NUC-7738 in combination with pembrolizumab in patients with metastatic melanoma: Phase 2 results from the NuTide:701 study

Sarah P Blagden[1], Stefan N Symeonides[2], Aglaia Skolariki[1], Noor Md Haris[3], Zhuang Boh[2], In Hwa Um[4], Mustafa Elshani[4,5], Alison L Dickson[4,5], Ying Zhang[4], David J Harrison[4,5], Fiona G McKissock[5], Elisabeth Oelmann[5], Jeffrey D Bloss[5], Natalie Cook[6], TR Jeffry Evans[7], E. Ruth Plummer[3]

1) Early Phase Clinical Trials Unit, Churchill Hospital, University of Oxford, Oxford, UK 2) Edinburgh ECMC, Western General Hospital, Edinburgh, UK 3) Newcastle ECMC, Northern Centre for Cancer Care, Freeman Hospital, Newcastle upon Tyne, UK 4) University of St Andrews, St Andrews, UK 5) NuCana plc, Edinburgh, UK 6) The Christie NHS Trust/University Manchester, Manchester, UK 7) Beatson West of Scotland Cancer Centre/University of Glasgow, UK

Poster Number: C032 **Email:** sarah.blagden@oncology.ox.ac.uk

BACKGROUND

Metastatic Melanoma & Immune Checkpoint Inhibitors

- Patients with metastatic melanoma have limited treatment options after progression on immune checkpoint inhibitors (ICIs) and BRAF/MEK inhibitors
 - Particular unmet need for patients who are primary refractory to ICIs
- Resistance to ICIs may be associated with secreted forms of PD-L1, which are partially controlled by alternative polyadenylation[1]

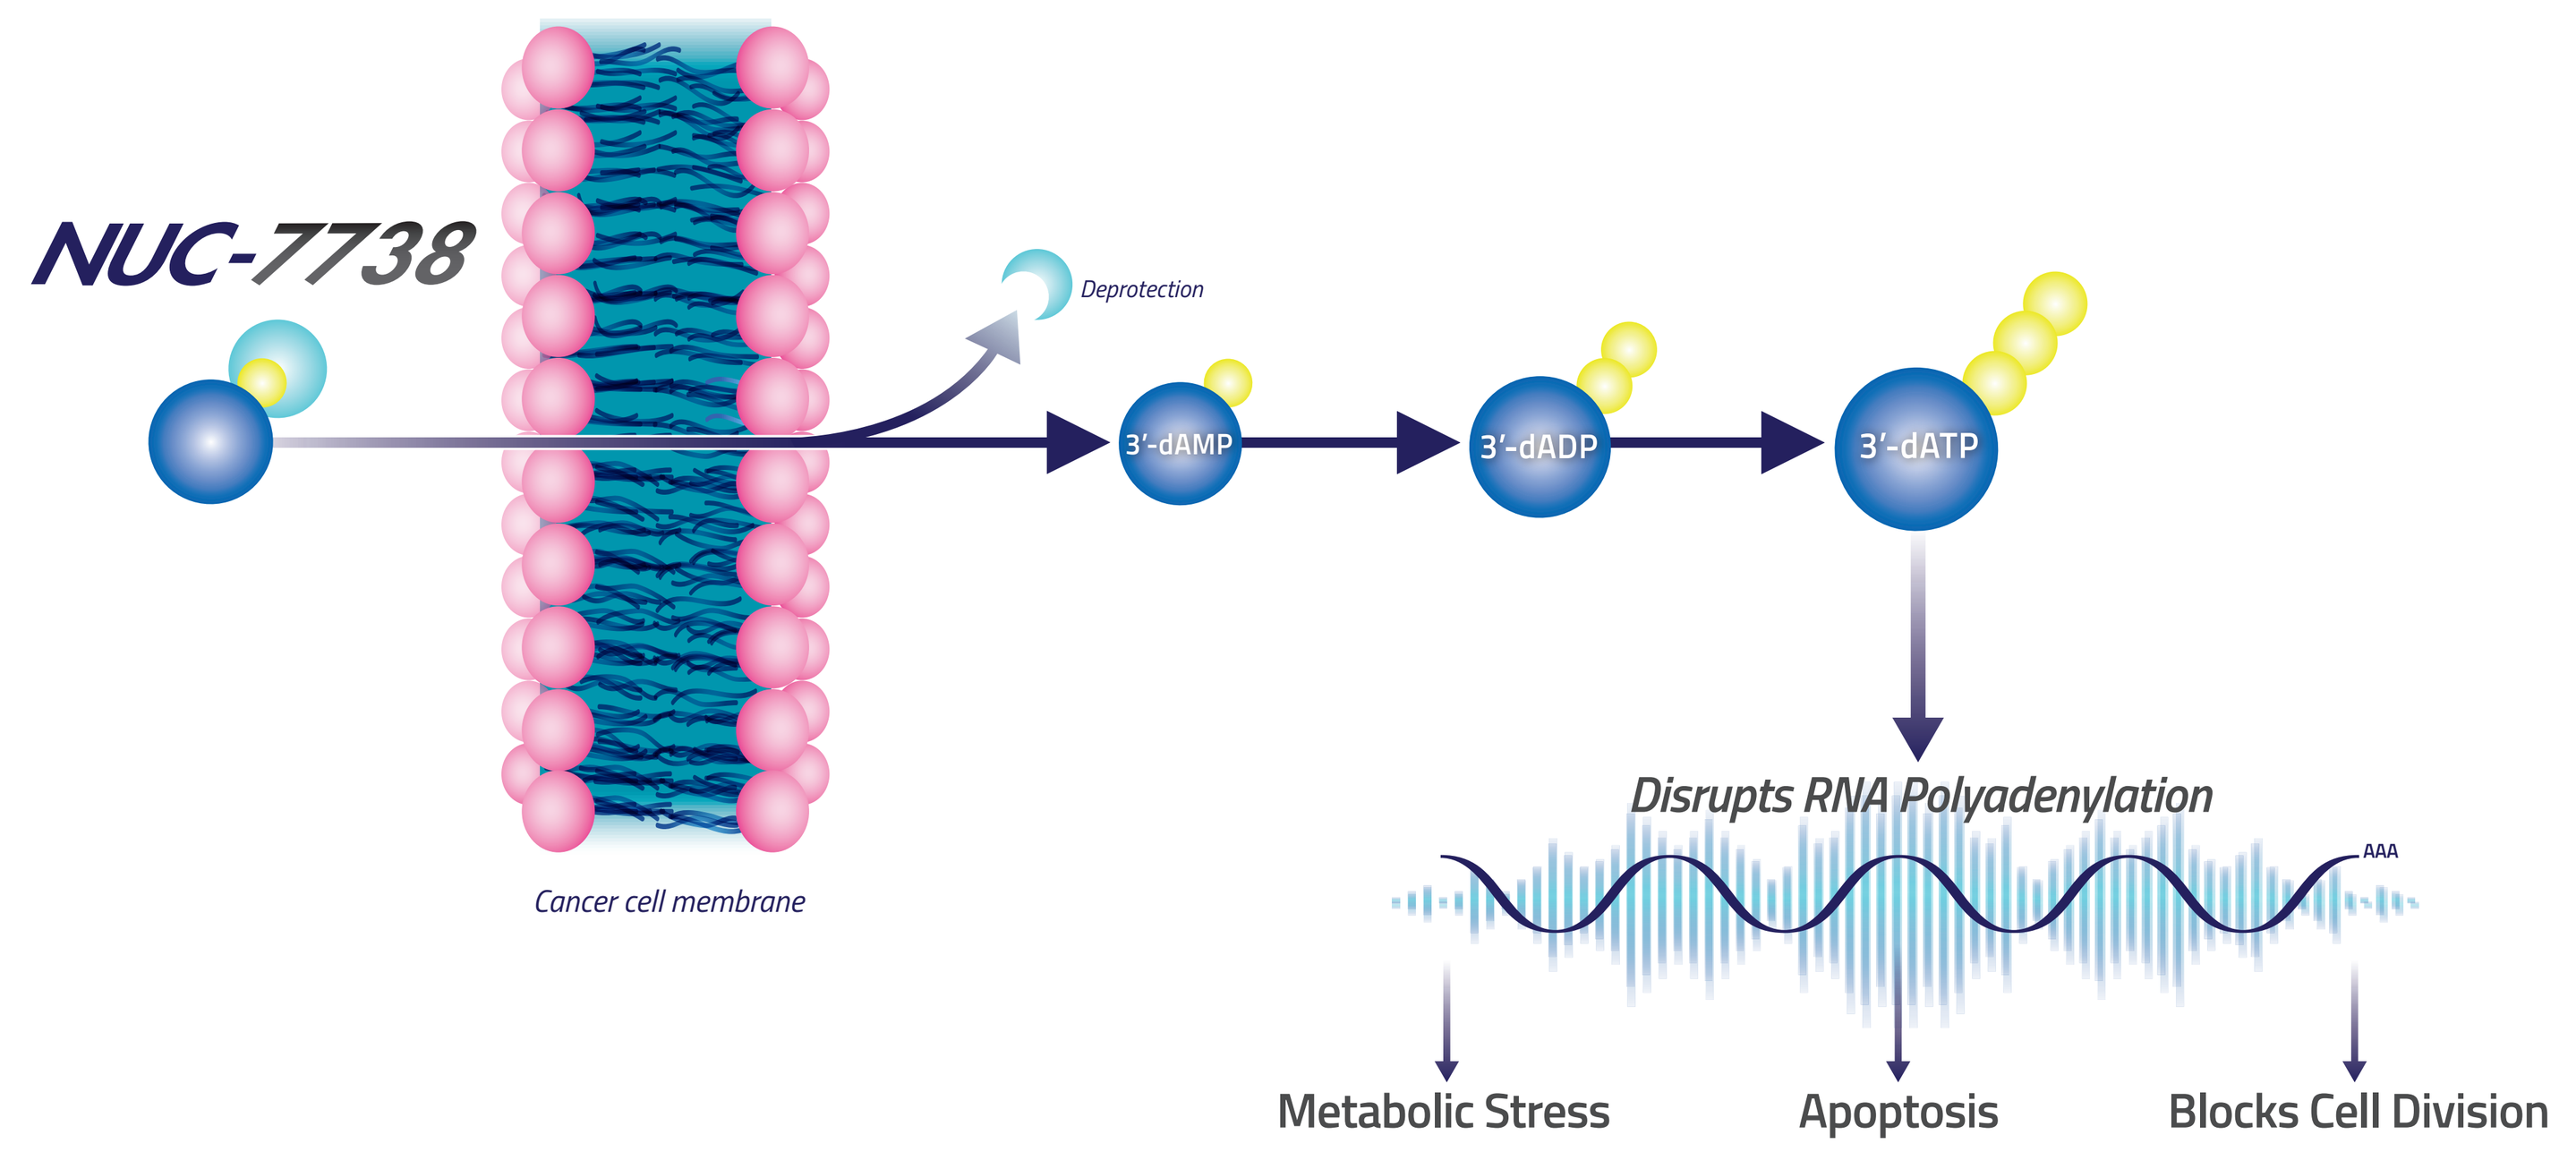
NUC-7738
Deprotection
3′-dAMP
3′-dADP
3′-dATP
Cancer cell membrane
Disrupts RNA Polyadenylation
AAA
Metabolic Stress
Apoptosis
Blocks Cell Division

NUC-7738: ProTide transformation of 3′-dA

- Generates high intracellular levels of active anti-cancer metabolite (3′-dATP)
- 3′-dATP induces transcriptional changes in genes involved in key cellular processes including metabolism, apoptosis, cell differentiation[2-5]
- 3′-dATP is associated with changes in polyadenylation
- Reduces secreted forms of PD-L1 and may enhance the clinical utility of PD-(L)1 agents[6]

NuTide:701 Study Design

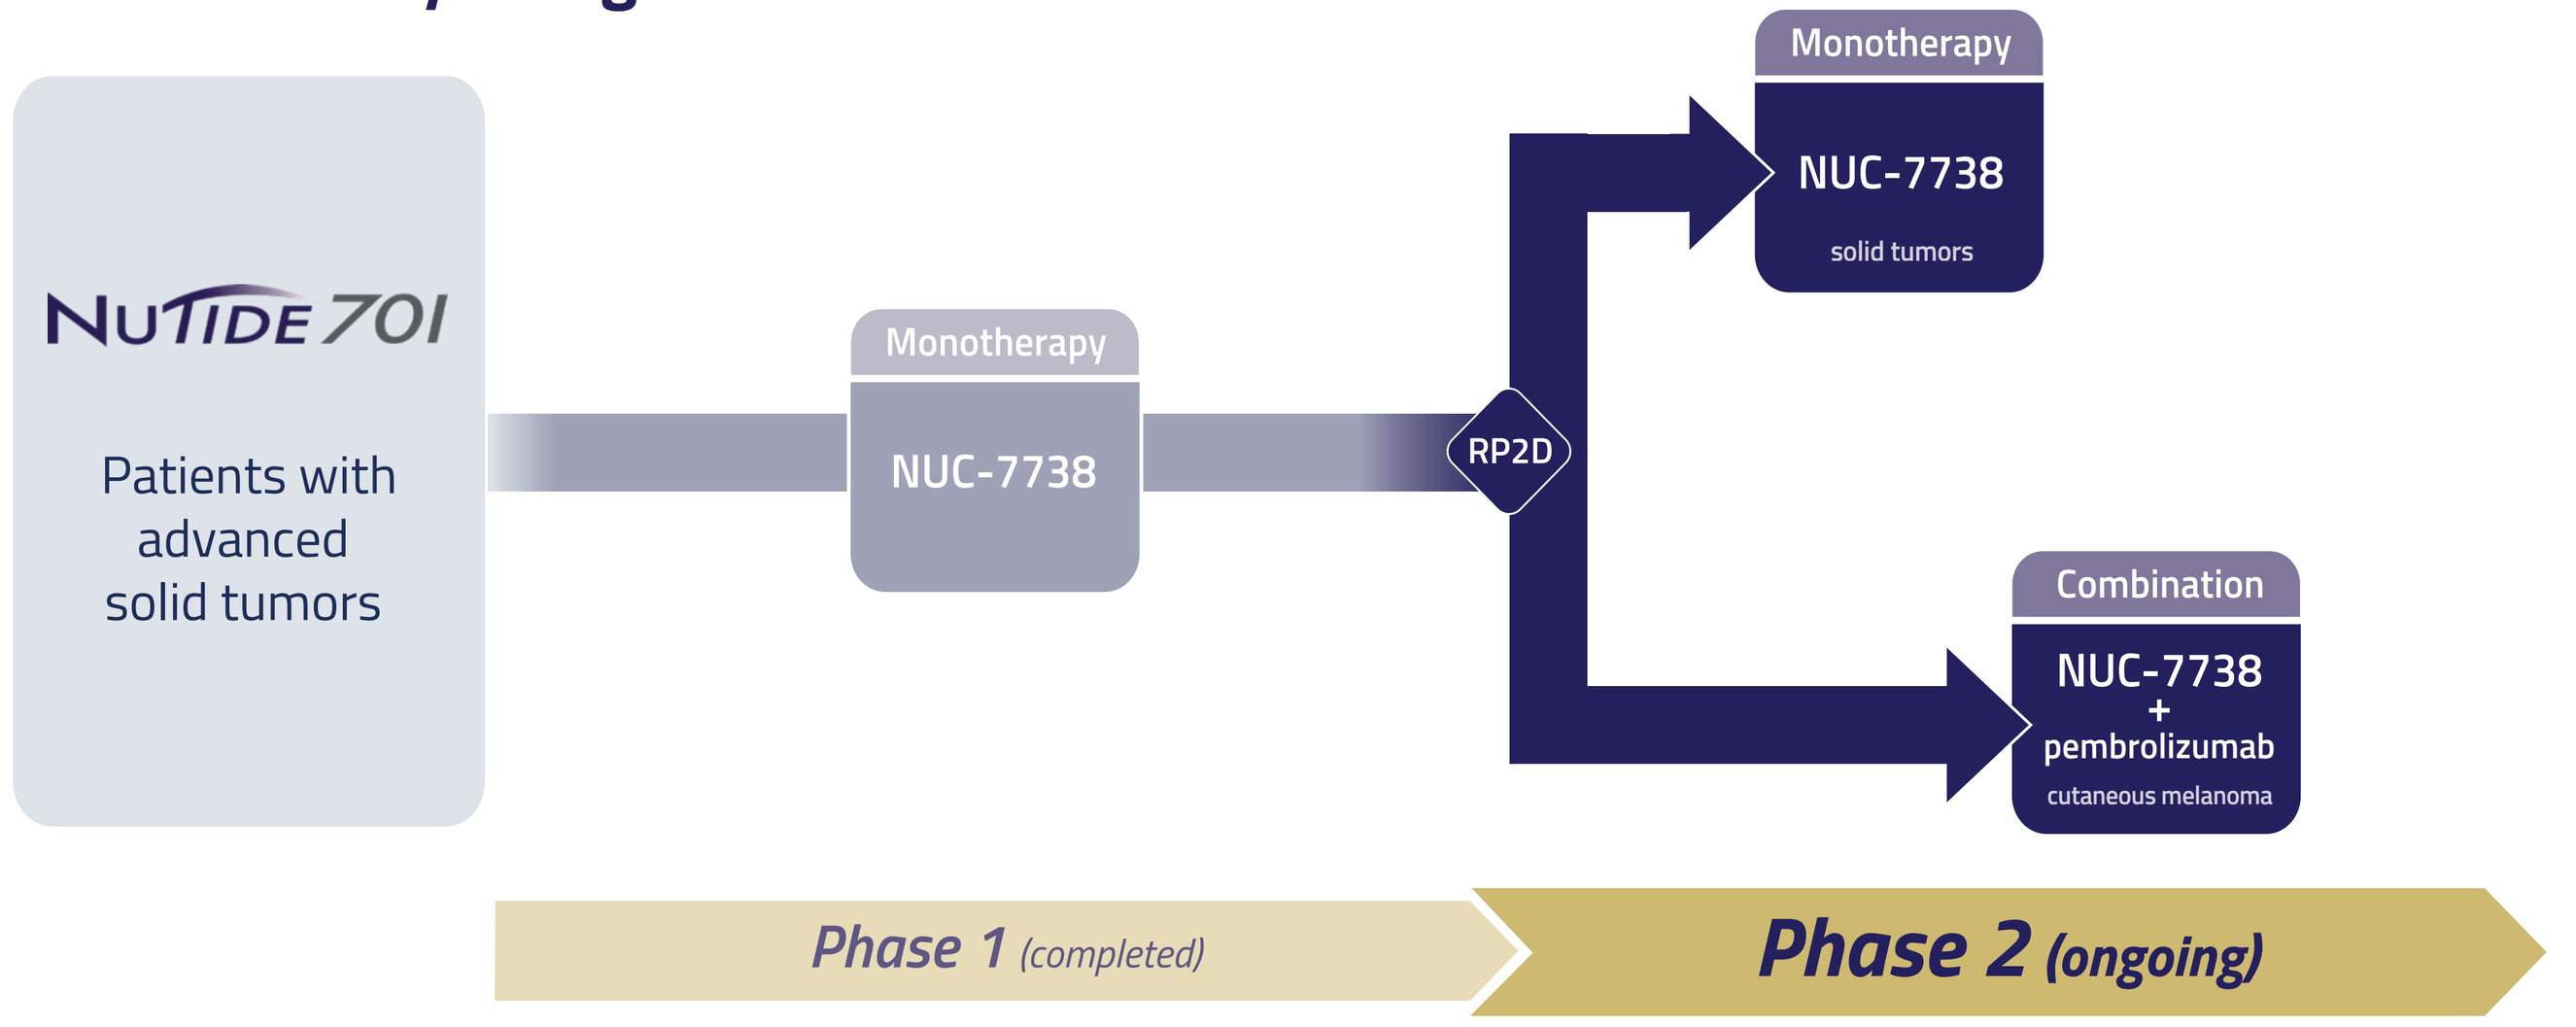
NuTide 701
Patients with advanced solid tumors
Monotherapy
NUC-7738
RP2D
Monotherapy
NUC-7738
solid tumors
Combination
NUC-7738 + pembrolizumab
cutaneous melanoma
Phase 1 (completed)
Phase 2 (ongoing)

Objectives: Anti-cancer activity, safety, PK of NUC-7738 in combination with pembrolizumab

Methods

Cell cultures: Human melanoma cells (SK-MEL28) treated with DMSO or NUC-7738 (IC_{50} levels). RNA extracted 24 h post-treatment.

Patient paired tumor biopsies: Patients received either NUC-7738 monotherapy (ranging from 900-1350 mg/m² on days 1 & 8 of a 14-day cycle) or NUC-7738 (1125 mg/m² IV infusion over 2 h on days 1, 8, & 15 of a 21-day cycle) in combination with pembrolizumab (200 mg IV over 30 min prior to NUC-7738 on day 1). Biopsies collected pre- and post-drug infusion (≤ 6 h post infusion).

PBMCs samples: NuTide:701 patient (2000 mg/m² NUC-7738) and healthy volunteers (treated *in vitro* with NUC-7738).

Intracellular metabolites: NUC-7738 and 3′-dATP levels determined by LC-MS (Biopsy tissue: NUC-7738 & 3'-dATP LLOQ = 5 ng/g; PBMCs: NUC-7738 LLOQ = 0.5 nM, 3'-dATP LLOQ = 20 nM).

PolyATail size: Determined with tailfindr R package.

Gene expression mapping and counting: Sequencing reads aligned to transcriptomic reference. Gene-level expression quantification performed with Salmon long-read counting mode *-ont*. Differential gene expression performed using R package edgeR with *-exactTest* option for samples without biological replicates. Heatmaps generated using hierarchical clustering to group genes based on expression similarity.

JESS Western blot: RAB27A and MITF protein levels immunoprobed using capillary Western blots.

Immunofluorescence: Paraffin embedded sections including antibodies specific to RAB27A, MITF, CD8 and PD-1.

RESULTS

NUC-7738 generates 3′-dATP in patients' biopsies

Patient biopsies	Concentration (ng/g) NUC-7738	3′-dATP
Cutaneous Melanoma	51	376
Endometrial	20	22
Mesothelioma	<LLOQ*	72

*LLOQ (NUC-7738) = 5 ng/g

NUC-7738 generates equivalent levels in patient PBMCs vs *in vitro* PBMCs

PBMCs	C_{max} (pmol/10⁶ cells) NUC-7738	3′-dATP
From patient (2000 mg/m²)	2	36
From volunteer *in vitro* (10 μM)	2	55
From volunteer *in vitro* (50 μM)	9	181

NUC-7738 causes marked effects on polyadenylation & transcription

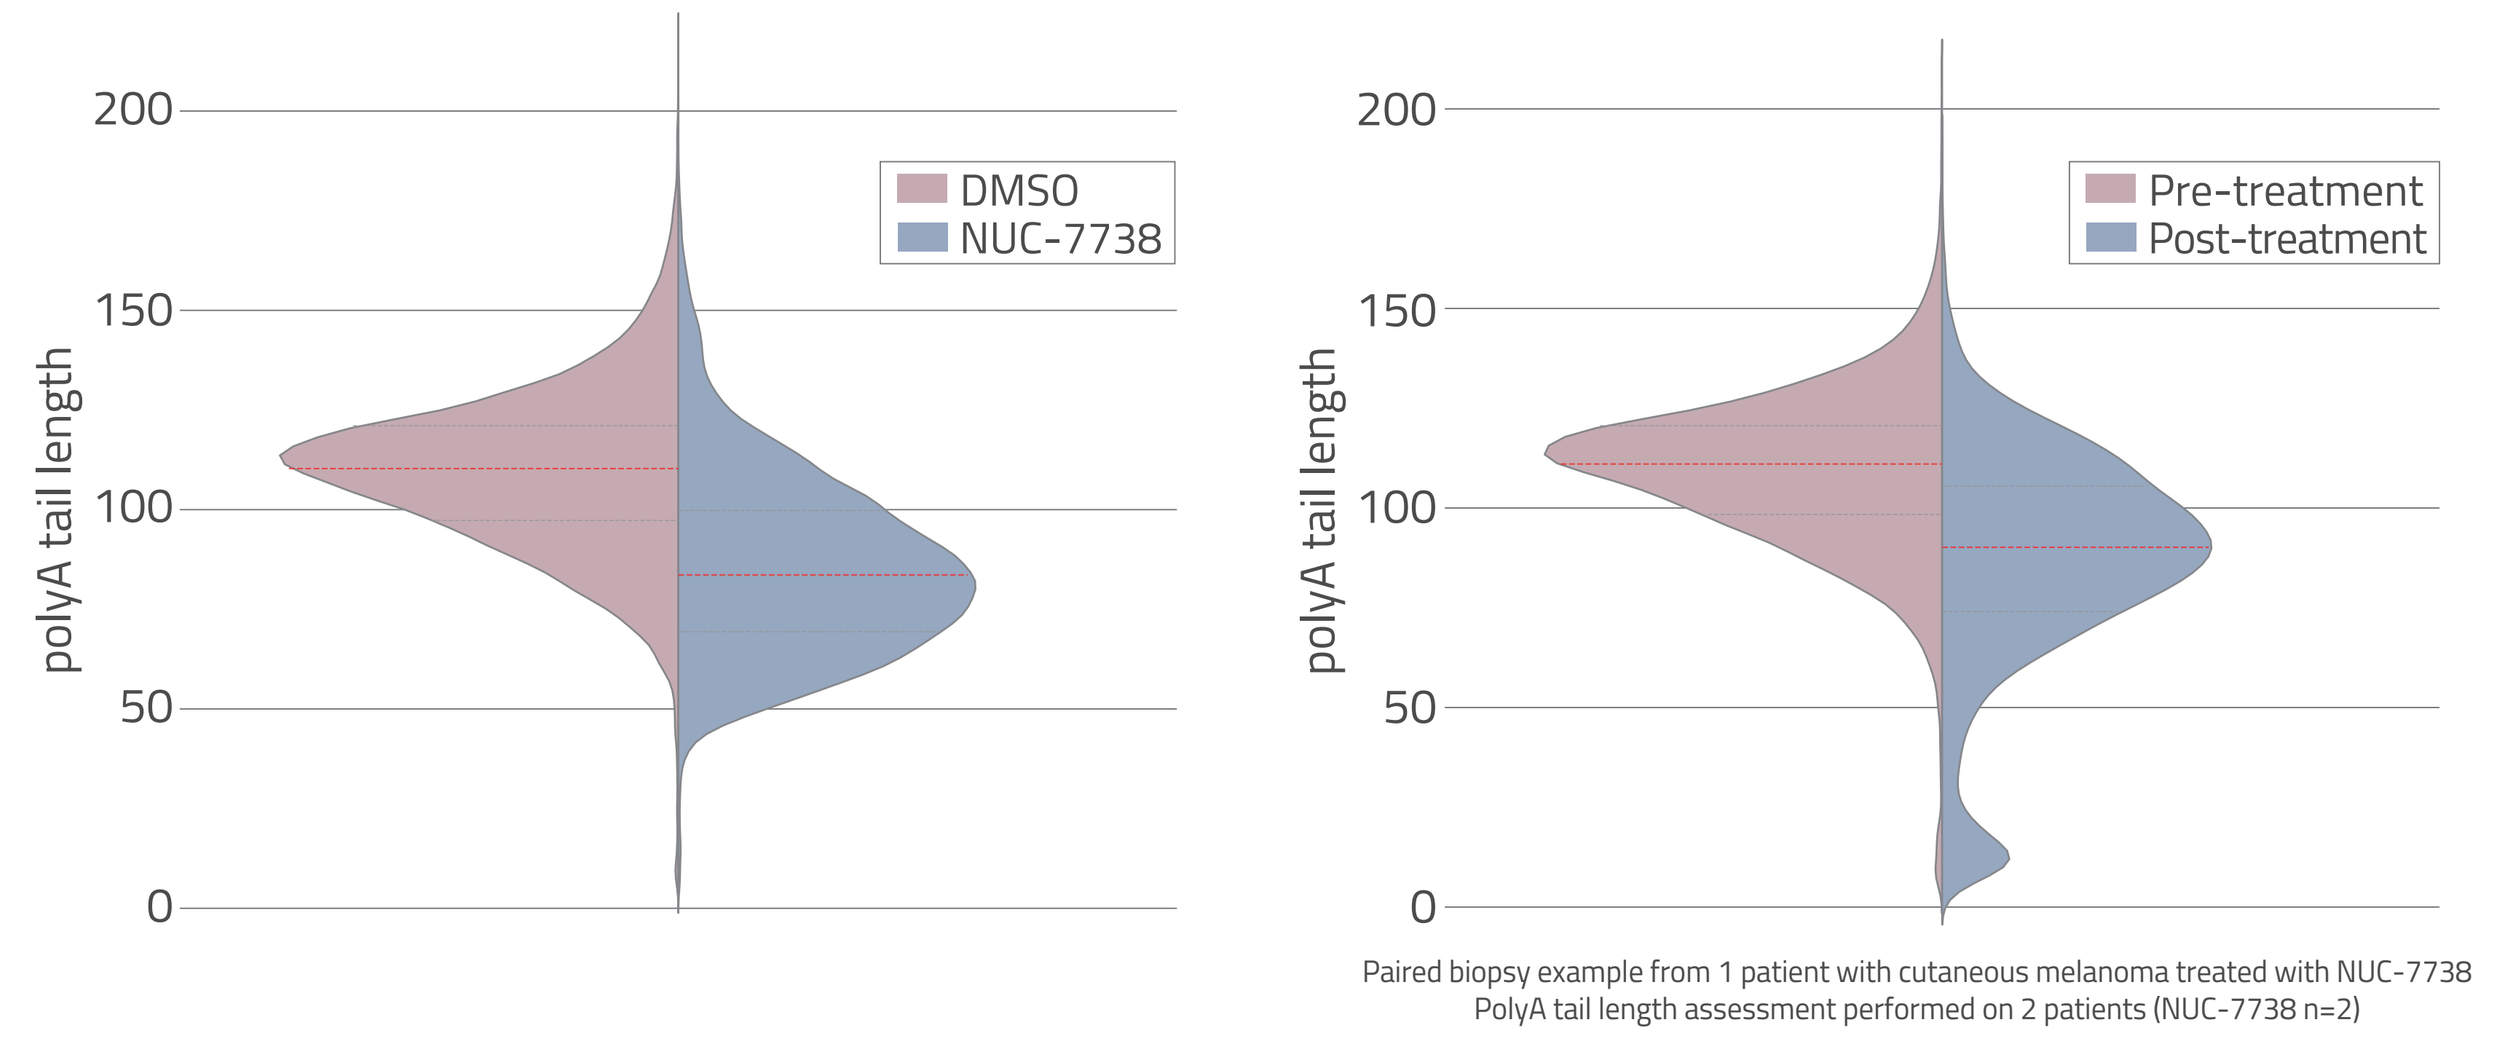
Melanoma cell line
Cutaneous melanoma biopsy
polyA tail length
DMSO
NUC-7738
Pre-treatment
Post-treatment

Paired biopsy example from 1 patient with cutaneous melanoma treated with NUC-7738
PolyA tail length assessment performed on 2 patients (NUC-7738 n=2)

- NUC-7738 caused a shortening of polyA tails (blue vs red) in both a tumor biopsy from a patient and a melanoma cell line

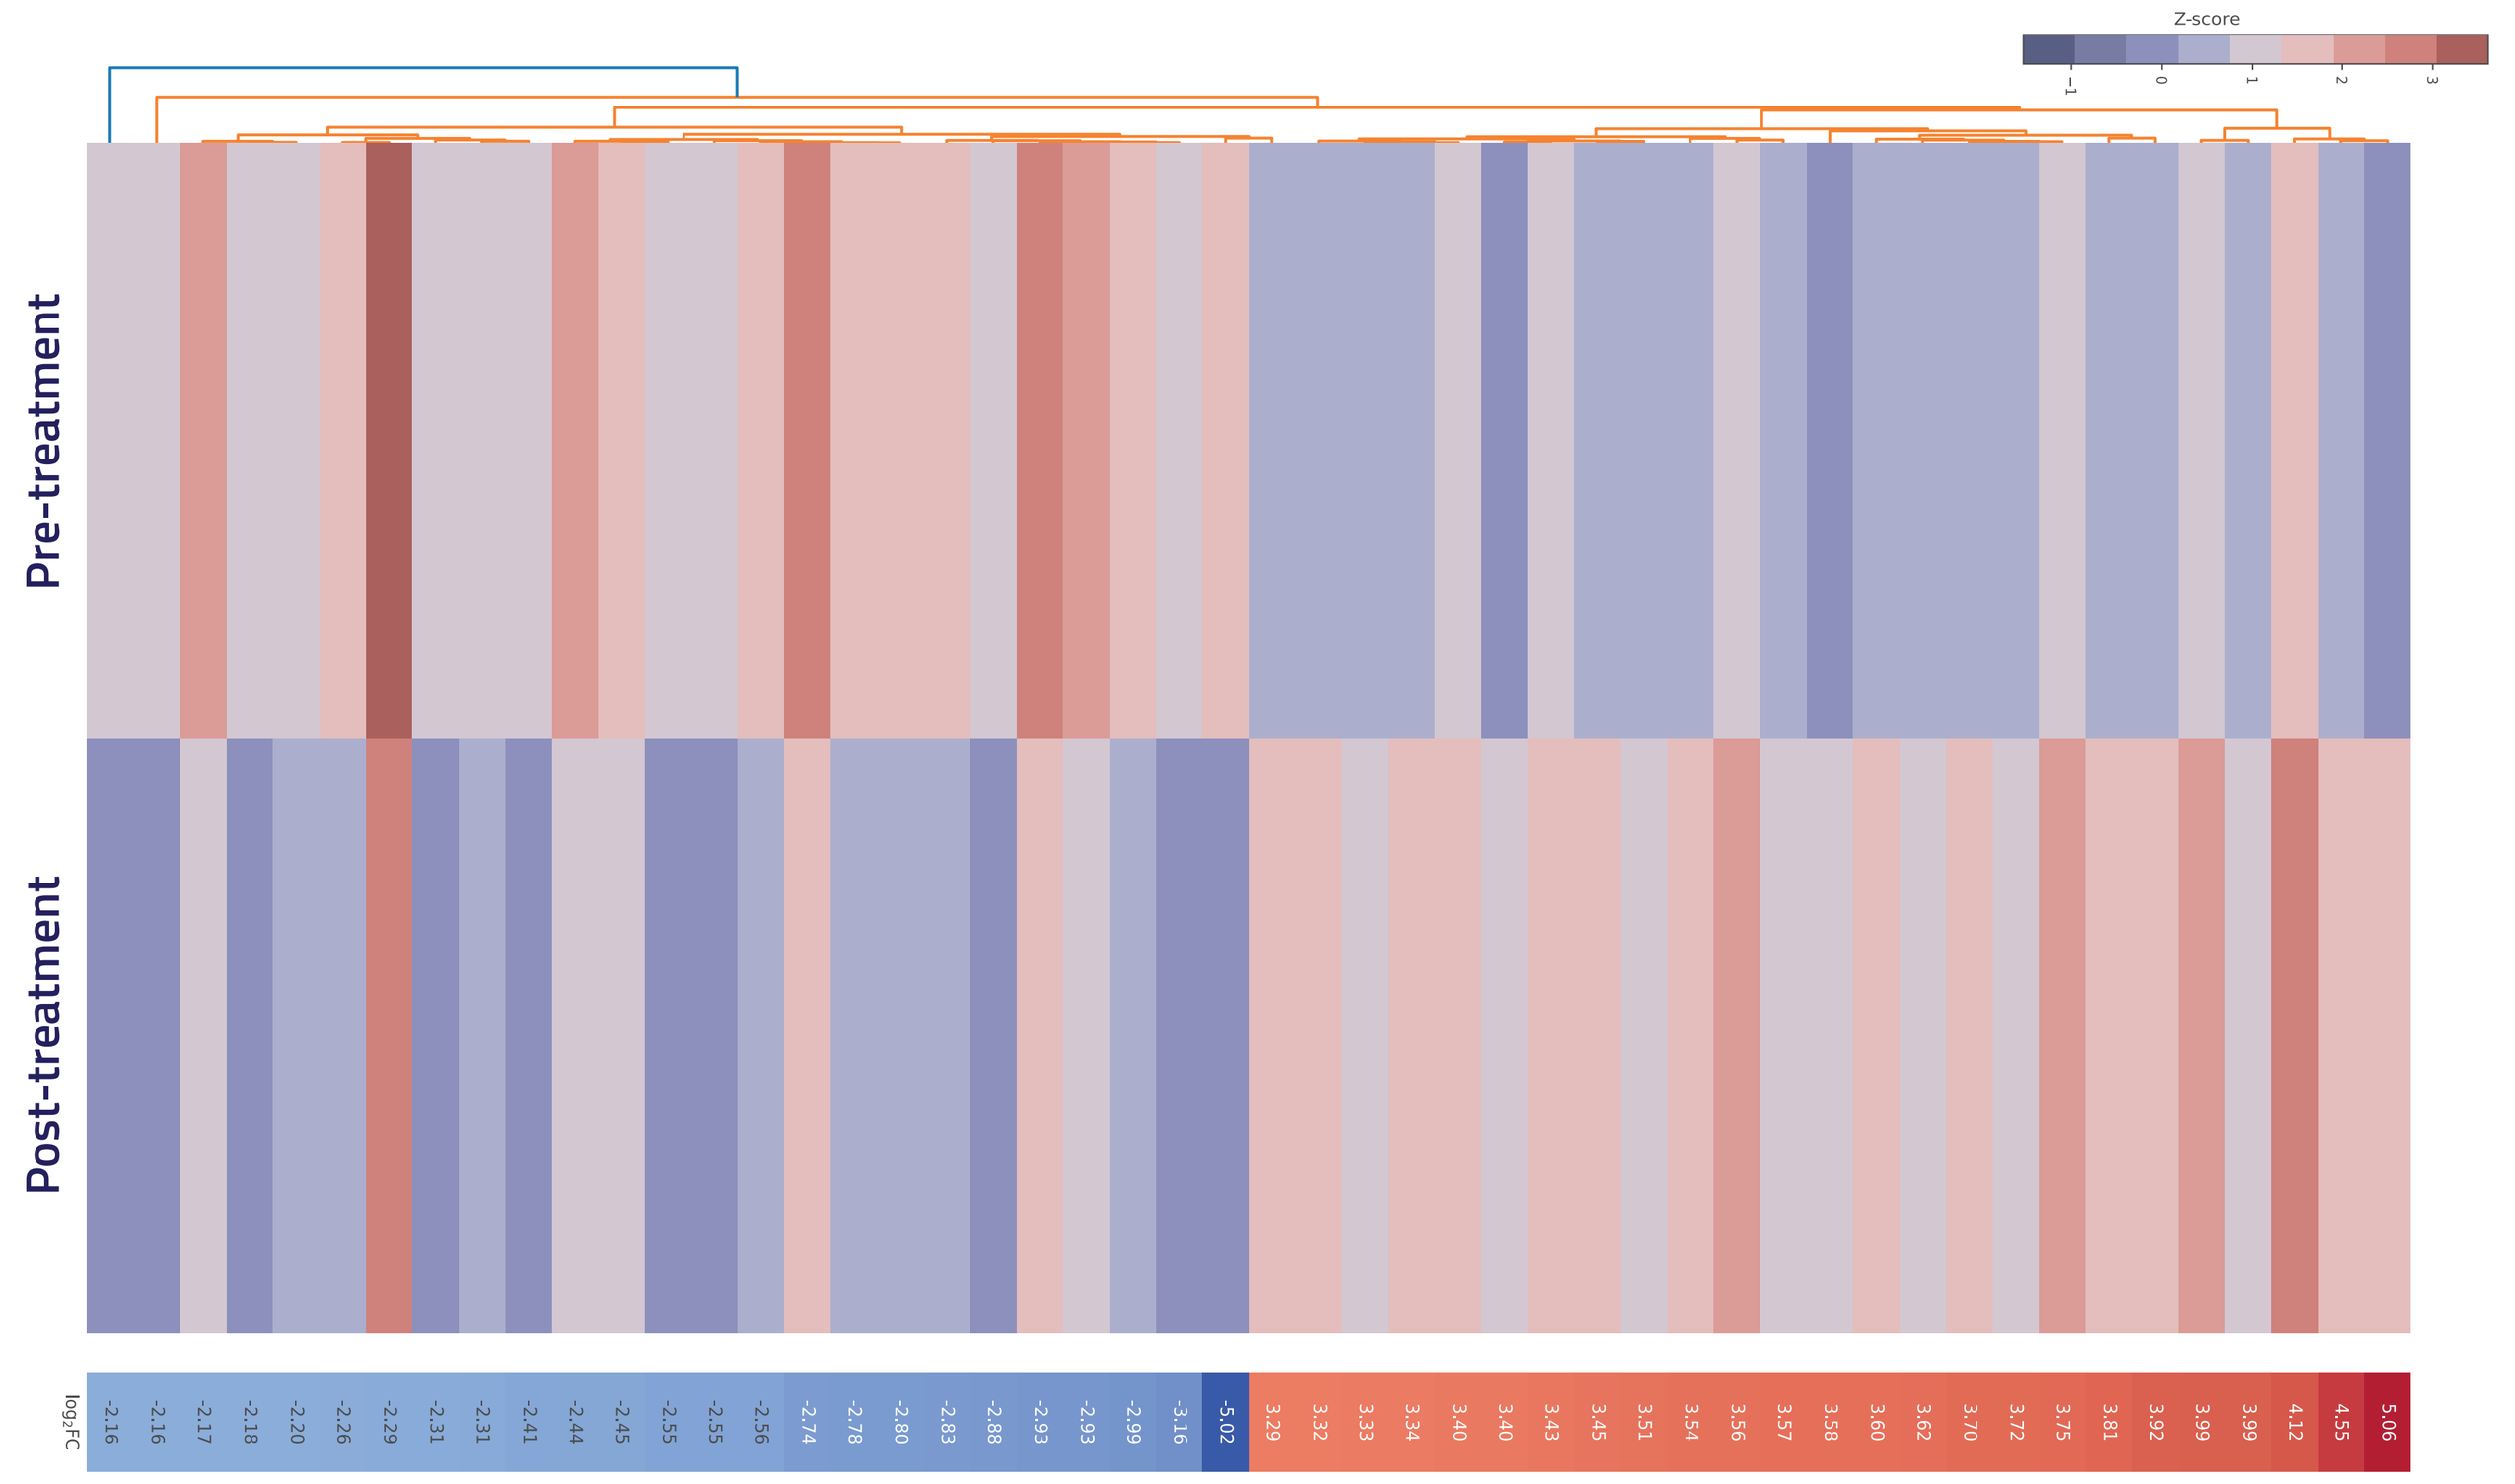
Z-score
Pre-treatment
Post-treatment
log2FC

Paired biopsy example from 1 patient with cutaneous melanoma treated with NUC-7738 + pembro
mRNA levels performed on 4 patients (NUC-7738 n=2; NUC-7738 + pembro n=2)

- NUC-7738 ± pembrolizumab causes major changes in mRNA expression in patients' biopsies

NUC-7738 reduces pro-tumorigenic factors RAB27A & MITF

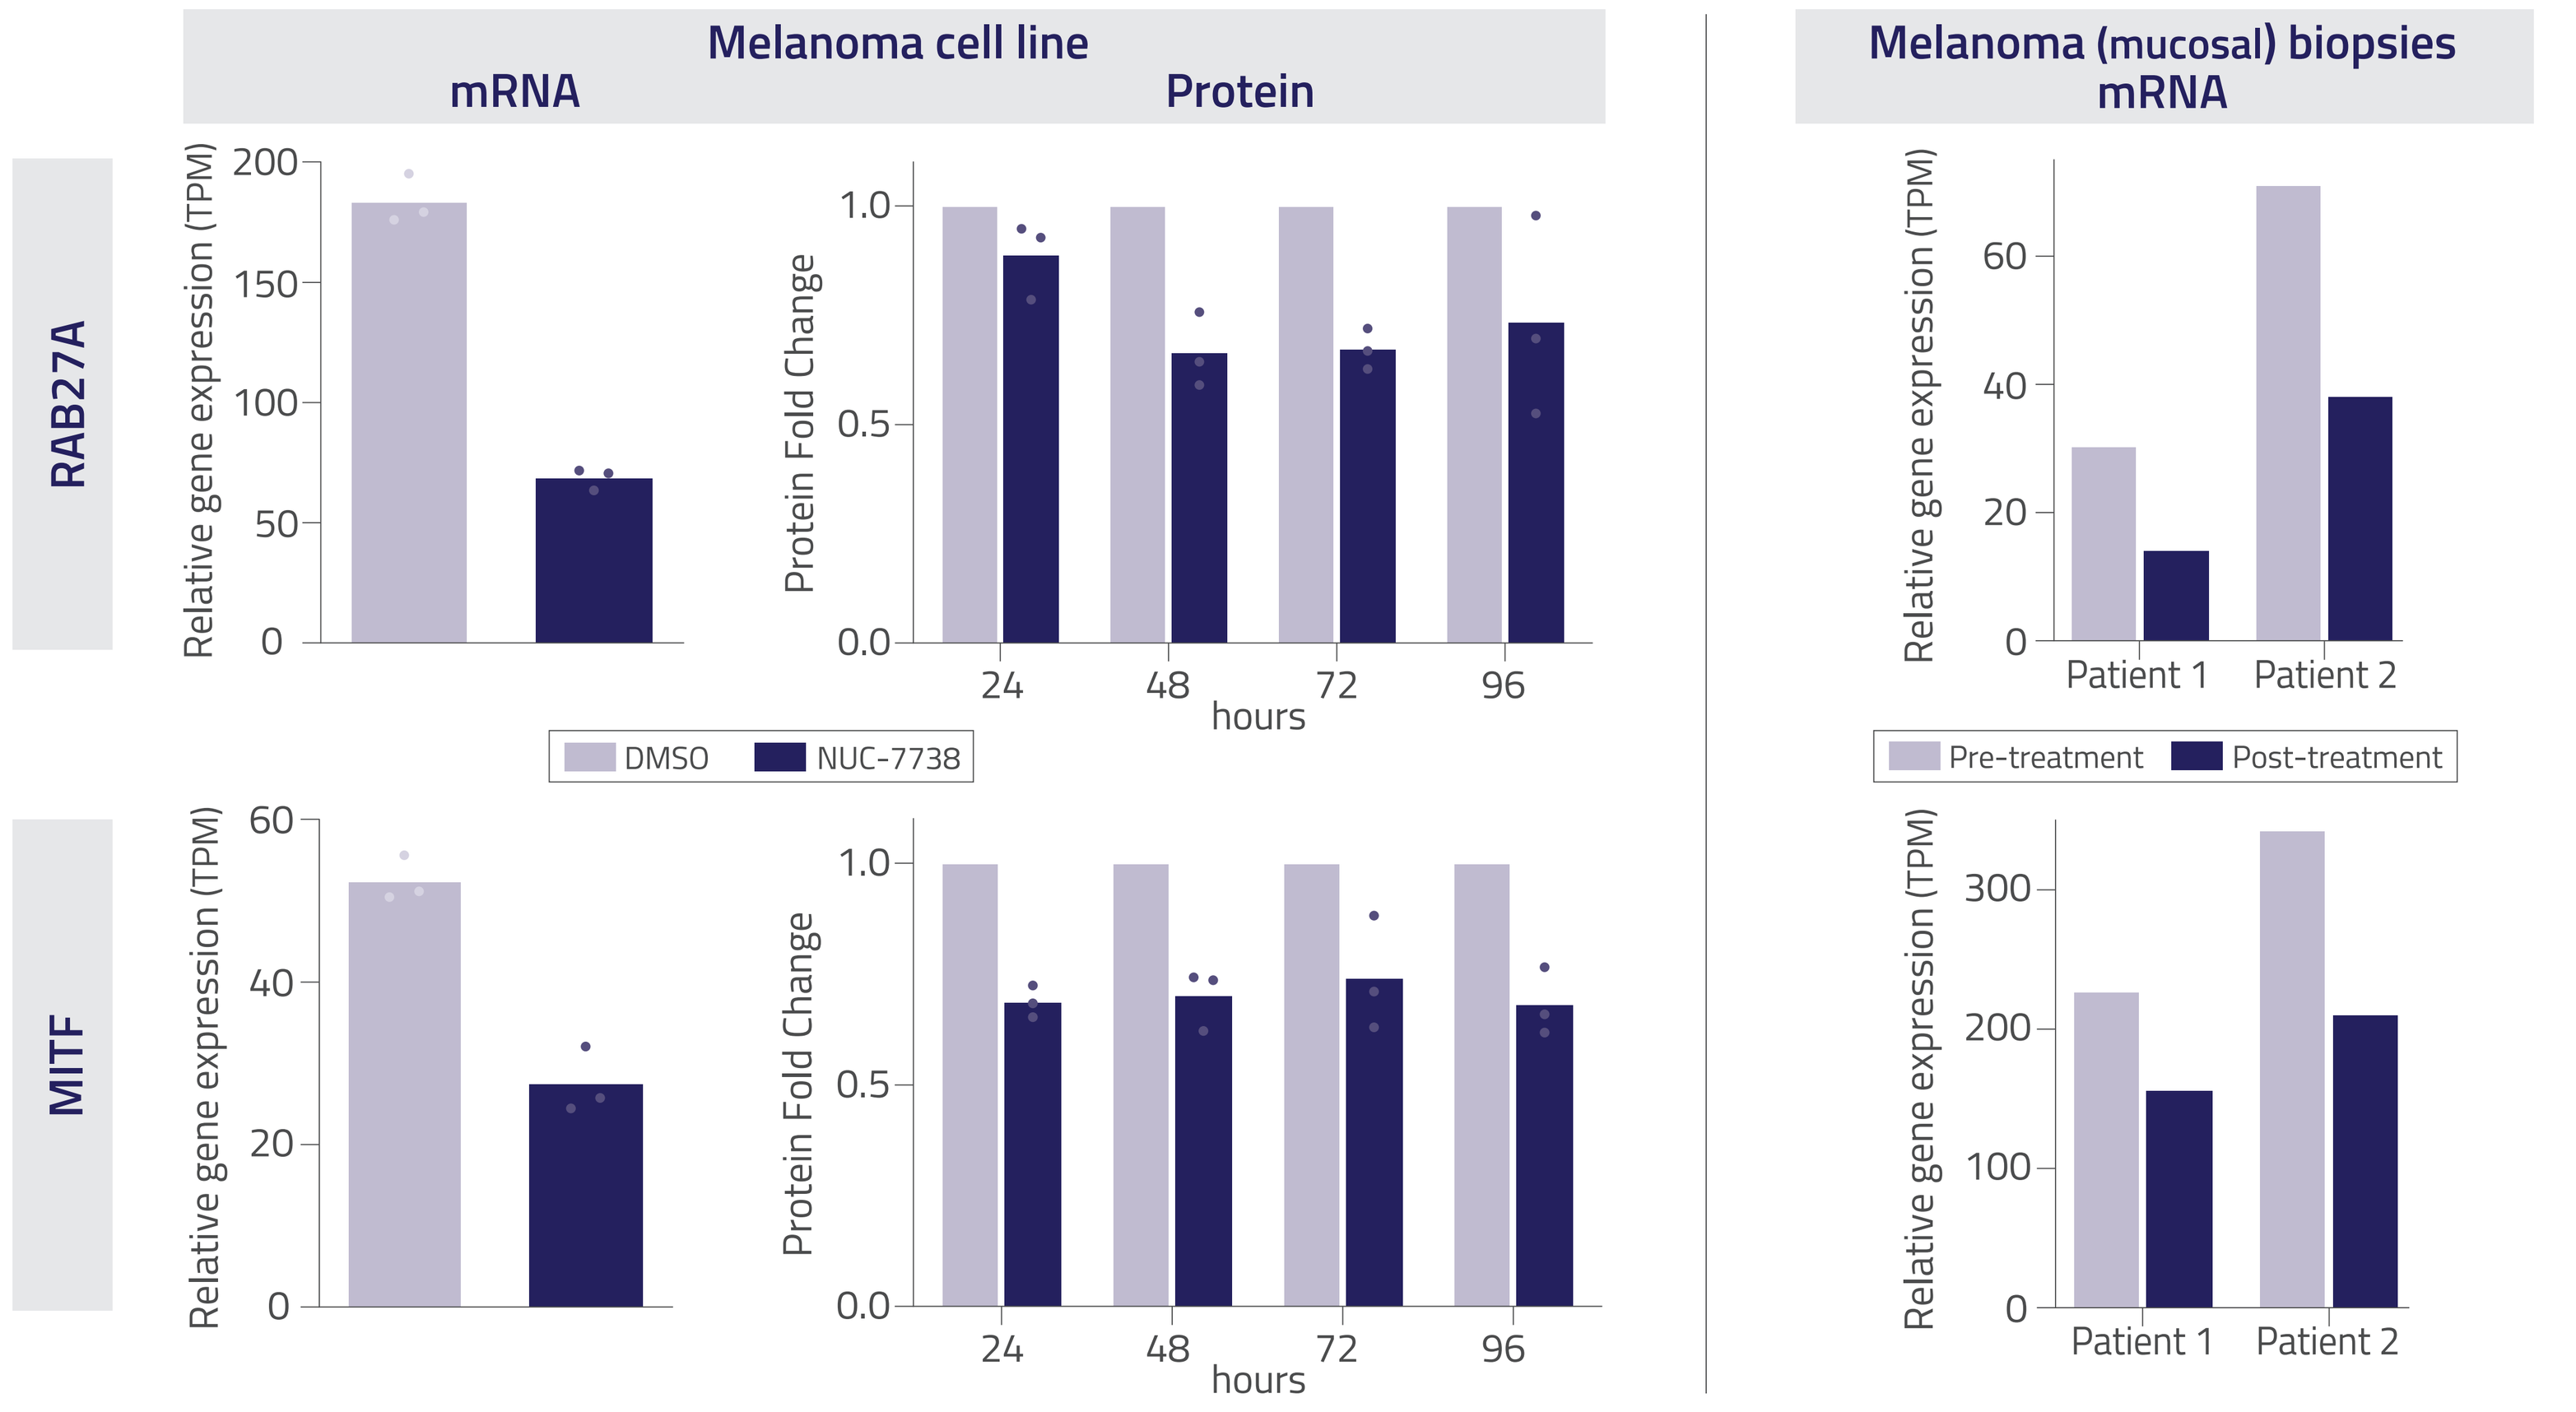
Melanoma cell line
mRNA
Protein
Melanoma (mucosal) biopsies
mRNA
RAB27A
MITF
Relative gene expression (TPM)
Protein Fold Change
hours
DMSO
NUC-7738
Patient 1
Patient 2
Pre-treatment
Post-treatment

- NUC-7738 reduces RAB27A and MITF mRNA in patients' biopsies, similar to that observed in melanoma cell lines[6]

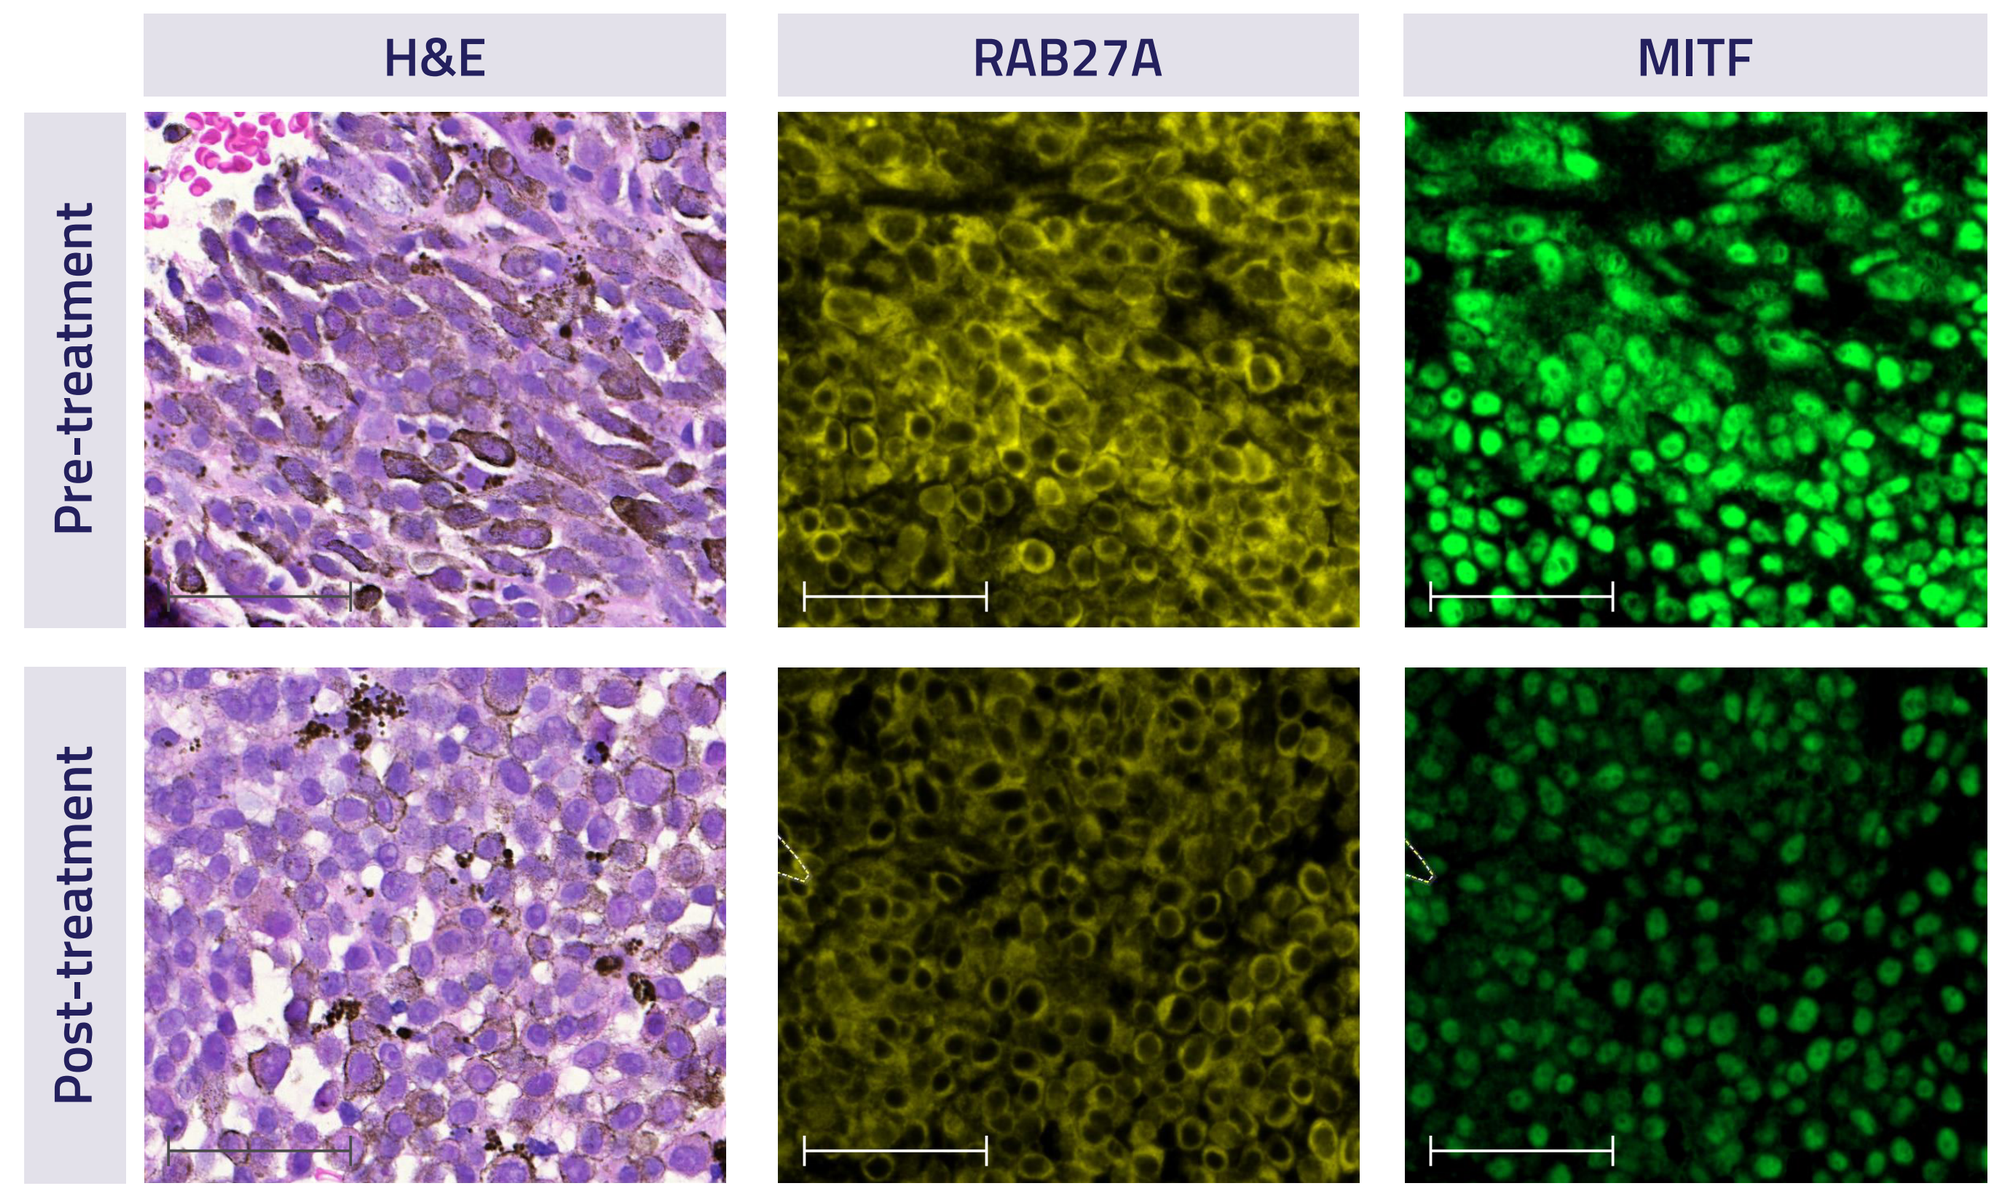
H&E
RAB27A
MITF
Pre-treatment
Post-treatment

Paired biopsy example from 1 patient with cutaneous melanoma treated with NUC-7738
Full immuno panel performed on 7 patients (NUC-7738 n=5; NUC-7738 + pembro n=2)

scale bar = 50 μM

- H&E sections show similar histologically pigmented melanoma (lymphocyte poor)
- RAB27A and MITF protein levels reflect changes observed in mRNA after NUC-7738 treatment

NUC-7738 modulates tumor morphology & immune cell infiltration

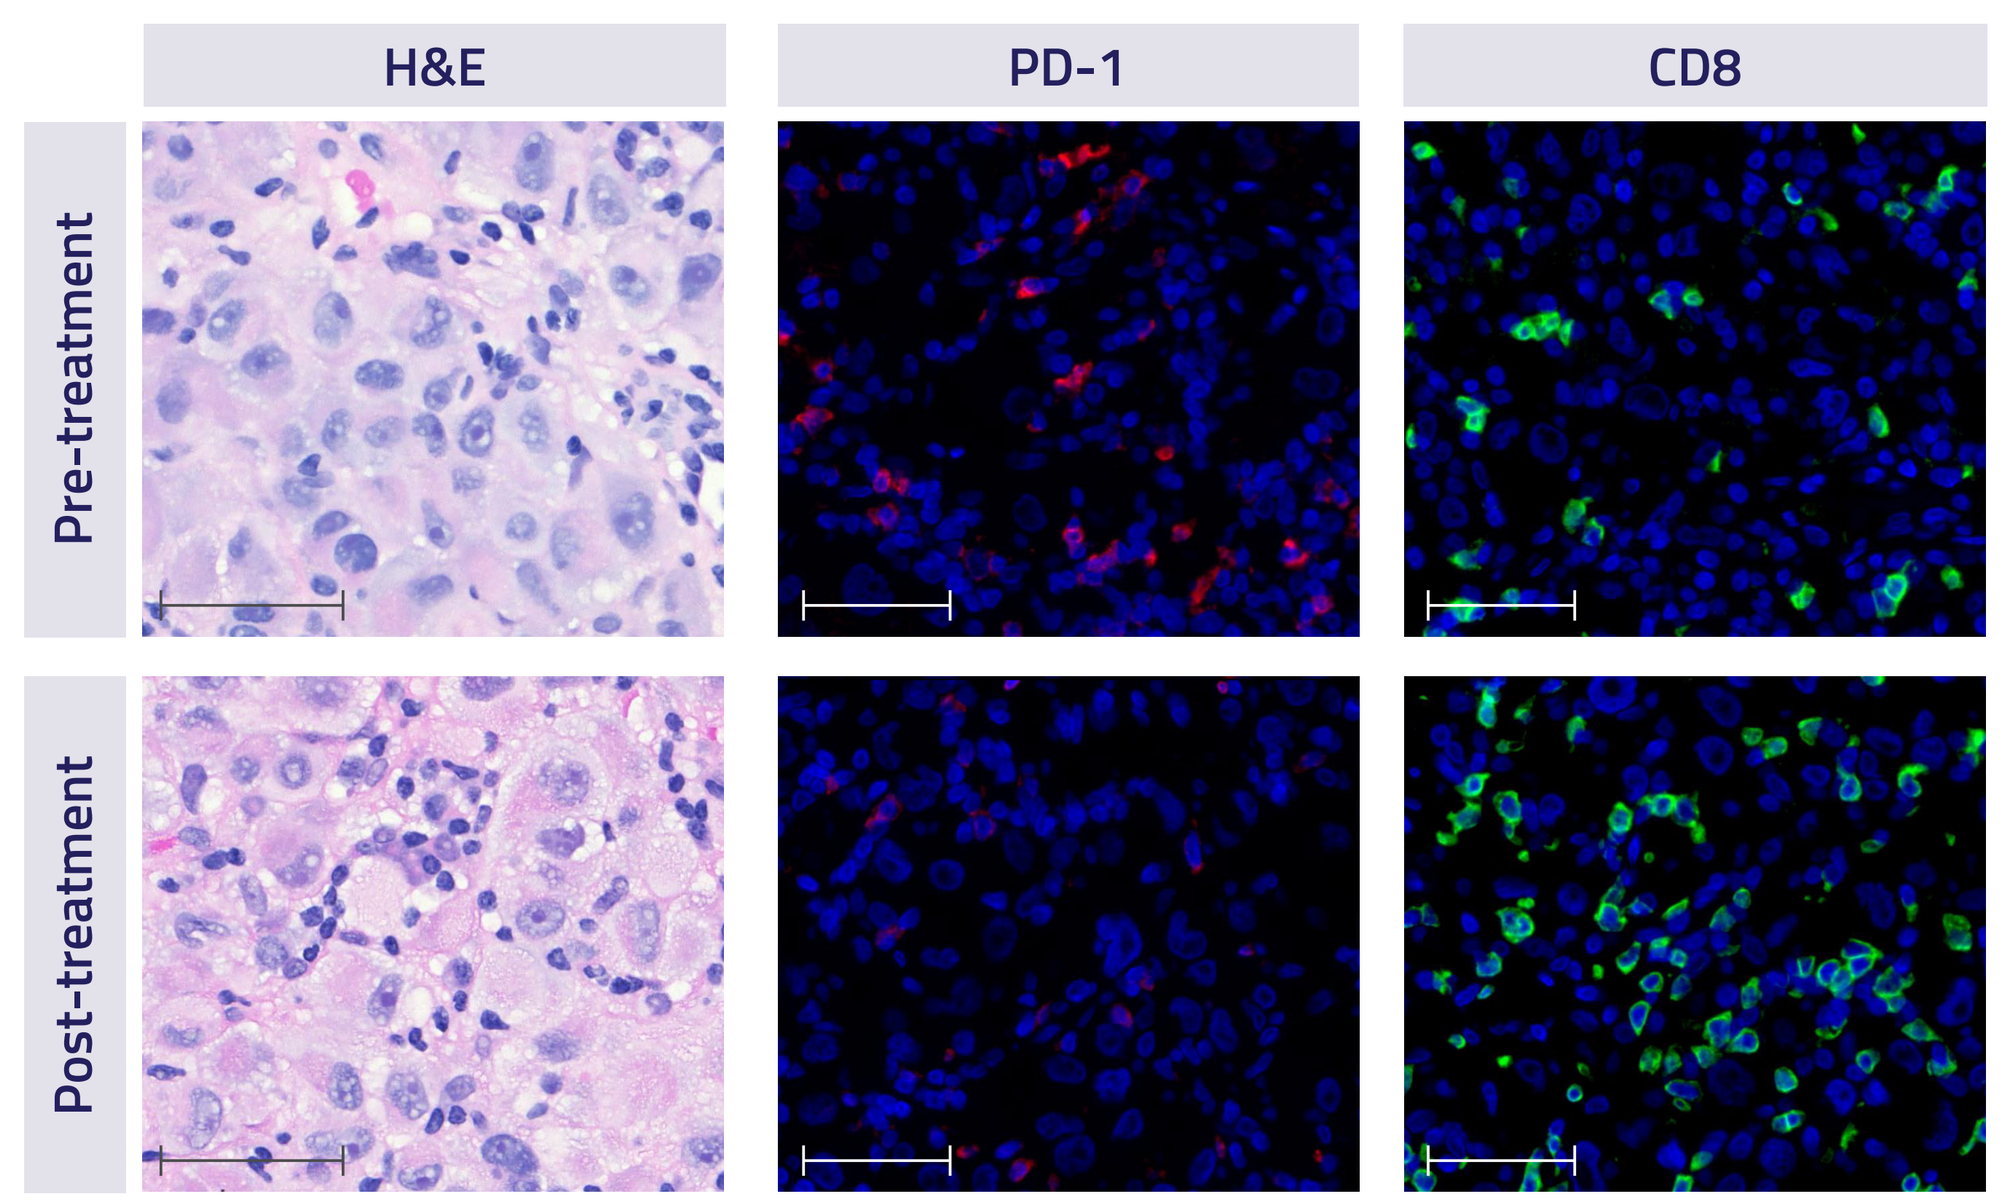
H&E
PD-1
CD8
Pre-treatment
Post-treatment

Paired biopsy example from 1 patient with cutaneous melanoma treated with NUC-7738 + pembro

scale bar = 50 μM

- NUC-7738 + pembrolizumab results in morphological cell changes (more prominent nucleoli and cytoplasmic vacuolation) and lymphoid infiltration (H&E sections of inflamed tumor)
- NUC-7738 + pembrolizumab results in a decreased number of PD-1 expressing lymphocytes (red)
- NUC-7738 + pembrolizumab results in an increased number of CD8⁺ T-cells (green)

Phase 1 (completed)

NUC-7738 monotherapy dose finding study in previously treated patients with advanced solid tumors

- MTD established: 1350 mg/m²

Phase 2 (ongoing)

- Monotherapy (completed): Intra-patient dose refinement (900-1350 mg/m² NUC-7738) in previously treated patients with advanced solid tumors. Starting dose established: 1125 mg/m²
- Combination therapy (ongoing): 1125 mg/m² NUC-7738* + 200 mg pembrolizumab in patients with advanced cutaneous melanoma previously treated with anti-PD-1 therapy

*escalation to 1350 mg/m² NUC-7738 permitted

Phase 2: NUC-7738 monotherapy (completed)

Treatment related AEs (n=11)

Preferred term	All Grades	Grade 3
Nausea	5 (46%)	0
Anemia	5 (46%)	1 (9%)
Vomiting	4 (36%)	0
Fatigue	4 (36%)	1 (9%)
Diarrhea	3 (27%)	0
Constipation	2 (18%)	0
Blood creatinine increased	2 (18%)	0
Flushing	2 (18%)	0
Infusion related reaction	2 (18%)	0

Most frequent (≥10% population) NUC-7738 related AEs

- No G4 TRAEs
- 1 patient with treatment related SAE; G3 acute kidney injury
- No discontinuations due to AEs

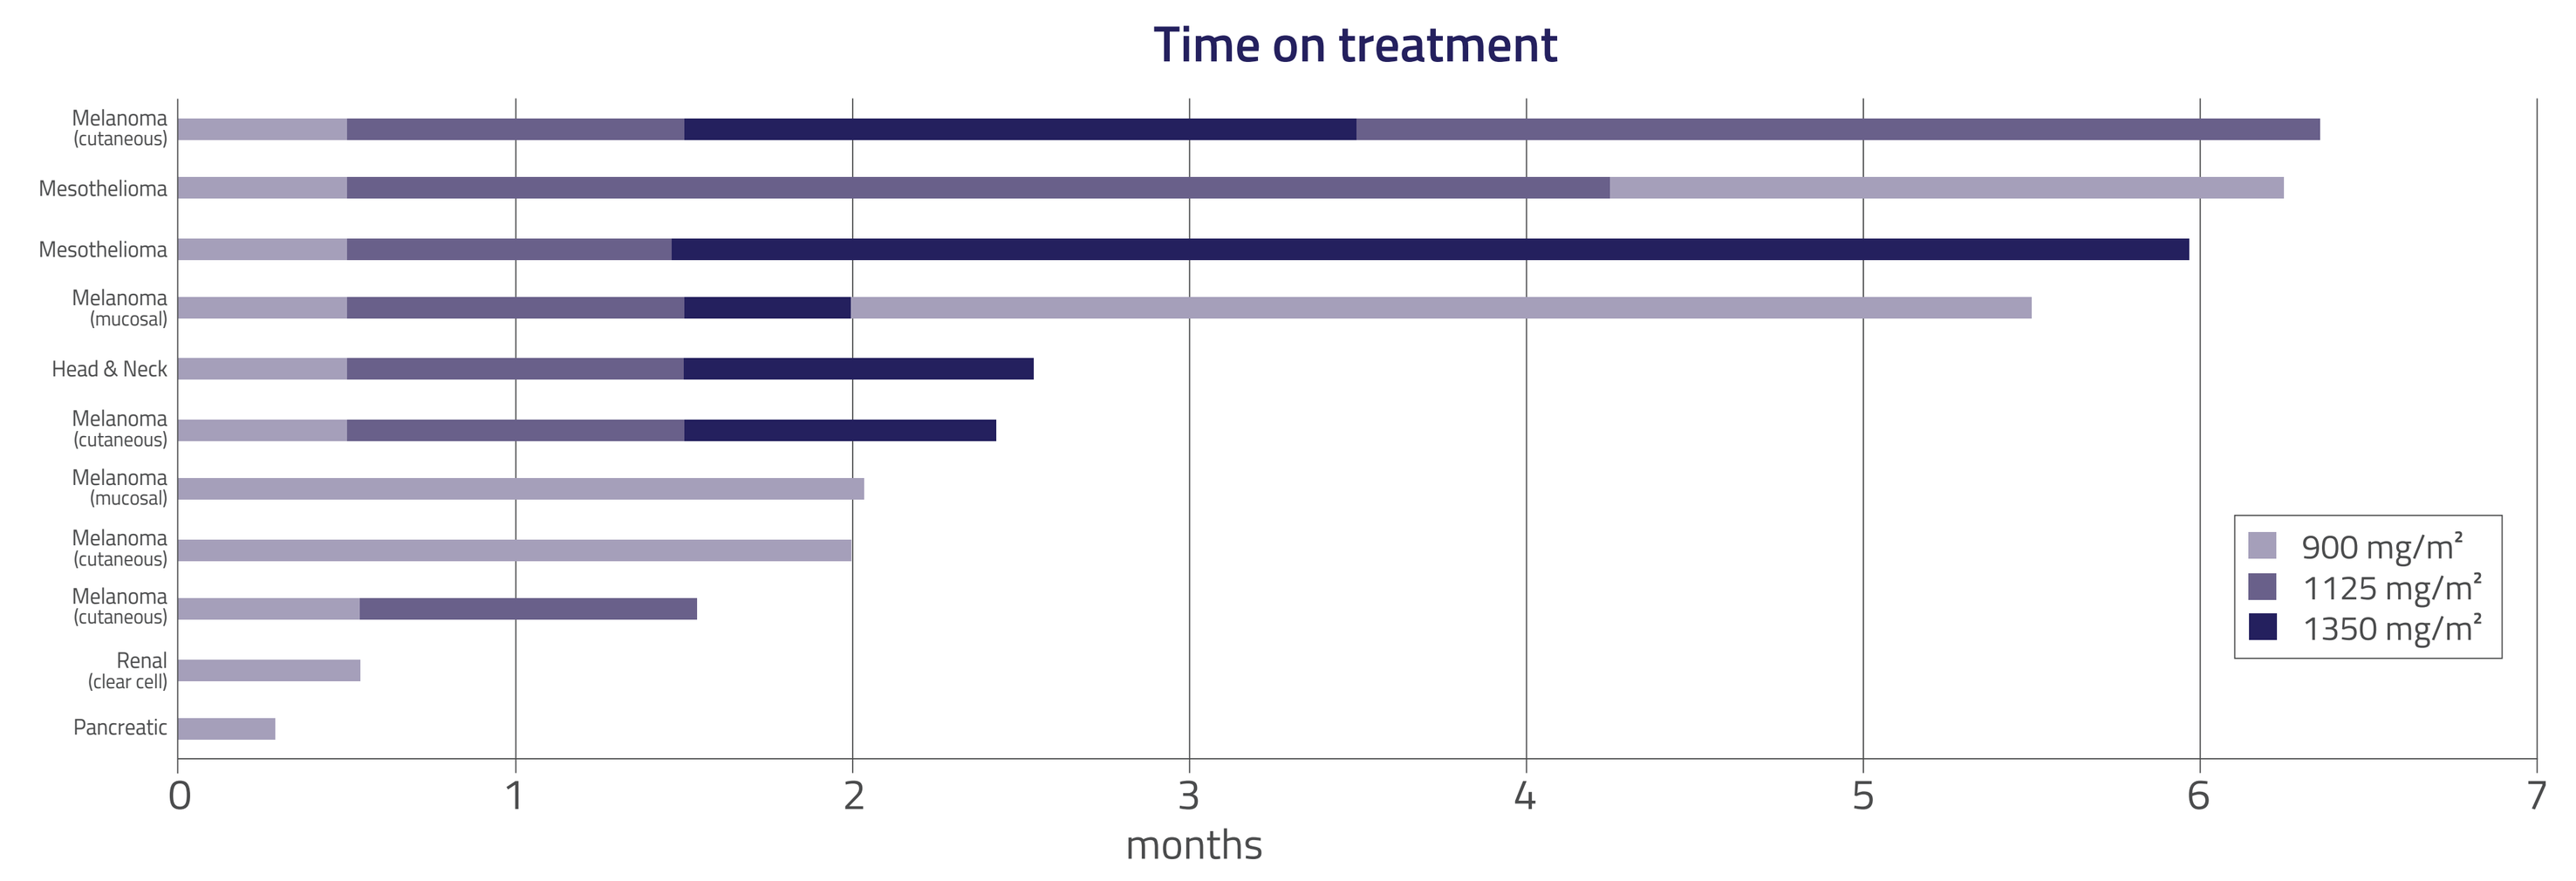
Time on treatment
Melanoma (cutaneous)
Mesothelioma
Mesothelioma
Melanoma (mucosal)
Head & Neck
Melanoma (cutaneous)
Melanoma (mucosal)
Melanoma (cutaneous)
Melanoma (cutaneous)
Renal (clear cell)
Pancreatic
900 mg/m²
1125 mg/m²
1350 mg/m²
months

Phase 2: NUC-7738 + pembrolizumab (ongoing)

Treatment related AEs (n=11)

Preferred term	All Grades	Grade 3/4
Nausea	7 (64%)	0
ALT increased	4 (36%)	1 (9%)
Diarrhea	4 (36%)	1 (9%)
Vomiting	4 (36%)	1 (9%)
Anemia	3 (27%)	0
Fatigue	3 (27%)	0
AST increased	2 (18%)	1 (9%)
Blood magnesium decreased	2 (18%)	0
Blood potassium decreased	2 (18%)	0

Most frequent (≥10% population) NUC-7738 ± pembro related AEs

- 2 patients with G3 TRAEs; abdominal pain & diarrhea (1); vomiting (1)
- 1 patient with G4 TRAEs; ALT increased & AST increased
- 3 patients with treatment related SAEs; G3 diarrhea (1), G4 ALT & AST increased (1), G3 vomiting (1)
- 1 patient discontinued due to AE

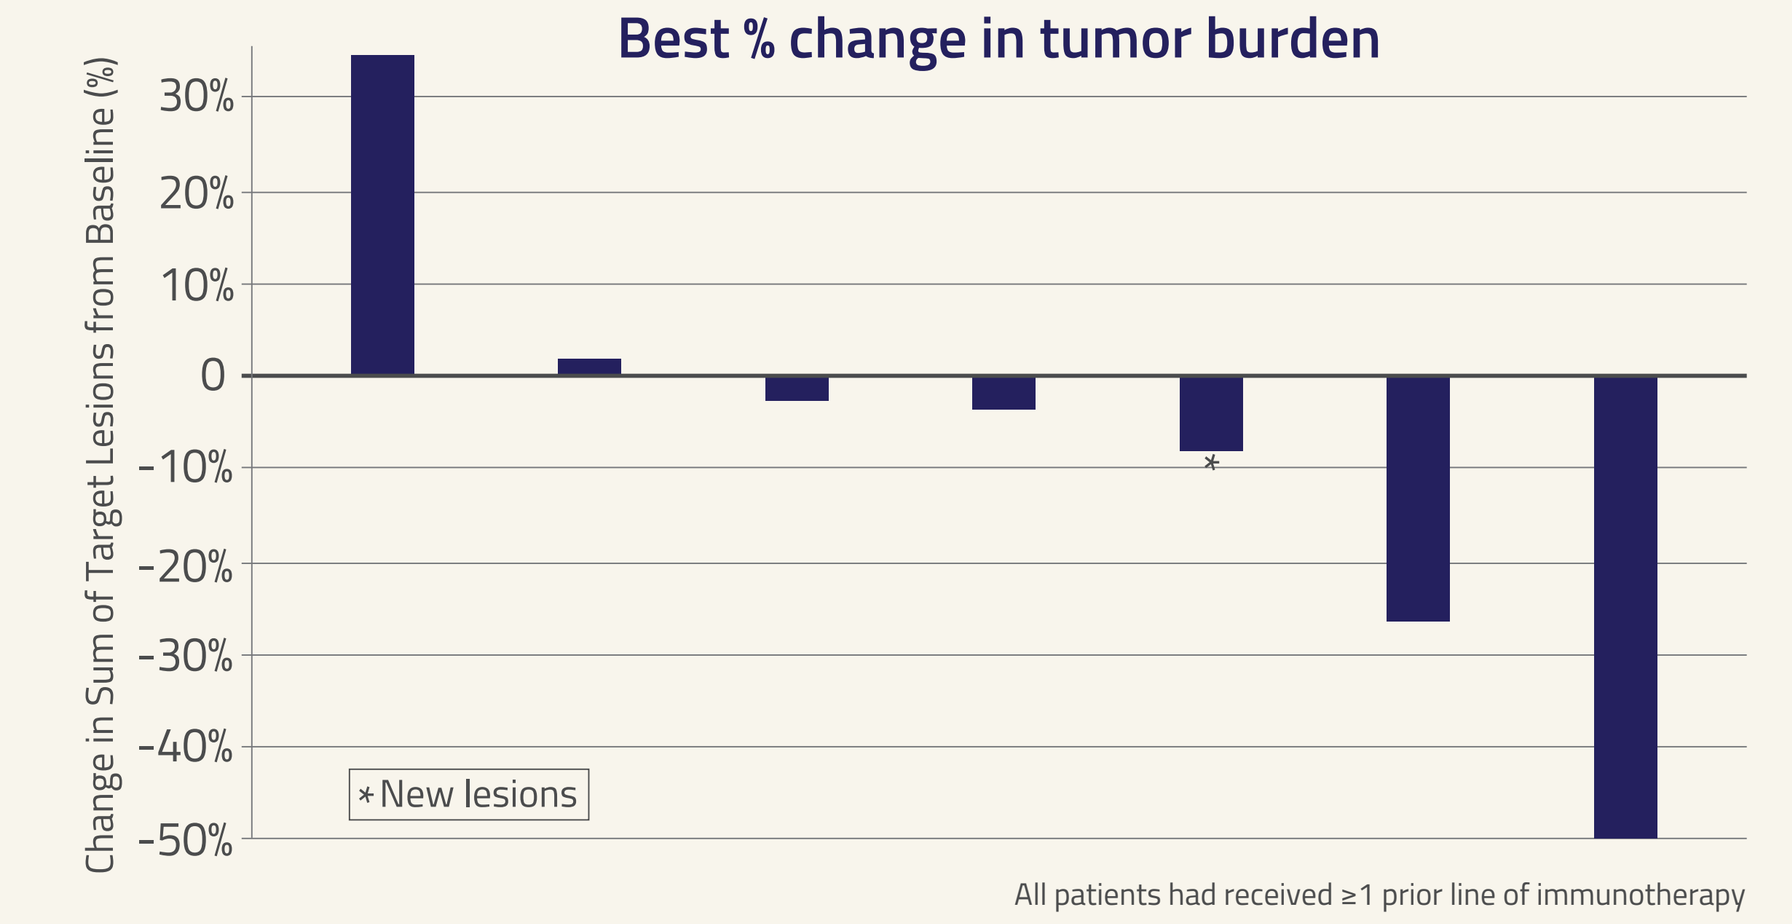
Best % change in tumor burden
Change in Sum of Target Lesions from Baseline (%)
*New lesions

All patients had received ≥1 prior line of immunotherapy

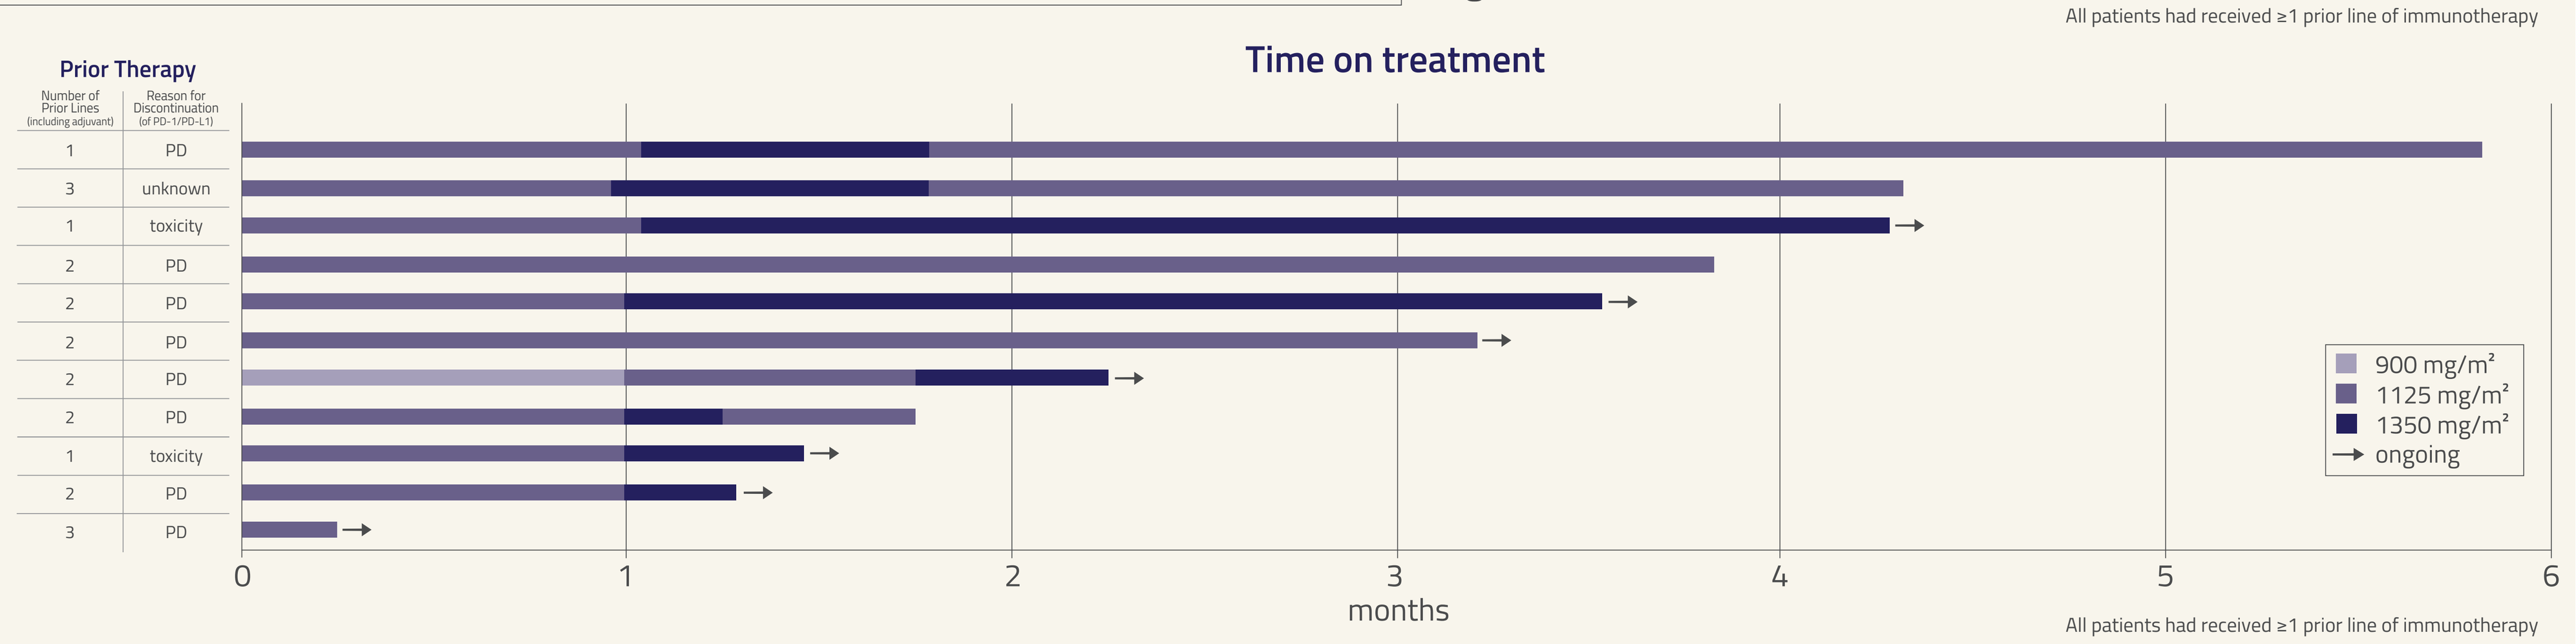
Prior Therapy
Number of Prior Lines (including adjuvant)
Reason for Discontinuation (of PD-1/PD-L1)
Time on treatment
1 PD
3 unknown
1 toxicity
2 PD
2 PD
2 PD
2 PD
2 PD
1 toxicity
2 PD
3 PD
900 mg/m²
1125 mg/m²
1350 mg/m²
ongoing
months

All patients had received ≥1 prior line of immunotherapy

CONCLUSION

- NUC-7738 anti-cancer metabolite, 3′-dATP, concentrations in patient PBMCs are comparable to those achieved *in vitro*
- NUC-7738 induced molecular changes observed in patient biopsies are similar to those identified *in vitro*
- NUC-7738 ± pembrolizumab has a tolerable safety profile
- NUC-7738 ± pembrolizumab demonstrates encouraging signs of anti-tumor activity in patients with advanced melanoma who have received prior immunotherapies

REFERENCES: 1. Mahoney, *et al.* 2019, *Cancer Immunol Immunother;* 68(3): 421-432 2. Kudsy *et al.* 2019, Mol Cancer Ther, 18(12_Suppl):C122 3. Schwenzer *et al.* 2021, *Clin Cancer Res,* 27(23):6500-6513 4. Shahid *et al.* 2021, *Mol Cancer Ther,* 20(12_Suppl): P026 5. Shahid *et al.* 2022, *PLoS ONE,* 17(12):e0278209 6. Elshani *et al.* 2023, *Cancer Res* (2023) 83 (7_Supplement): 5962.

ABBREVIATIONS: 2′-dATP: 2′-deoxyadenosine triphosphate 3′-dA: 3′-deoxyadenosine 3′-dADP: 3′-deoxyadenosine diphosphate 3′-dAMP: 3′-deoxyadenosine monophosphate 3′-dATP: 3′-deoxyadenosine triphosphate AE: adverse event ALT: alanine aminotransferase AST: aspartate aminotransferase BRAF: v-raf murine sarcoma viral oncogene homolog B1 CD8: cluster of differentiation 8 C_{max}: maximal concentration DMSO: dimethyl sulfoxide H&E: hematoxylin & eosin IC_{50}: half maximal inhibitory concentration ICI: immune checkpoint inhibitor IV: intravenous LC-MS: liquid chromatography mass spectrometry LLOQ: lower limit of quantification MITF: microphthalmia-associated transcription factor MEK: mitogen-activated protein kinase kinase PBMC: peripheral blood mononuclear cell PD: progressive disease per RECISTv1.1 PD-1: programmed cell death protein 1 PD-L1: programmed death-ligand 1 PK: pharmacokinetics RAB27A: ras-related protein Rab-27A RNA: ribonucleic acid TPM: transcripts per million TRAE: treatment related adverse event SAE: serious adverse event

Copies of this poster obtained through QR (Quick Response) and/or text key codes are for personal use only and may not be reproduced without written permission of the authors.

Data cleaning ongoing: data cut-off date 19 September 2023