Exposing the Heterogeneity of the Lipidome in the TME of Cutaneous Melanoma Following Treatment with NUC-7738 in Combination with anti-PD-1 Therapy

Alison L. Dickson[1,2], Greice M. Zickuhr[1], In Hwa Um[1], Ying Zhang[1], Ruth Plummer[3], Sarah P. Blagden[4], Stefan N. Symeonides[5], Natalie Cook[6], T.R. Jeffry Evans[7], Mustafa Elshani[1,2] & David J. Harrison[1,2]

1) University of St Andrews, St Andrews, UK 2) NuCana plc, Edinburgh, UK 3) Newcastle ECMC, Northern Centre for Cancer Care, Freeman Hospital, Newcastle Upon Tyne, UK 4) Oxford ECMC, Churchill Hospital, University of Oxford, UK 5) Edinburgh ECMC, Western General Hospital, Edinburgh, UK 6) The Christie NHS Trust/University of Manchester, UK 7) The Beatson West of Scotland Cancer Centre/University of Glasgow, UK

Abstract Number: 6222

Email: alisondickson@nucana.com

Introduction

- Cancer cells adapt to hypoxic and nutrient deprived environments by reprogramming lipid biosynthesis to accelerate malignant behaviour[1]
- Lipids are not only structural components of cell membranes but also serve as signaling molecules within the tumor microenvironment (TME) to modulate immune cells[2]
- Cancer cells transitioning to a malignant phenotype demonstrate marked changes in lipid metabolism including:
 - Changes in the ratio of monounsaturated fatty acids (MUFAs) to polyunsaturated fatty acids (PUFAs)
 - Dysregulation of sphingolipids, crucial for tumor progression and survival
- Reprogramming lipid metabolism within the TME is a recognised strategy for boosting the effects of immunotherapy[3]

Lipid Metabolism in the TME

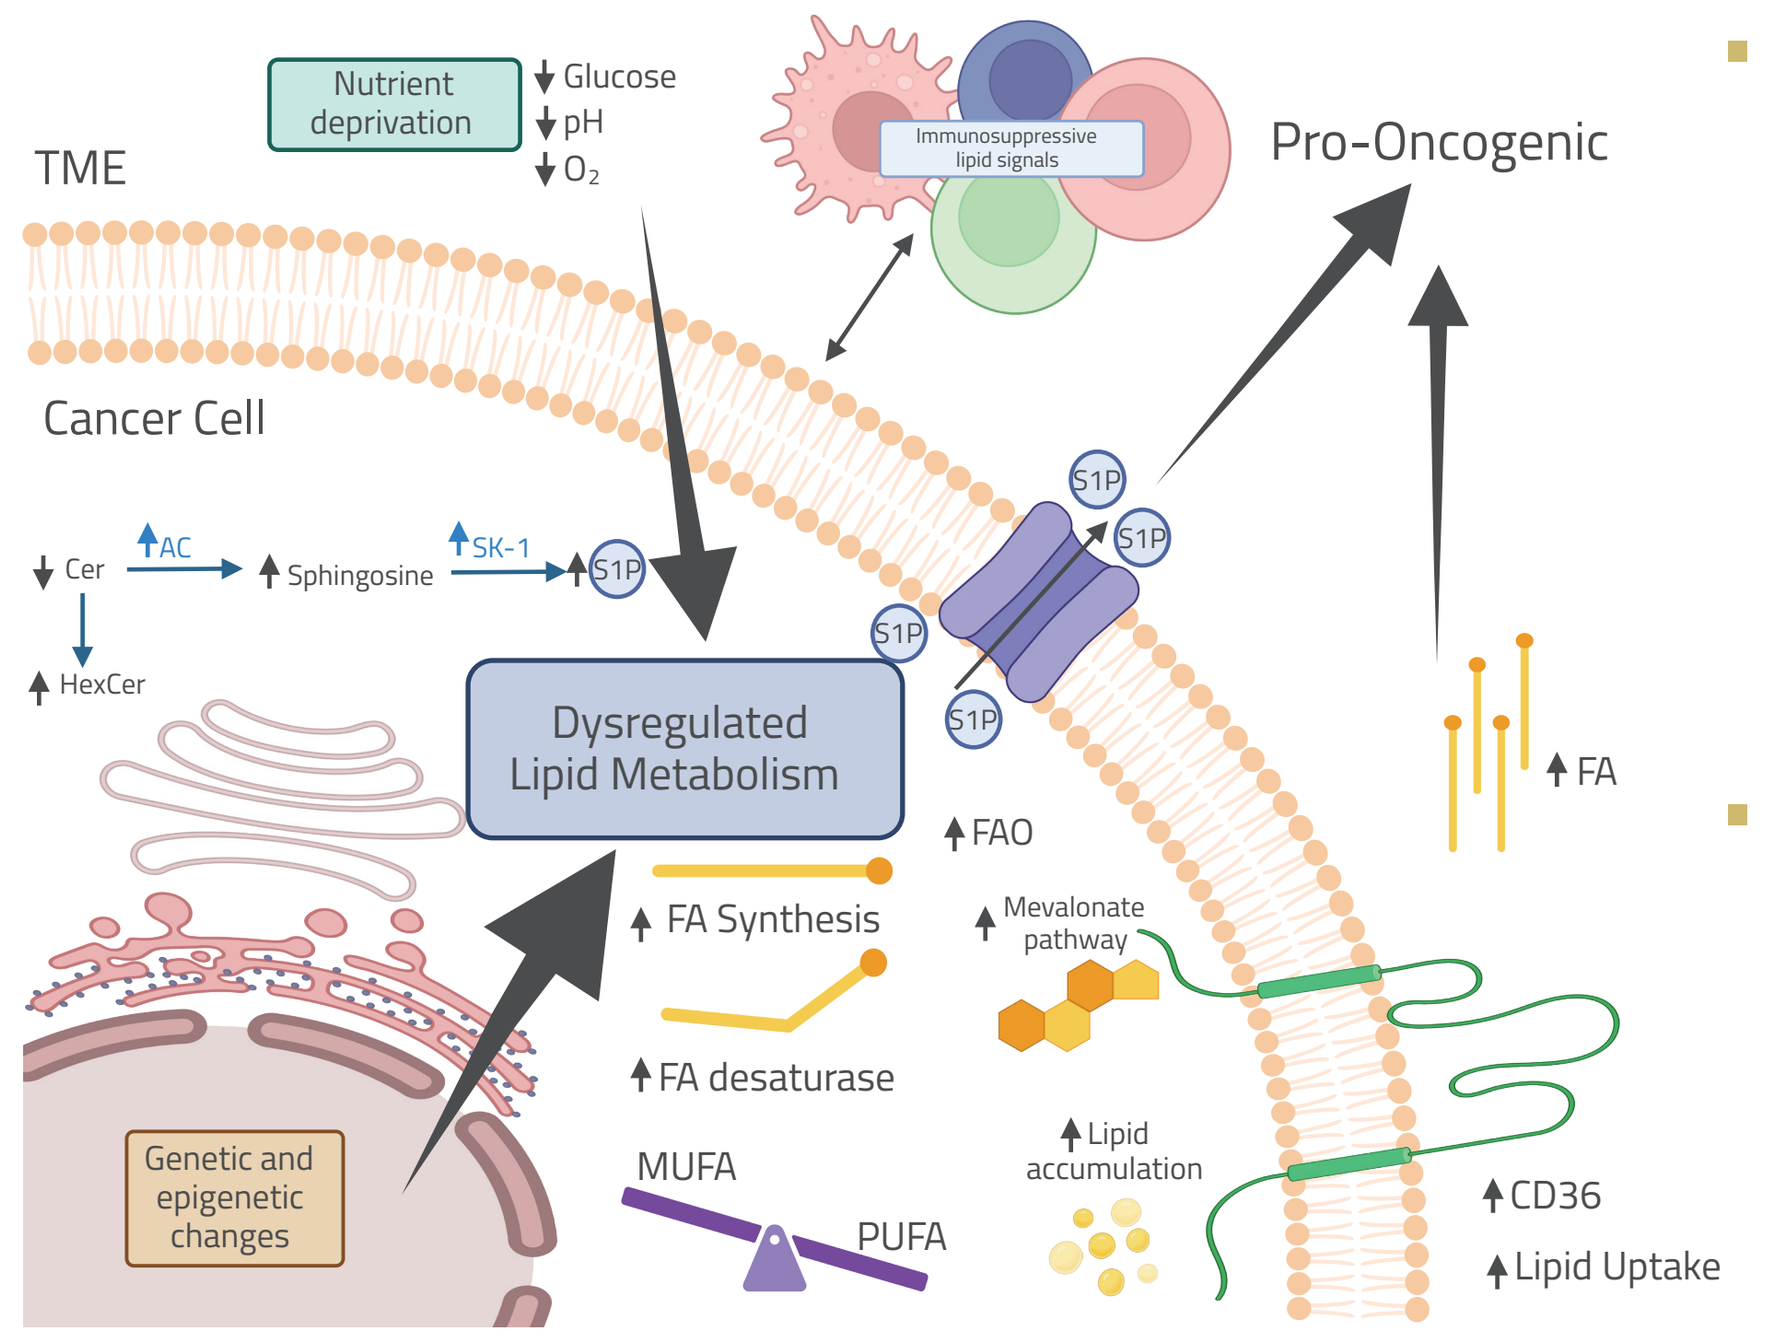

Nutrient deprivation
Glucose
pH
O_2
Immunosuppressive lipid signals
TME
Pro-Oncogenic
Cancer Cell
Cer
AC
Sphingosine
SK-1
S1P
HexCer
Dysregulated Lipid Metabolism
FAO
FA Synthesis
FA desaturase
Mevalonate pathway
Lipid accumulation
FA
CD36
Lipid Uptake
Genetic and epigenetic changes
MUFA
PUFA

- NUC-7738 generates sustained intracellular levels of 3' deoxyadenosine triphosphate (3'-dATP), which profoundly alters RNA regulatory processes in tumor cells, resulting in changes in expression of proteins related to lipid metabolism
- Using paired biopsies from patients treated with treated with NUC-7738 ± pembrolizumab, we aim to investigate the potential role of NUC-7738 as a lipid reprogramming agent

Figure 1: In nutrient deprived conditions cancer cells increase lipogenesis, fatty acid uptake via CD36 and fatty acid oxidation (FAO). Cancer cells rich in monounsaturated fatty acids (MUFA) prevent ferroptosis. Alterations in sphingolipid biosynthesis cause accumulation of the pro-tumorigenic and immunosuppressive sphingosine-1-phosphate (S1P) in the TME.

Methods

- Snap frozen and FFPE biopsies were collected from 6 patients (1 x mucosal melanoma and 5x cutaneous melanoma) treated with NUC-7738 ± pembrolizumab. Biopsies were collected pre- and post- drug infusion (≤6h post infusion).
- TIC normalised data was exported into MetaboAnalyst 5.0 for statistical analysis (SAM, PCA, fold-change). LIPID MAPS® Structure Database was used to tentatively assign lipid species.

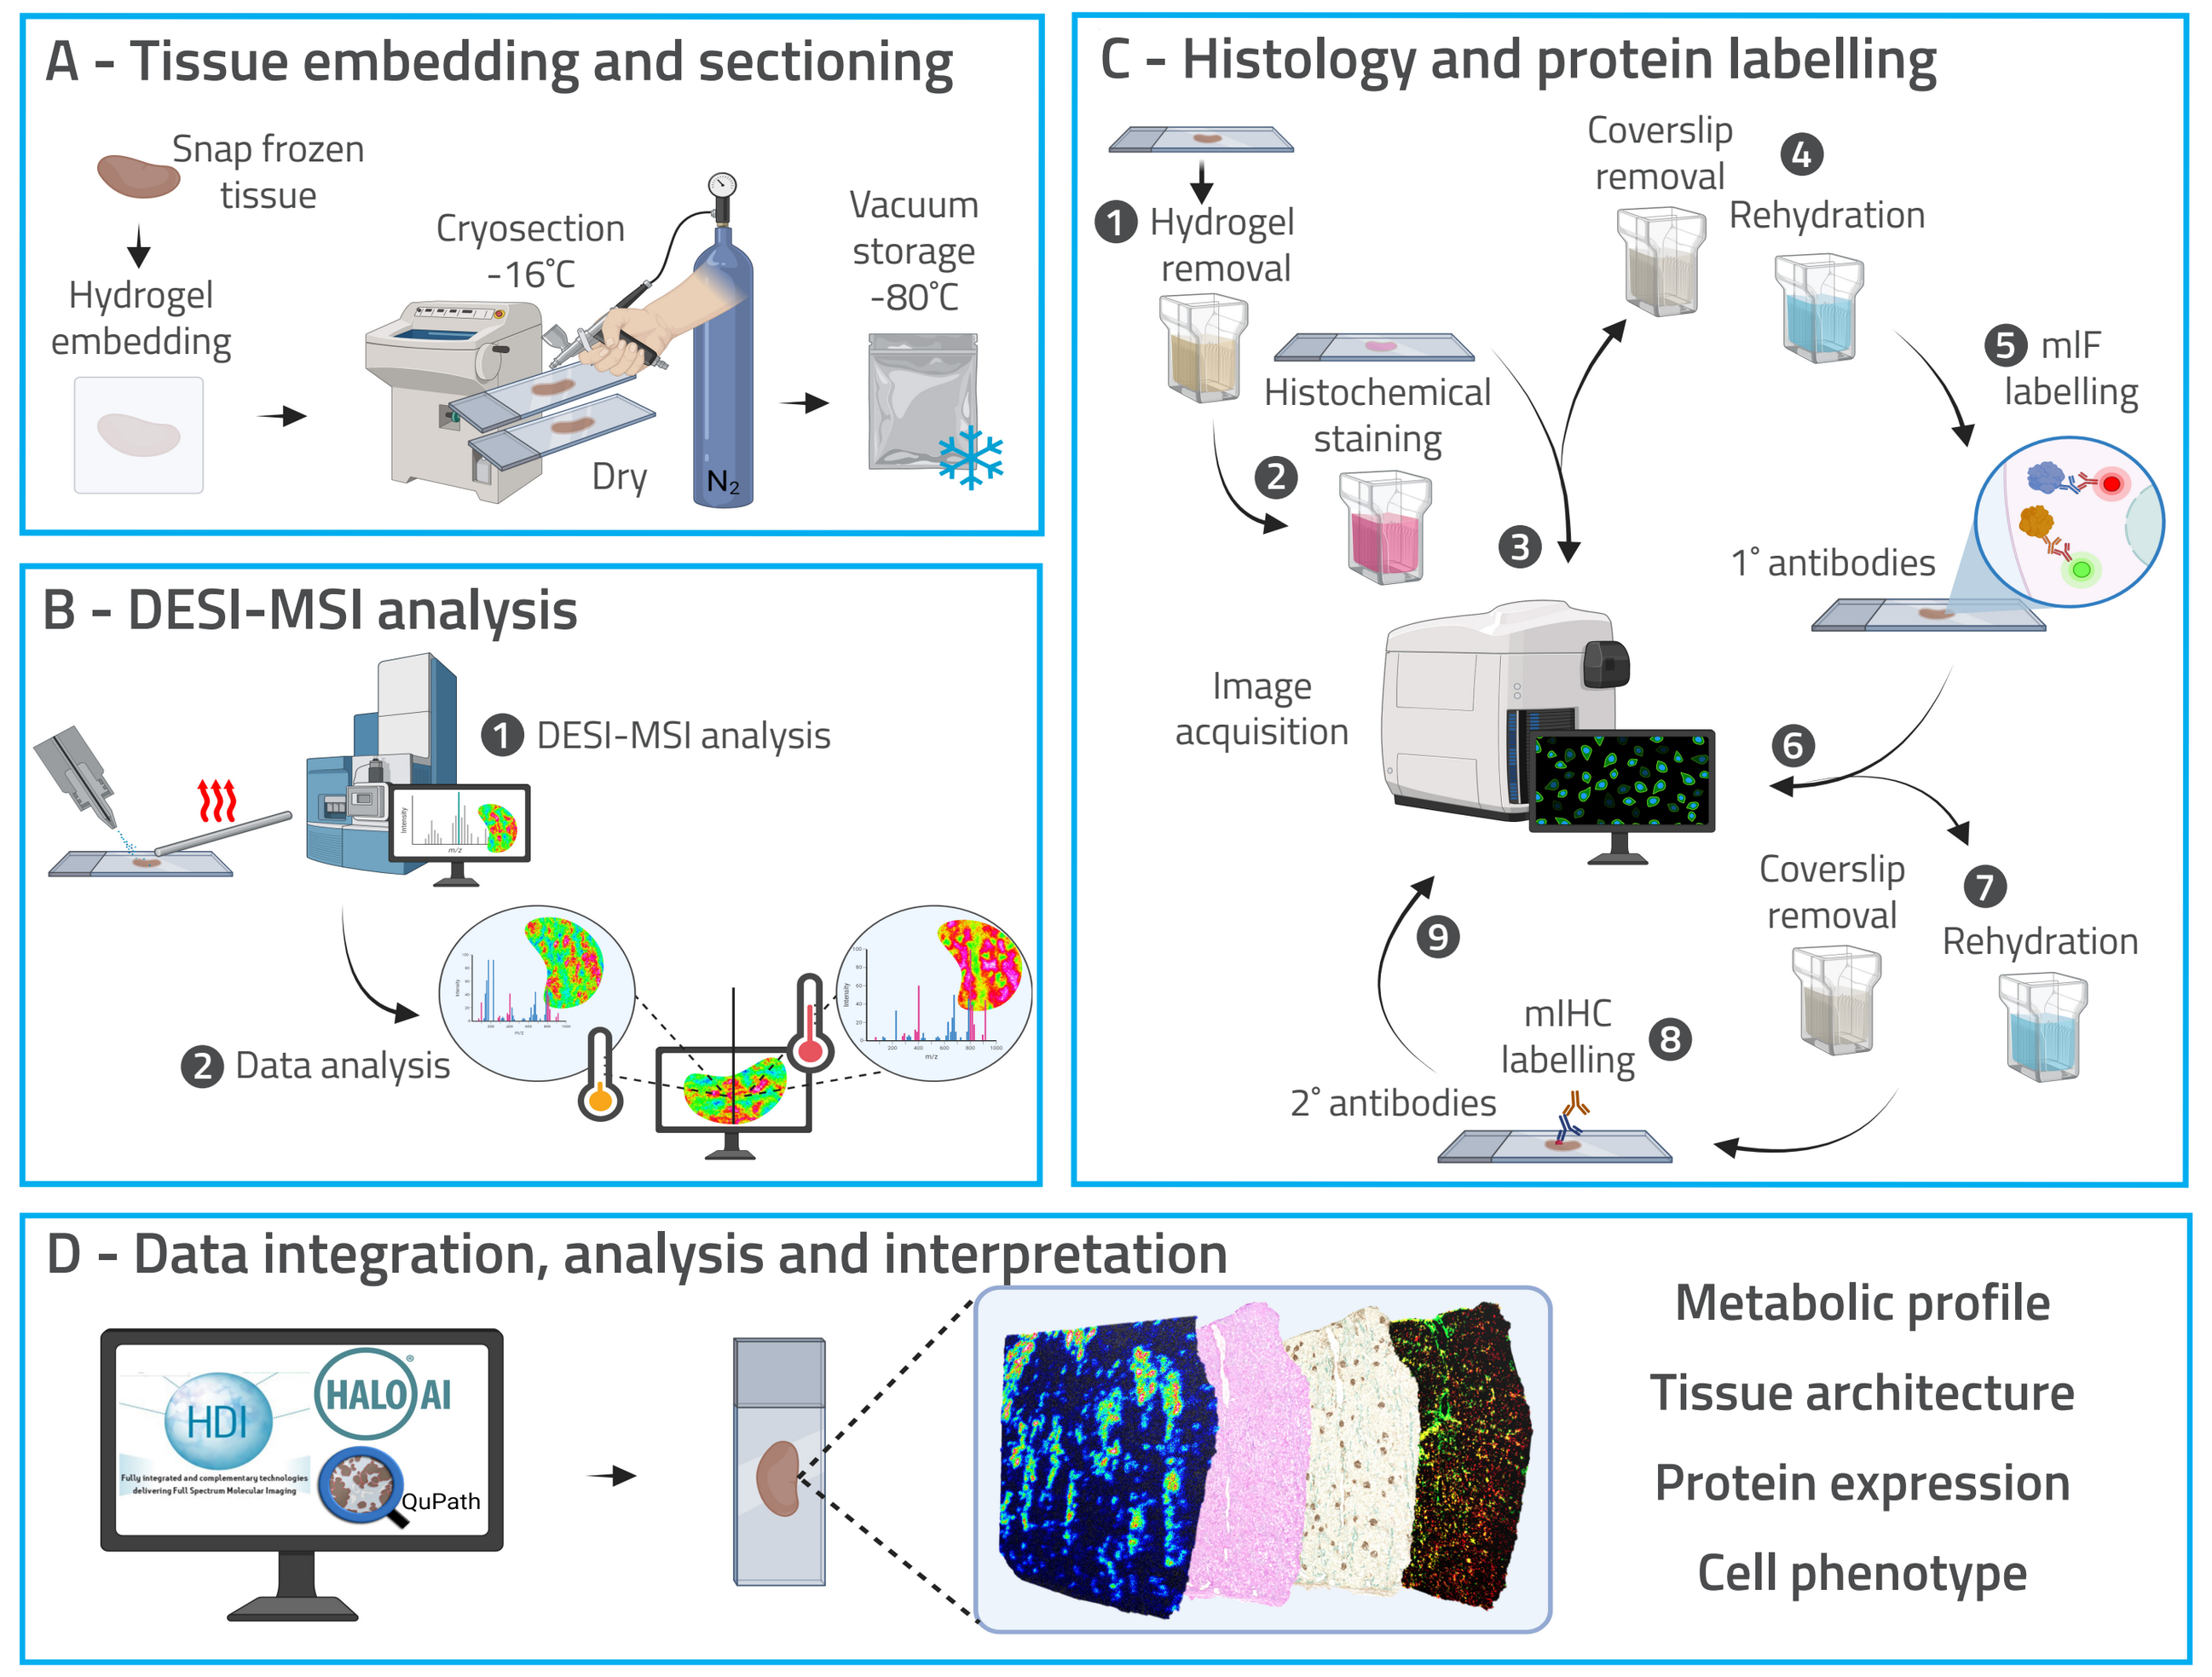

A - Tissue embedding and sectioning
Snap frozen tissue
Hydrogel embedding
Cryosection -16°C
Dry
N_2
Vacuum storage -80°C
B - DESI-MSI analysis
1 DESI-MSI analysis
2 Data analysis
C - Histology and protein labelling
1 Hydrogel removal
2 Histochemical staining
3
Coverslip removal
4 Rehydration
5 mIF labelling
1° antibodies
Image acquisition
6
Coverslip removal
7 Rehydration
8 mIHC labelling
9
2° antibodies
D - Data integration, analysis and interpretation
HDI
HALO AI
QuPath
Metabolic profile
Tissue architecture
Protein expression
Cell phenotype

Figure 2: Multi-Modal Imaging Workflow for 10μm single-section tissue analysis

Lipid Profiles of Melanoma Cells Change Following Treatment

- The emergence of two distinct classes of lipids suggests NUC-7738 ± pembrolizumab changes the lipidome in the melanoma TME

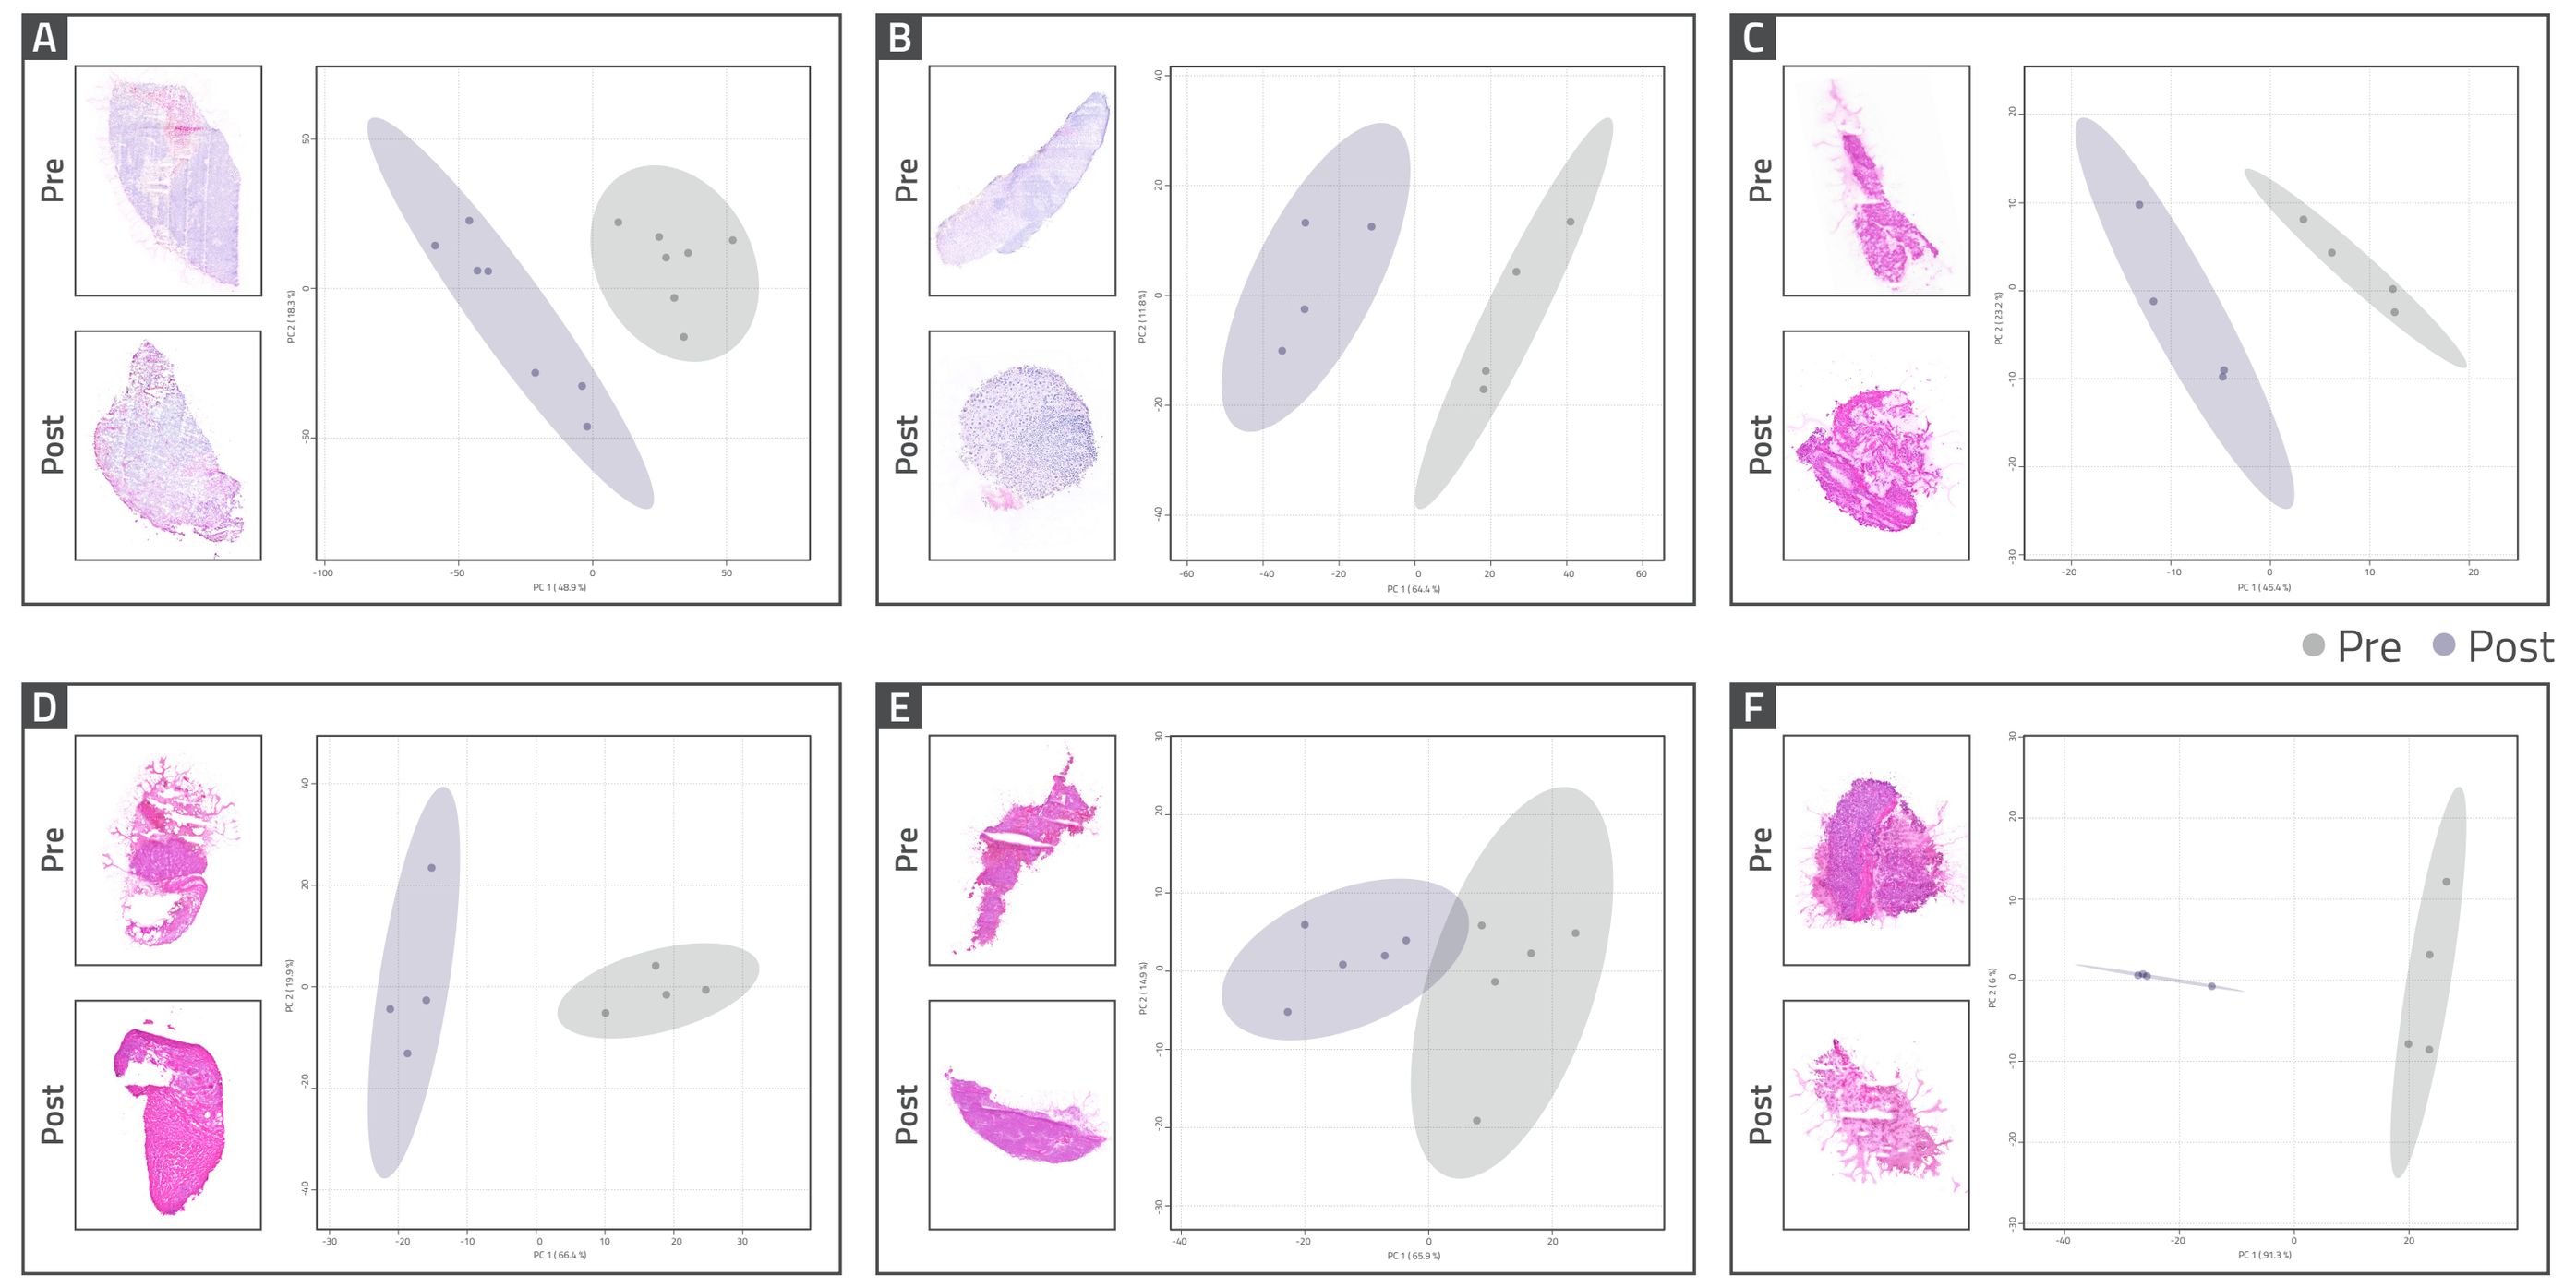

A
B
C
D
E
F
Pre
Post

Figure 3: PCA 2D scores plots of lipids in tumor regions from biopsies pre- and post-treatment (A, mucosal and B-F, cutaneous melanoma). ROI were selected by high-magnification analysis of viable tumor regions of H&E-stained sections following DESI-MSI.

Heterogeneity of Lipids and Immune Cells in the TME

- Dimension reduction analysis (UMAP) of data (>1000 lipids) reveals distinct regions within each biopsy

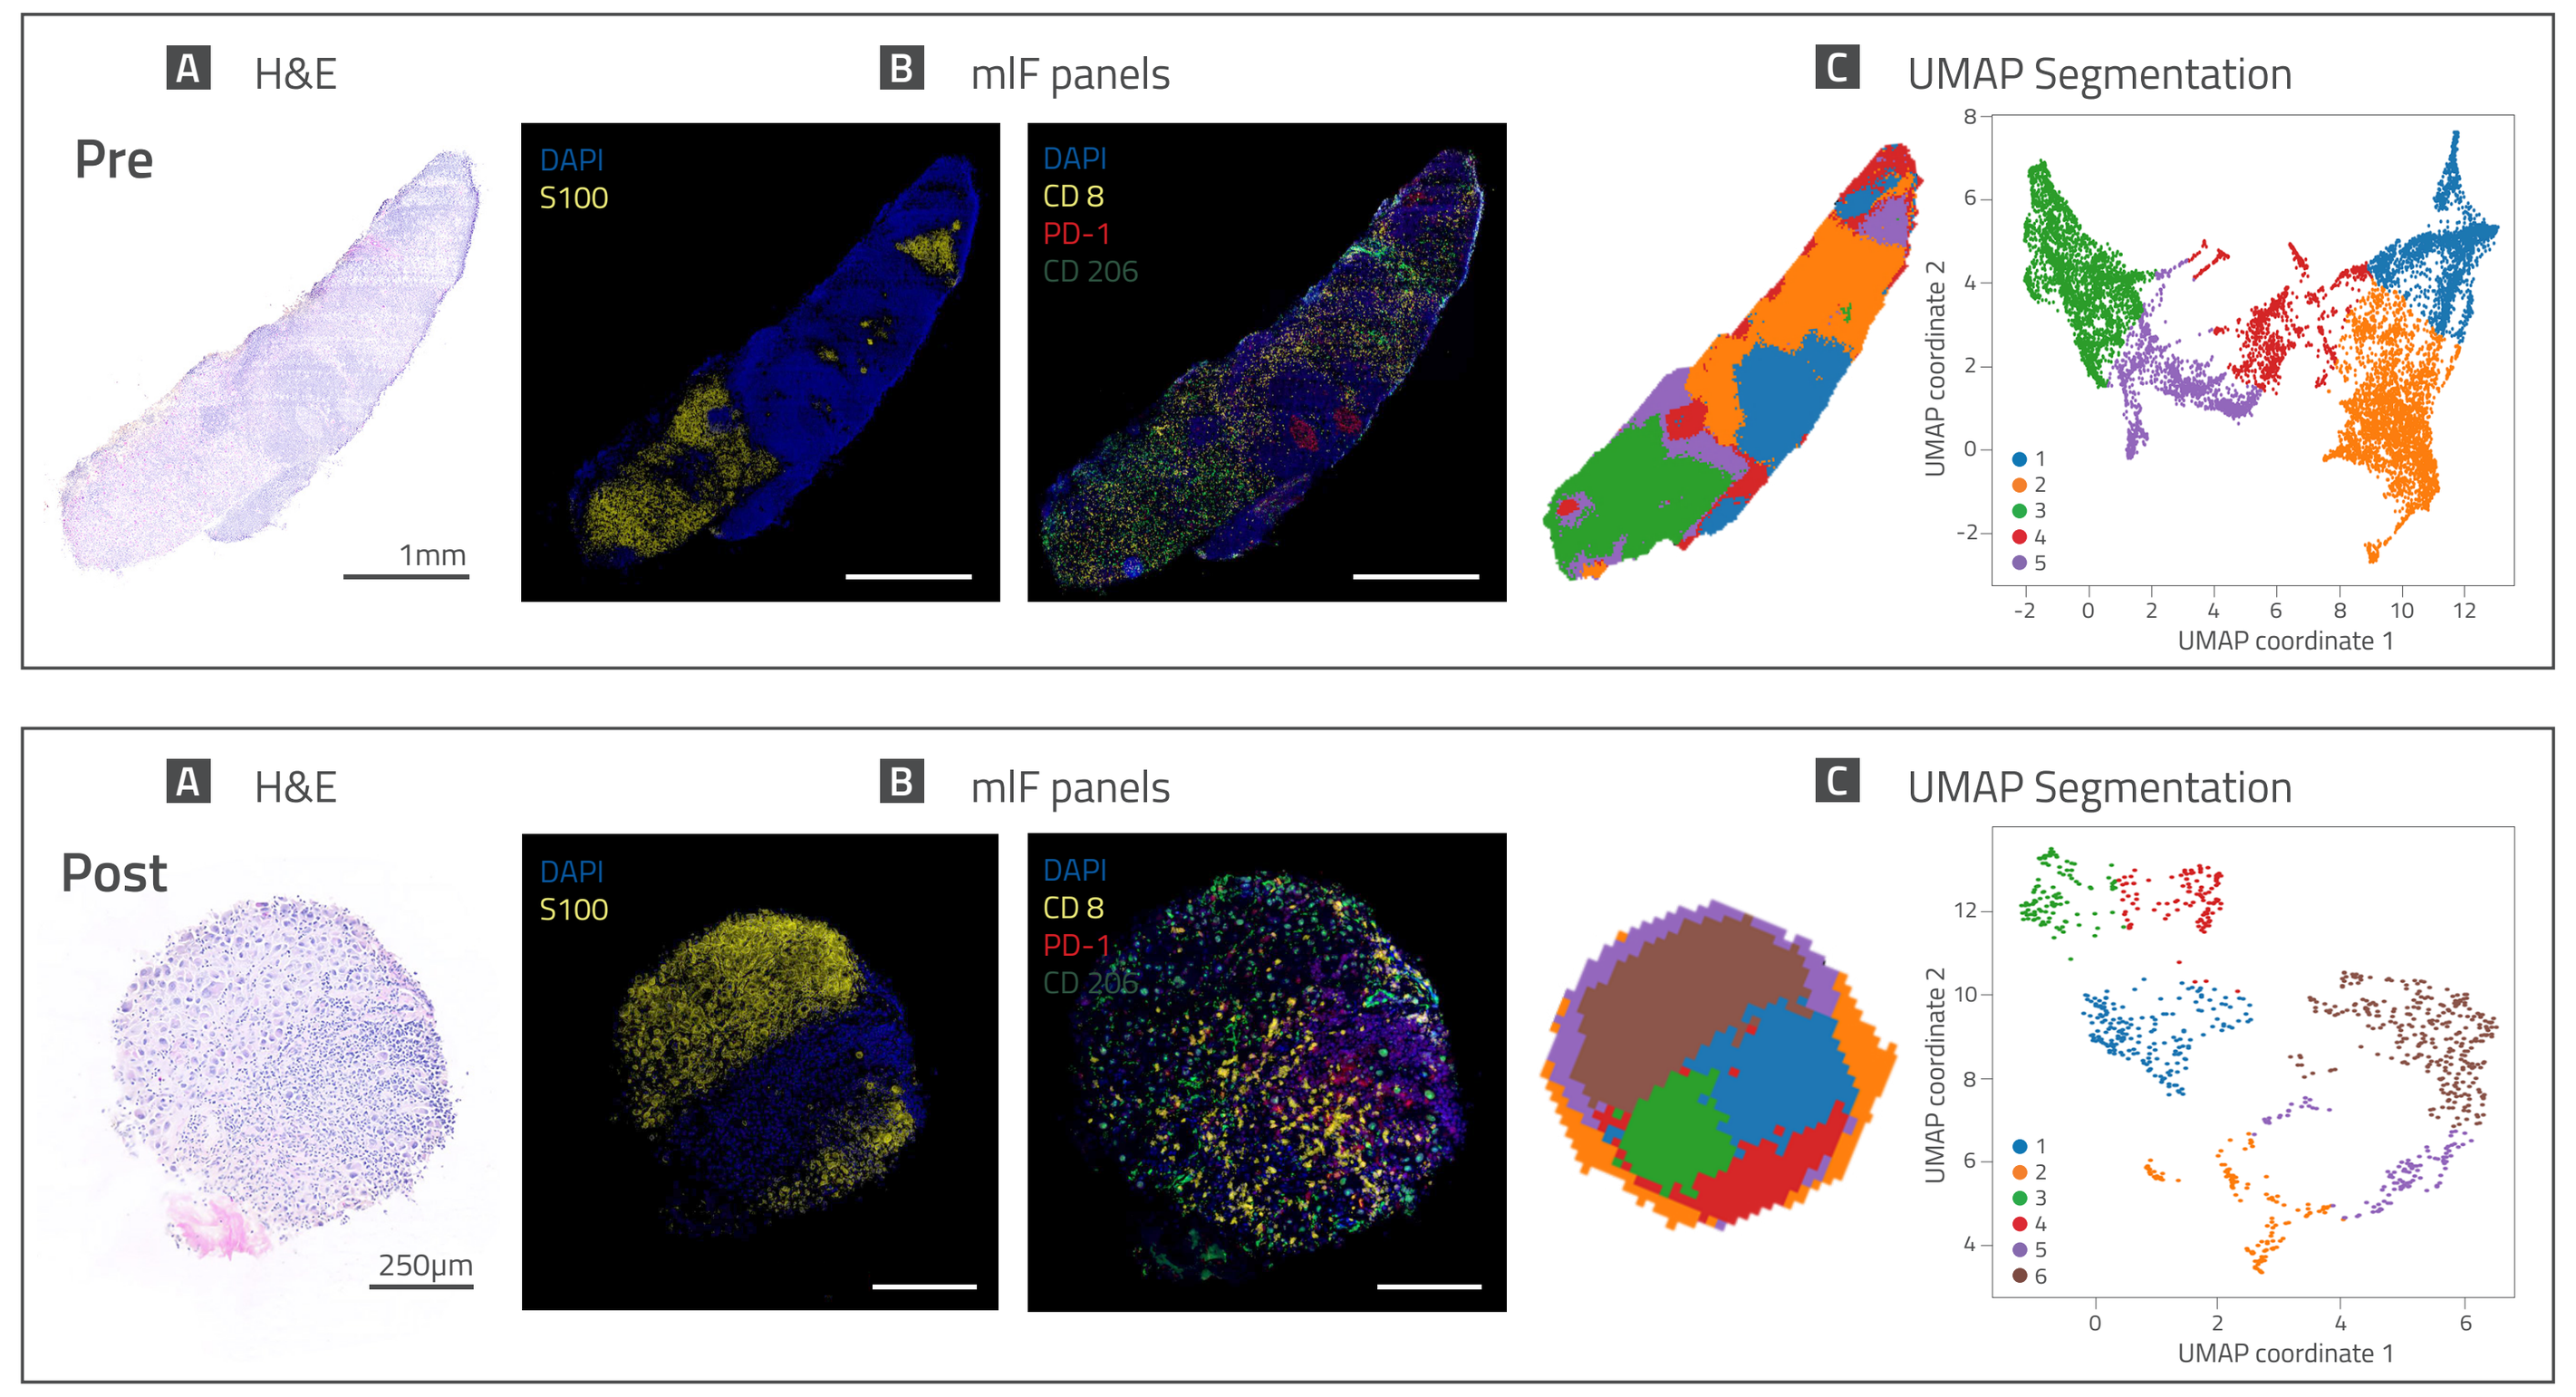

A H&E
B mIF panels
C UMAP Segmentation
Pre
1mm
DAPI
S100
DAPI
CD 8
PD-1
CD 206
UMAP coordinate 2
UMAP coordinate 1
Post
250μm

Figure 4: A) Post DESI-MSI H&E section, B) multiplex immunofluorescence (mIF) of DAPI co-localised with; s100 positive cells (indicative of malignant melanoma), CD 8 positive cells (T cells), CD 206 positive cells (macrophages) and PD-1 positive cells. C) UMAP segmentation of single 10μm section based on UMAP cluster data.

Spatial Co-registration for Quantitative Image Analysis

- Quantification of individual cell types within distinct metabolic regions of the tissue show which cell types may be driving changes in lipid signatures

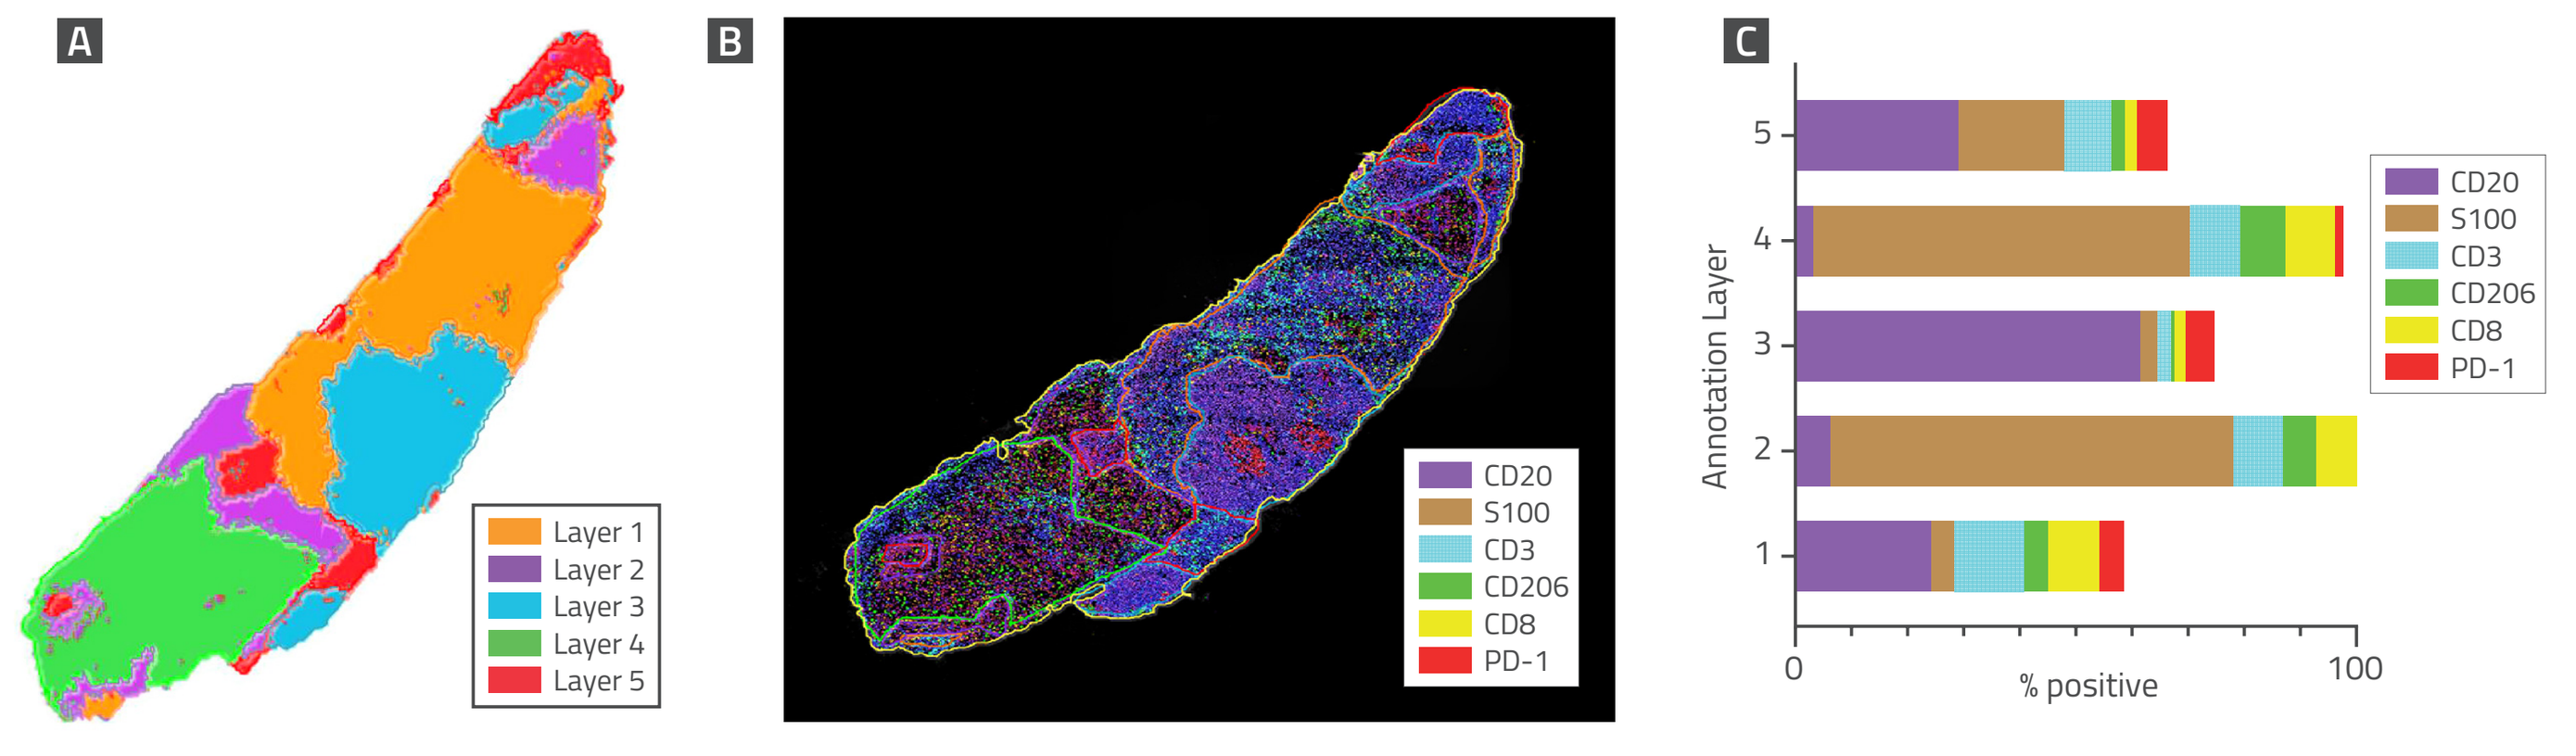

A
Layer 1
Layer 2
Layer 3
Layer 4
Layer 5
B
CD20
S100
CD3
CD206
CD8
PD-1
C
Annotation Layer
% positive

Figure 5: A) UMAP segmentation integrated into HALO AI analysis software as annotation layers. B) annotation layers applied to fused mIF and mIHC images. C) population of cells in each annotation layer determined by random forest classification.

Fatty Acid Reprogramming

- NUC-7738 + pembrolizumab changes the balance of fatty acids in favor of PUFAs, indicative of a shift towards a less aggressive oncogenic phenotype
- MUFAs have been linked to malignant behaviour and resistance to chemotherapy whilst PUFAs have been associated with susceptibility to reactive oxygen species and ferroptosis[4]

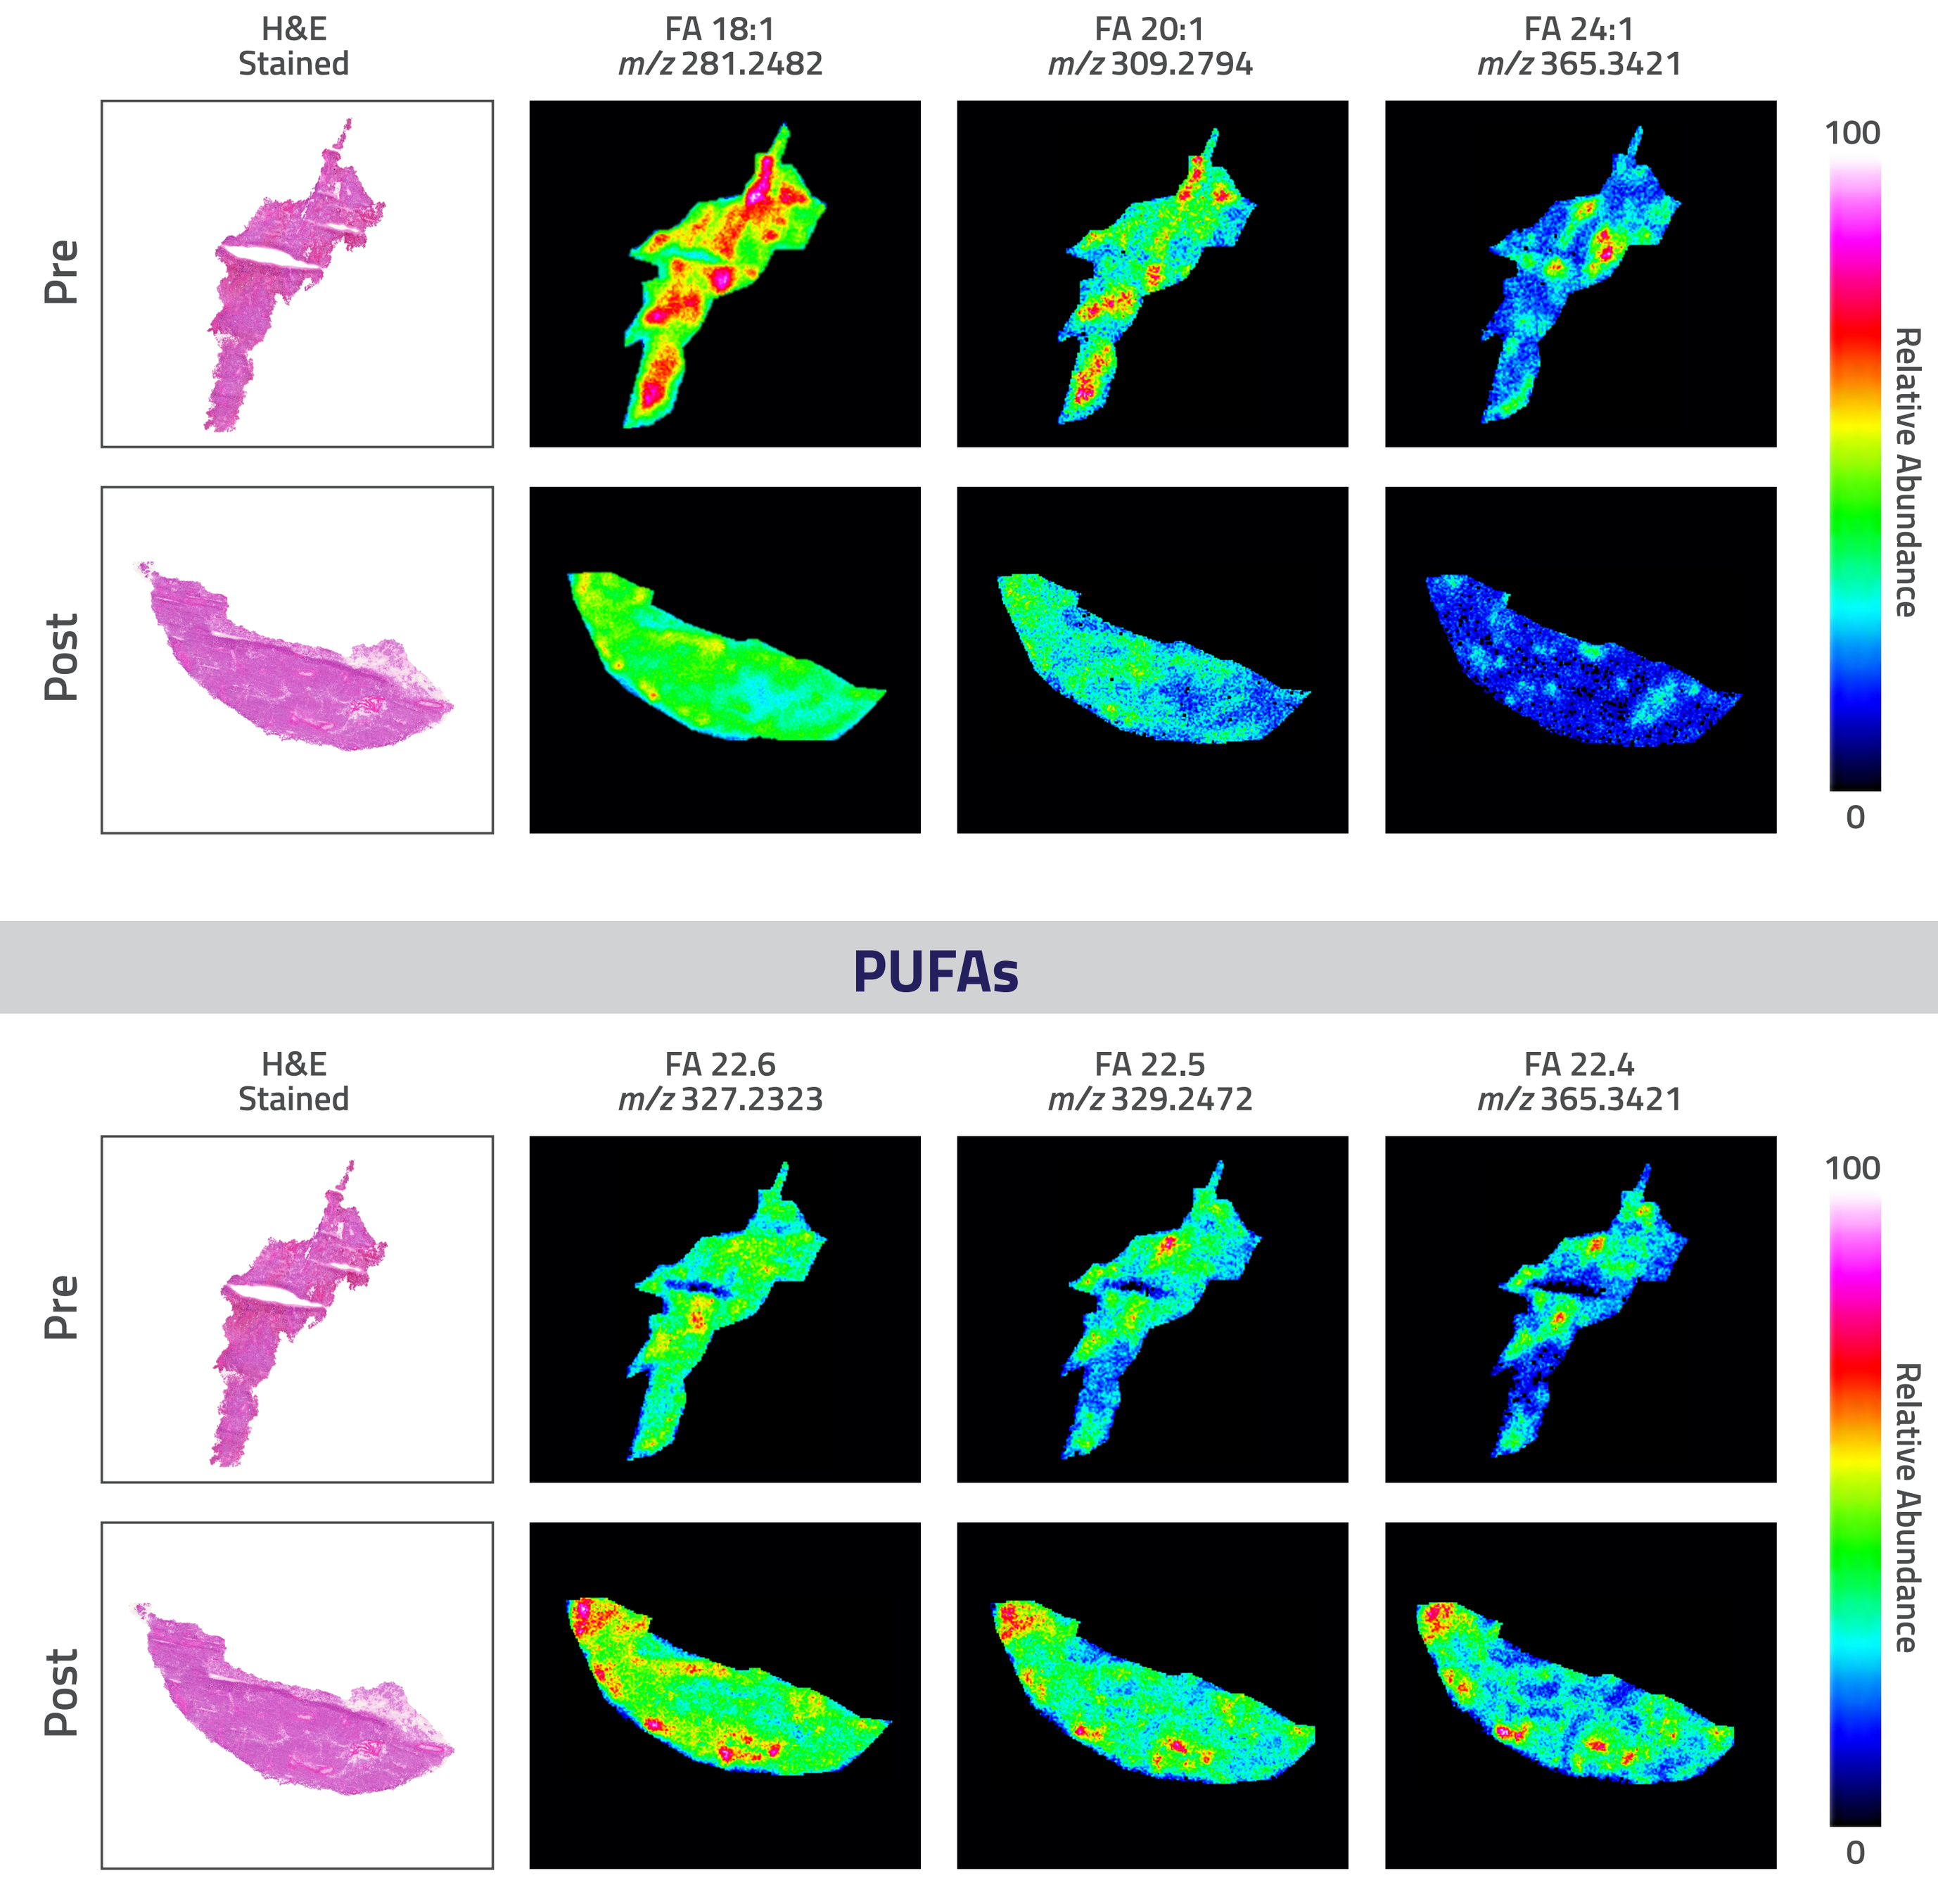

MUFAs
H&E Stained
FA 18:1 m/z 281.2482
FA 20:1 m/z 309.2794
FA 24:1 m/z 365.3421
Pre
Post
Relative Abundance
100
0
PUFAs
H&E Stained
FA 22.6 m/z 327.2323
FA 22.5 m/z 329.2472
FA 22.4 m/z 365.3421

Figure 6: DESI-MSI ion heatmaps show spatial distribution and a decrease in the relative abundance of three MUFA species and increase in three PUFA species in viable tumor regions following NUC-7738 + pembrolizumab treatment

Changes in Apoptotic Related Lipids

- NUC-7738 + pembrolizumab causes a reduction in levels of HexCer species in tumor area
- HexCer, a subclass of sphingolipids are anti-apoptotic and associated with chemoresistance in melanoma patients[5]

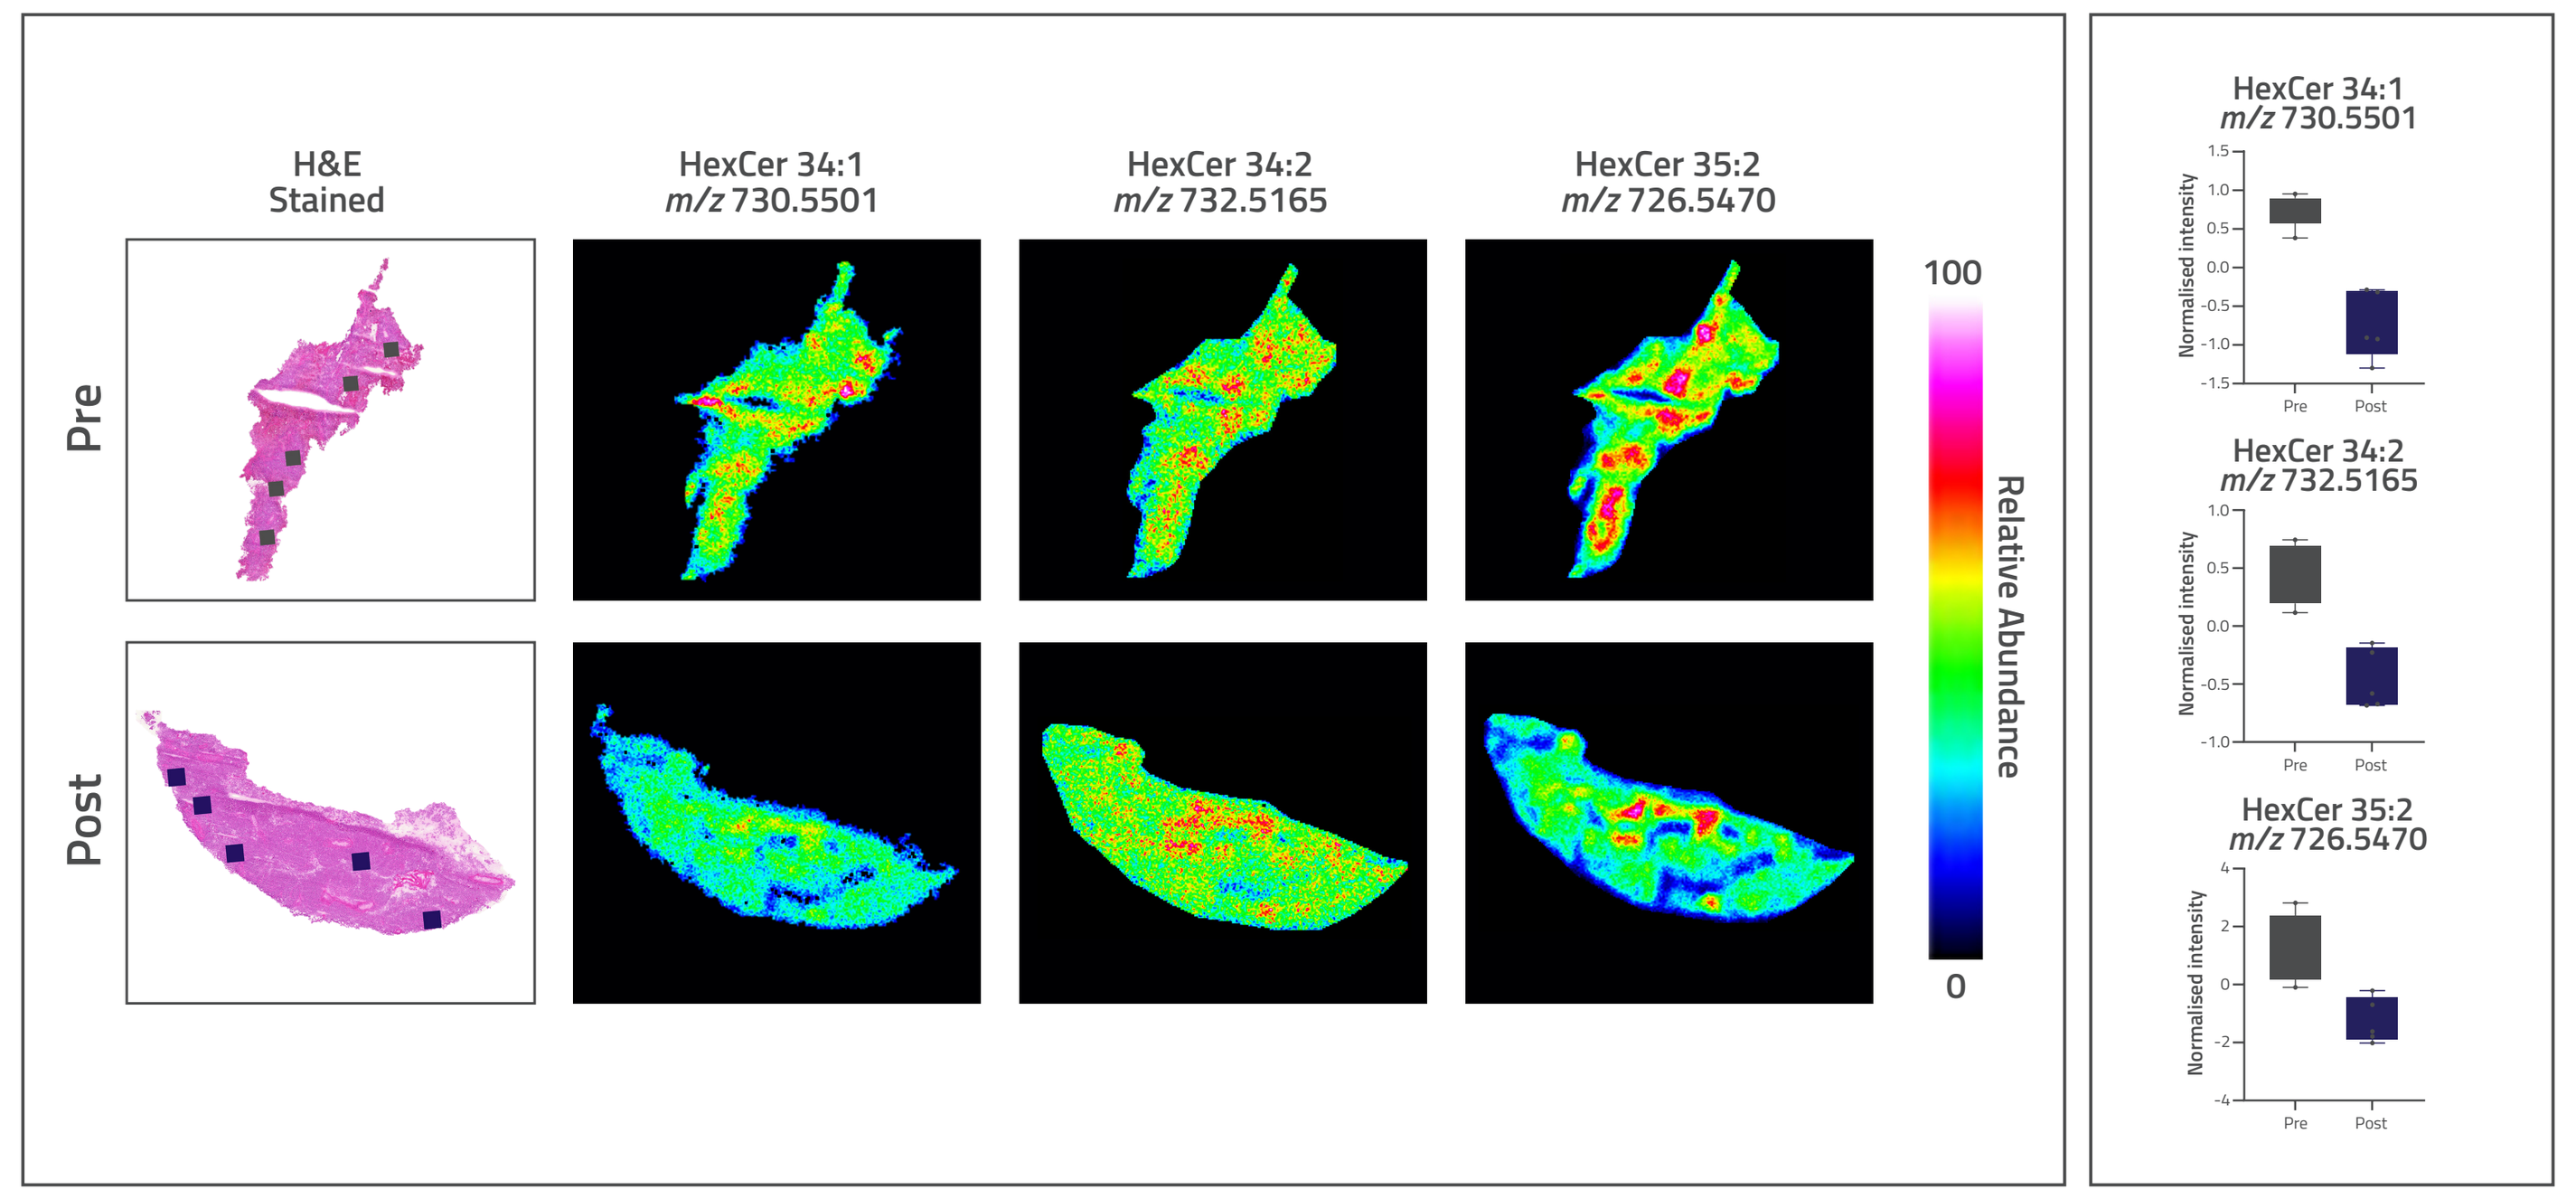

H&E Stained
HexCer 34:1 m/z 730.5501
HexCer 34:2 m/z 732.5165
HexCer 35:2 m/z 726.5470
Pre
Post
Relative Abundance
Normalised intensity

Figure 7: DESI-MSI ion heatmaps show spatial distribution and a significant decrease (|d| < 5) in the relative abundance of three HexCer species in viable tumor regions following NUC-7738 + pembrolizumab treatment

- Pro-apoptotic ceramide (Cer) species increase following treatment

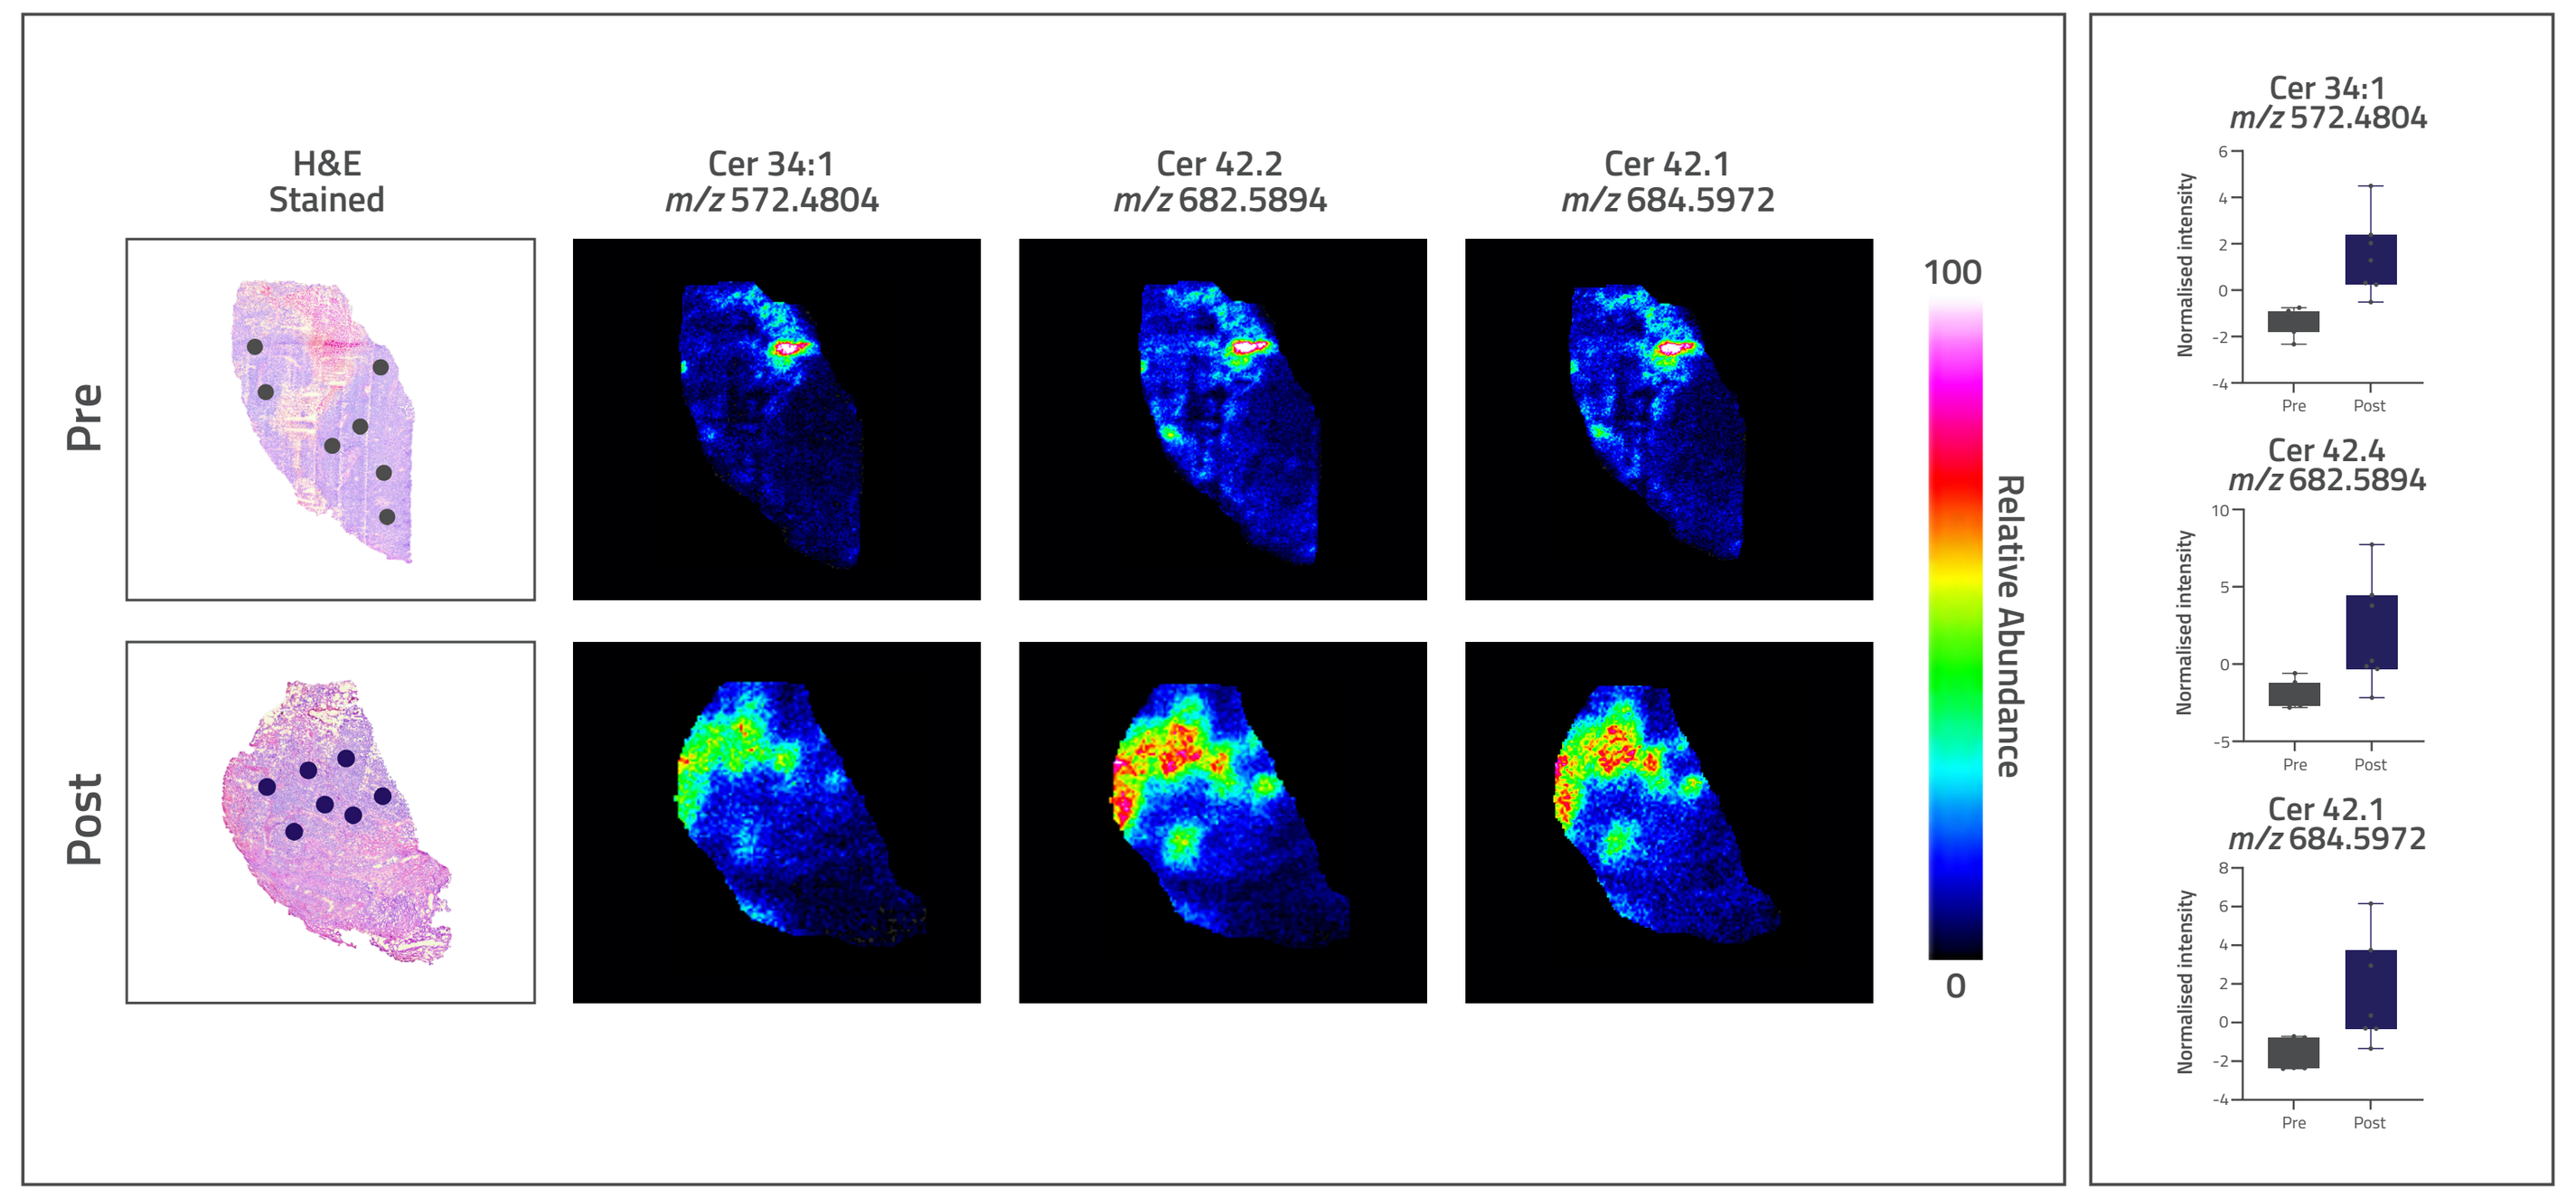

H&E Stained
Cer 34:1 m/z 572.4804
Cer 42.2 m/z 682.5894
Cer 42.1 m/z 684.5972
Cer 42.4 m/z 682.5894
Pre
Post
Relative Abundance
Normalised intensity

Figure 8: DESI-MSI ion heatmaps show spatial distribution and a significant increase (|d| > 3) in the relative abundance of three Cer species in viable tumor regions following NUC-7738 treatment

Multi-Modal Imaging Confirms Lipidomic Reprogramming

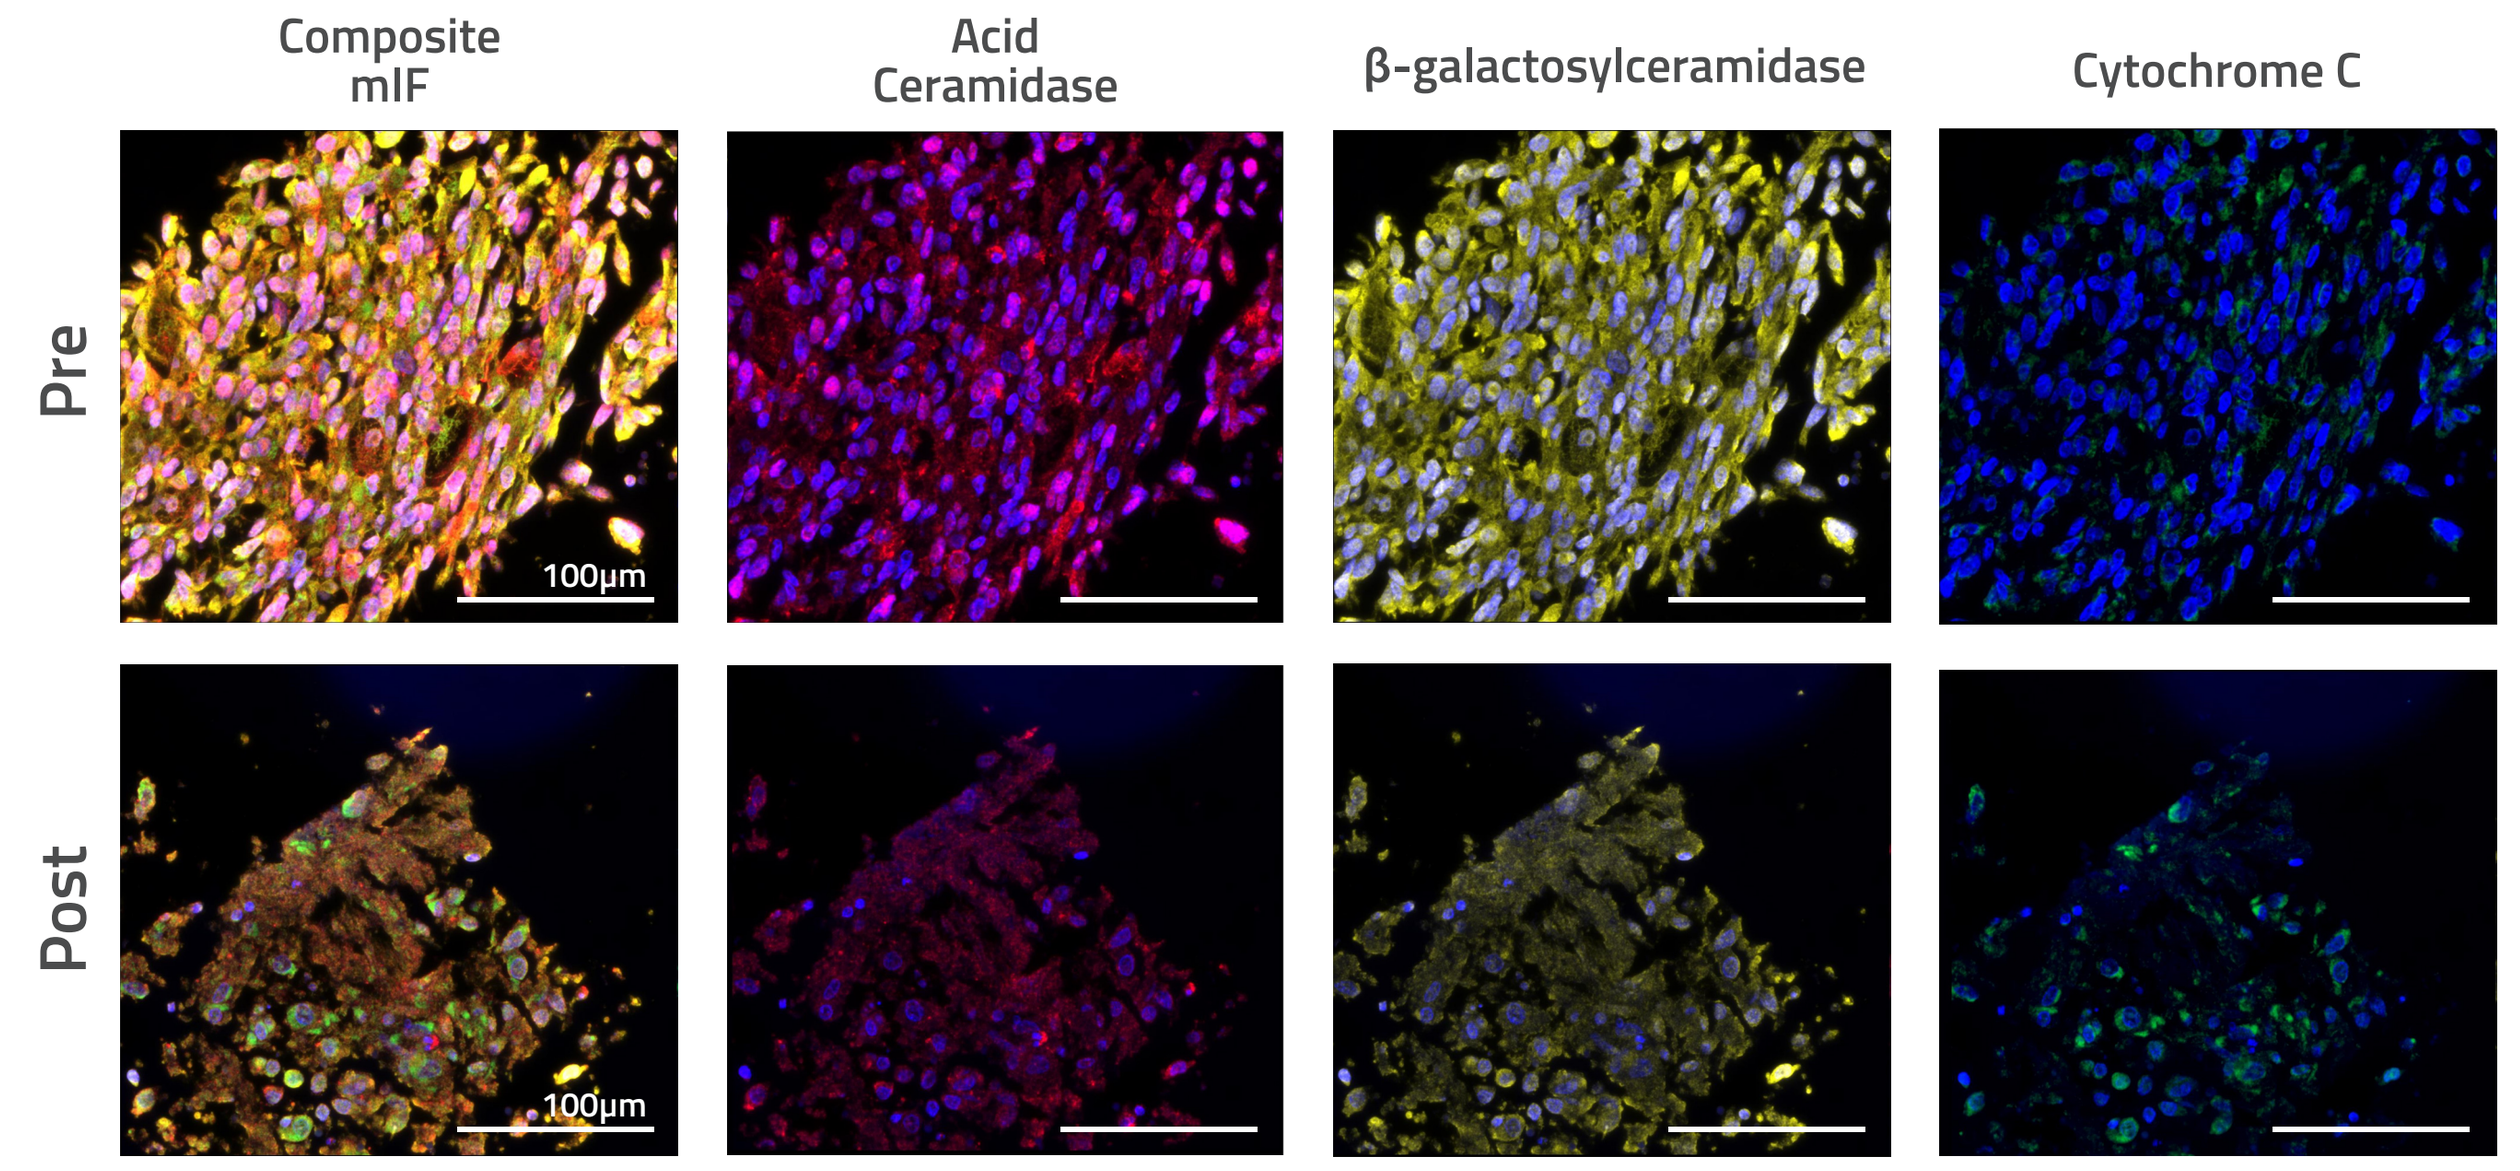

Composite mIF
Acid Ceramidase
β-galactosylceramidase
Cytochrome C
Pre
Post
100μm

- Pro-apoptotic ceramide (Cer) species increase following treatment with NUC-7738 corresponding with a decrease in protein expression of acid ceramidase (AC) which hydrolyses ceramides into sphingosine and is associated with tumor progression[6]
- β-galactosylceramidase, a key enzyme in sphingolipid metabolism often upregulated in melanoma progression and associated with poor prognosis[7] decreases following NUC-7738 treatment
- Protein expression of Cytochrome C, a marker of apoptosis increases following treatment

Figure 9: mIF of 3μm FFPE section show a decrease in the protein expression of key lipid metabolizing enzymes and an increase in the apoptotic marker cyt c following NUC-7738 treatment

CONCLUSION

- Novel methodology developed to analyse the spatial relationship between the lipidome and TME of patient biopsies within a single-tissue section
- Preliminary results suggest that NUC-7738 ± pembrolizumab reprograms cancer cell lipid metabolism in the TME
- This platform provides a powerful investigative tool to simultaneously explore the myriad of factors in the TME that contribute to tumorigenesis and therapeutic response

REFERENCES: 1. N. Koundouros, G. Poulogiannis, Br. J. Cancer 122 (2020) 4–22 2. H.-R. Jin, J. Wang, Z.-J. Wang, M.-J. Xi, B.-H. Xia, K. Deng, J.-L. Yang, J. Hematol. Oncol.J Hematol Oncol 16 (2023) 103 3. Y. Wu, X. Pu, X. Wang, M. Xu, Lipids Health Dis. 23 (2024) 35 4. M.E. King, R. Yuan, J. Chen, K. Pradhan, I. Sariol, S. Li, A. Chakraborty, O. Ekpenyong, J.H. Yearley, J.C. Wong, L. Zúñiga, D. Tomazela, M. Beaumont, J.-H. Han, L.S. Eberlin, J. Biol. Chem. 299 (2023) 102902 5. S. Reza, M. Ugorski, J. Suchański, Glycobiology 31 (2021) 1416–1434 6. N. Realini, F. Palese, D. Pizzirani, S. Pontis, A. Basit, A. Bach, A. Ganesan, D. Piomelli, J. Biol. Chem. 291 (2016) 2422–2434 7. M. Belleri, P. Chiodelli, M. Corli, M. Capra, M. Presta, Biochim. Biophys. Acta BBA - Rev. Cancer 1877 (2022) 188675

ABBREVIATIONS: 3'dATP: 3'deoxyadenosine Triphosphate AC: acid ceramidase β-galc: beta-galactosylceramidase CD36: cluster of differentiation 36 Cer: Ceramide Cyt C: cytochrome C DESI-MSI: desorption electrospray ionisation- mass spectrometry imaging FA: fatty acids FAO: fatty acid oxidation H&E: hematoxylin and eosin HexCer: hexosylceramide IHC: immunohistochemsitry miF: multiplex immunofluorescence MUFA: monosaturated fatty acids PCA: principal component analysis PUFA: polyunsaturated fatty acids ROI: region of interest S1P: sphingosine-1-phosphate SAM: significance of microarray SK-1: sphingosine kinase -1 TIC: total ion count TME: tumor microenvironment UMAP: uniform manifold approximation and projection

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.