylated H2A histone family member X

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.